Predictors of Hypomethylating Agent Discontinuation Among Patients With Higher-Risk Myelodysplastic Syndromes

Amer M Zeidan,[1] Namita Joshi,[2] Hrishikesh Kale,[2] Wei-Jhih Wang,[2] Shelby Corman,[2] Kala Hill,[3] Tehseen Salimi,[3] Robert S Epstein[4]

[1]Yale University and Yale Cancer Center, New Haven, CT, USA; [2]OPEN Health, Bethesda, MD, USA; [3]Taiho Oncology, Inc., Princeton, NJ, USA; [4]Epstein Health LLC, Woodcliff Lake, NJ, USA

Background

- Hypomethylating agents (HMAs) are the standard of care for the treatment of patients with higher-risk myelodysplastic syndromes (MDS).[1]
- Real-world studies have shown that persistence with intravenous (IV) and subcutaneous (SC) HMAs among patients with higher-risk MDS is poor, with over one-third of treated patients receiving <4 cycles or having a ≥90-day gap in therapy,[2-6] despite recommendations for at least 4–6 cycles to elicit response in the absence of progression or unacceptable toxicity.[7]
- Our prior analyses using the Surveillance, Epidemiology, and End Results (SEER)-Medicare linked database found that 44% of higher-risk MDS patients initiating HMAs were non-persistent to HMAs.[5,6]
 - Further, patients who were HMA non-persistent had worse health outcomes and incurred higher direct healthcare costs than those who were HMA persistent.
 - Patients who were HMA persistent had a higher overall survival (13.8 months) than patients who were HMA non-persistent (9.5 months) and those with no HMA use (3.8 months).[5]
 - Patients who were HMA non-persistent incurred significantly higher total per-patient-per-month healthcare costs compared with patients who were HMA persistent ($18,039 vs $13,893, p<0.05).[6]

Objective

- This study explored the factors associated with early discontinuation of HMA therapy in patients with higher-risk MDS.

Methods

- This was an observational, retrospective cohort study using the 2010–2016 SEER-Medicare linked database.
 - The SEER program covers patients from 18 cancer registries and includes data on clinical and demographic information, and cause of death for people with cancer.
 - The Medicare component provides information recorded on Medicare claims for covered healthcare services from the time of Medicare eligibility until death.
- Patients were included who had a new diagnosis of MDS and an International Classification of Diseases for Oncology (3rd edition) code for refractory anemia with excess blasts (RAEB; a surrogate for higher-risk MDS) from 2011–2015, were ≥66 years of age at diagnosis, had received HMA therapy, and had a continuous follow-up after diagnosis until death or end of study period (December 31, 2016), whichever occurred first (**Figure 1**).
- Discontinuation was defined as stopping HMA therapy before 4 cycles.
- Mean, standard deviation (SD), median, and ranges were reported for continuous variables, which were compared between groups using t-test or Wilcoxon-Mann-Whitney test.
- Multivariable logistic regression was conducted to assess potential predictors of HMA discontinuation.

Results

Patient population

- In total, 664 patients with RAEB and treated with HMAs were included, with a median follow-up time after HMA initiation of 9.9 months (interquartile range, 4.6–17.0 months).
- Among all patients, the mean (SD) age at diagnosis was 77.8 (6.3) years, 62.2% of patients were male, and 85.5% of patients were non-Hispanic White.
- Overall, 193 (29.1%) patients discontinued before 4 cycles; of these, 91 (47.2%) discontinued after 1 cycle, 57 (29.5%) after 2 cycles, and 45 (23.3%) after 3 cycles.

Figure 1. Cohort Selection for Patients With Higher-Risk MDS

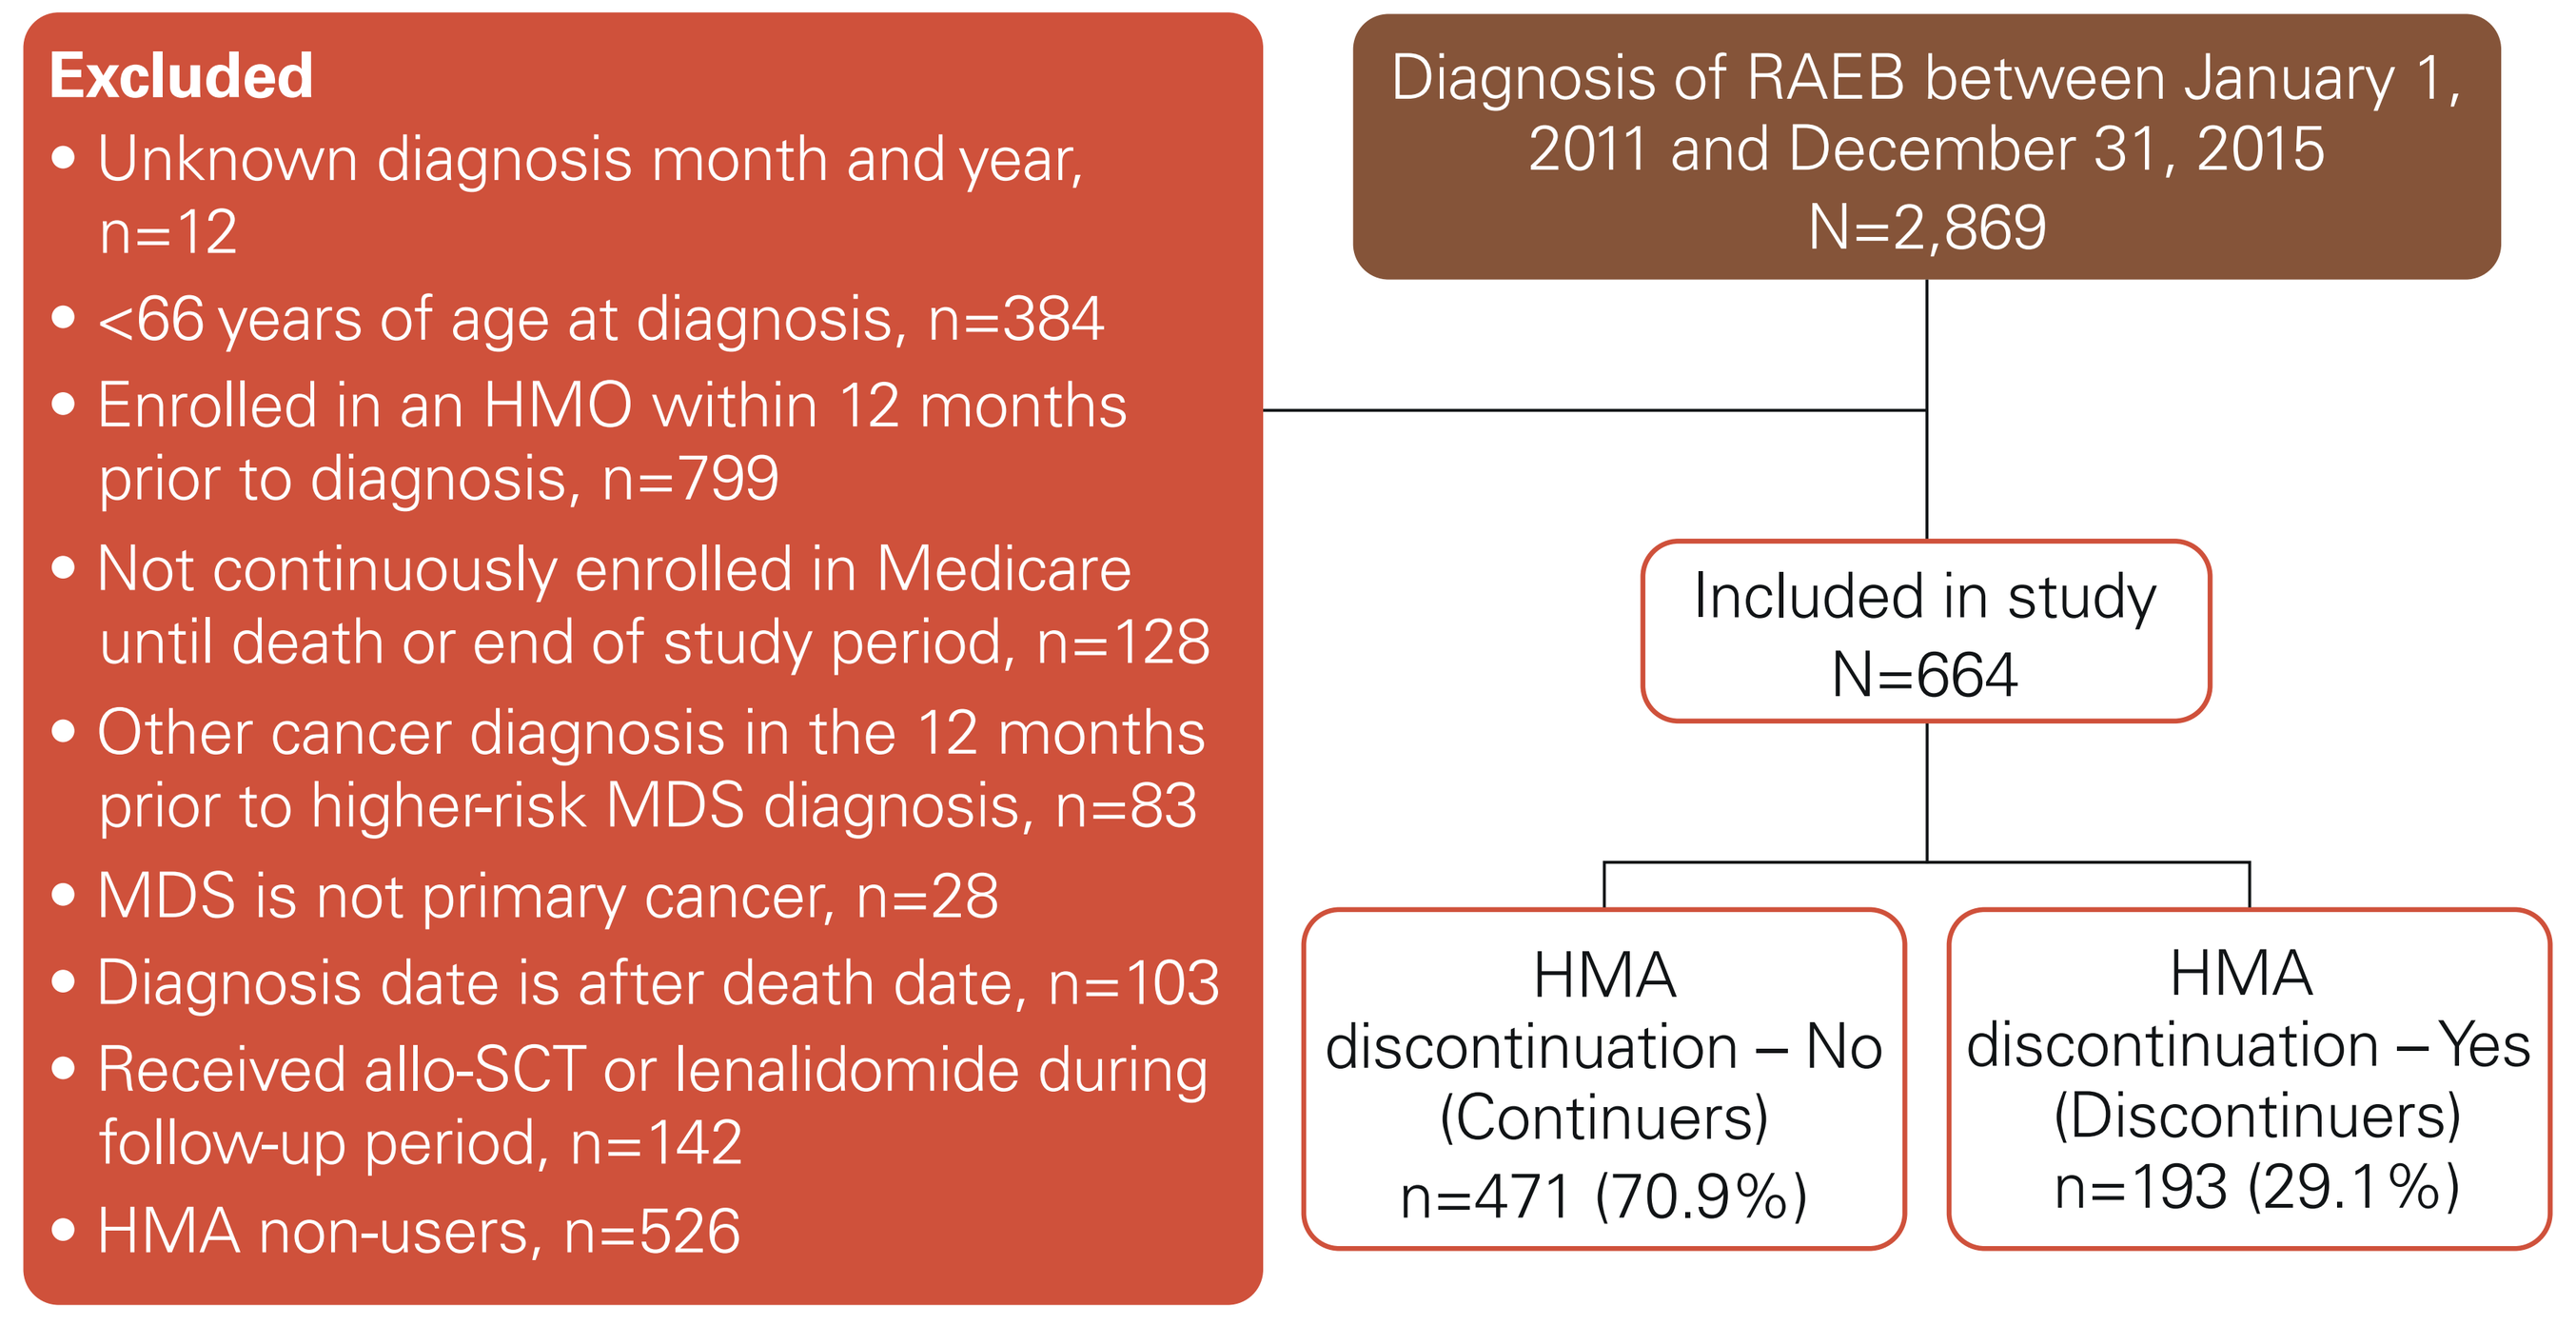
Excluded
• Unknown diagnosis month and year, n=12
• <66 years of age at diagnosis, n=384
• Enrolled in an HMO within 12 months prior to diagnosis, n=799
• Not continuously enrolled in Medicare until death or end of study period, n=128
• Other cancer diagnosis in the 12 months prior to higher-risk MDS diagnosis, n=83
• MDS is not primary cancer, n=28
• Diagnosis date is after death date, n=103
• Received allo-SCT or lenalidomide during follow-up period, n=142
• HMA non-users, n=526
Diagnosis of RAEB between January 1, 2011 and December 31, 2015 N=2,869
Included in study N=664
HMA discontinuation – No (Continuers) n=471 (70.9%)
HMA discontinuation – Yes (Discontinuers) n=193 (29.1%)

allo-SCT, allogeneic stem cell transplant; HMA, hypomethylating agent; HMO, health maintenance organization; MDS, myelodysplastic syndromes; RAEB, refractory anemia with excess blasts.

Predictors of HMA discontinuation (unadjusted)

- Compared with patients who continued HMA therapy for ≥4 cycles, patients who discontinued before 4 cycles were generally older and more likely to be single/separated/divorced/widowed, have more comorbidities, and have poor performance status (PS) (**Table 1**).
- These trends were most pronounced among patients who discontinued HMA therapy after only 1 cycle vs ≥4 cycles (**Table 1**).

Predictors of HMA discontinuation (multivariable regression)

- In multivariable analysis, age 71–75 vs ≥80 years (odds ratio [OR] 0.556, p=0.017) and poor PS (OR 1.585, p=0.019) remained significant predictors of HMA discontinuation (**Figure 2**).
- Among treatment-related factors, the most statistically significant association with HMA discontinuation was observed for granulocyte colony stimulating factor use (OR 0.453, p<0.001).
- Number of pills per day was not a predictor of HMA discontinuation (OR 1.009, p=NS).

Figure 2. Multivariable Logistic Regression Analysis of Predictors of Early HMA Discontinuation

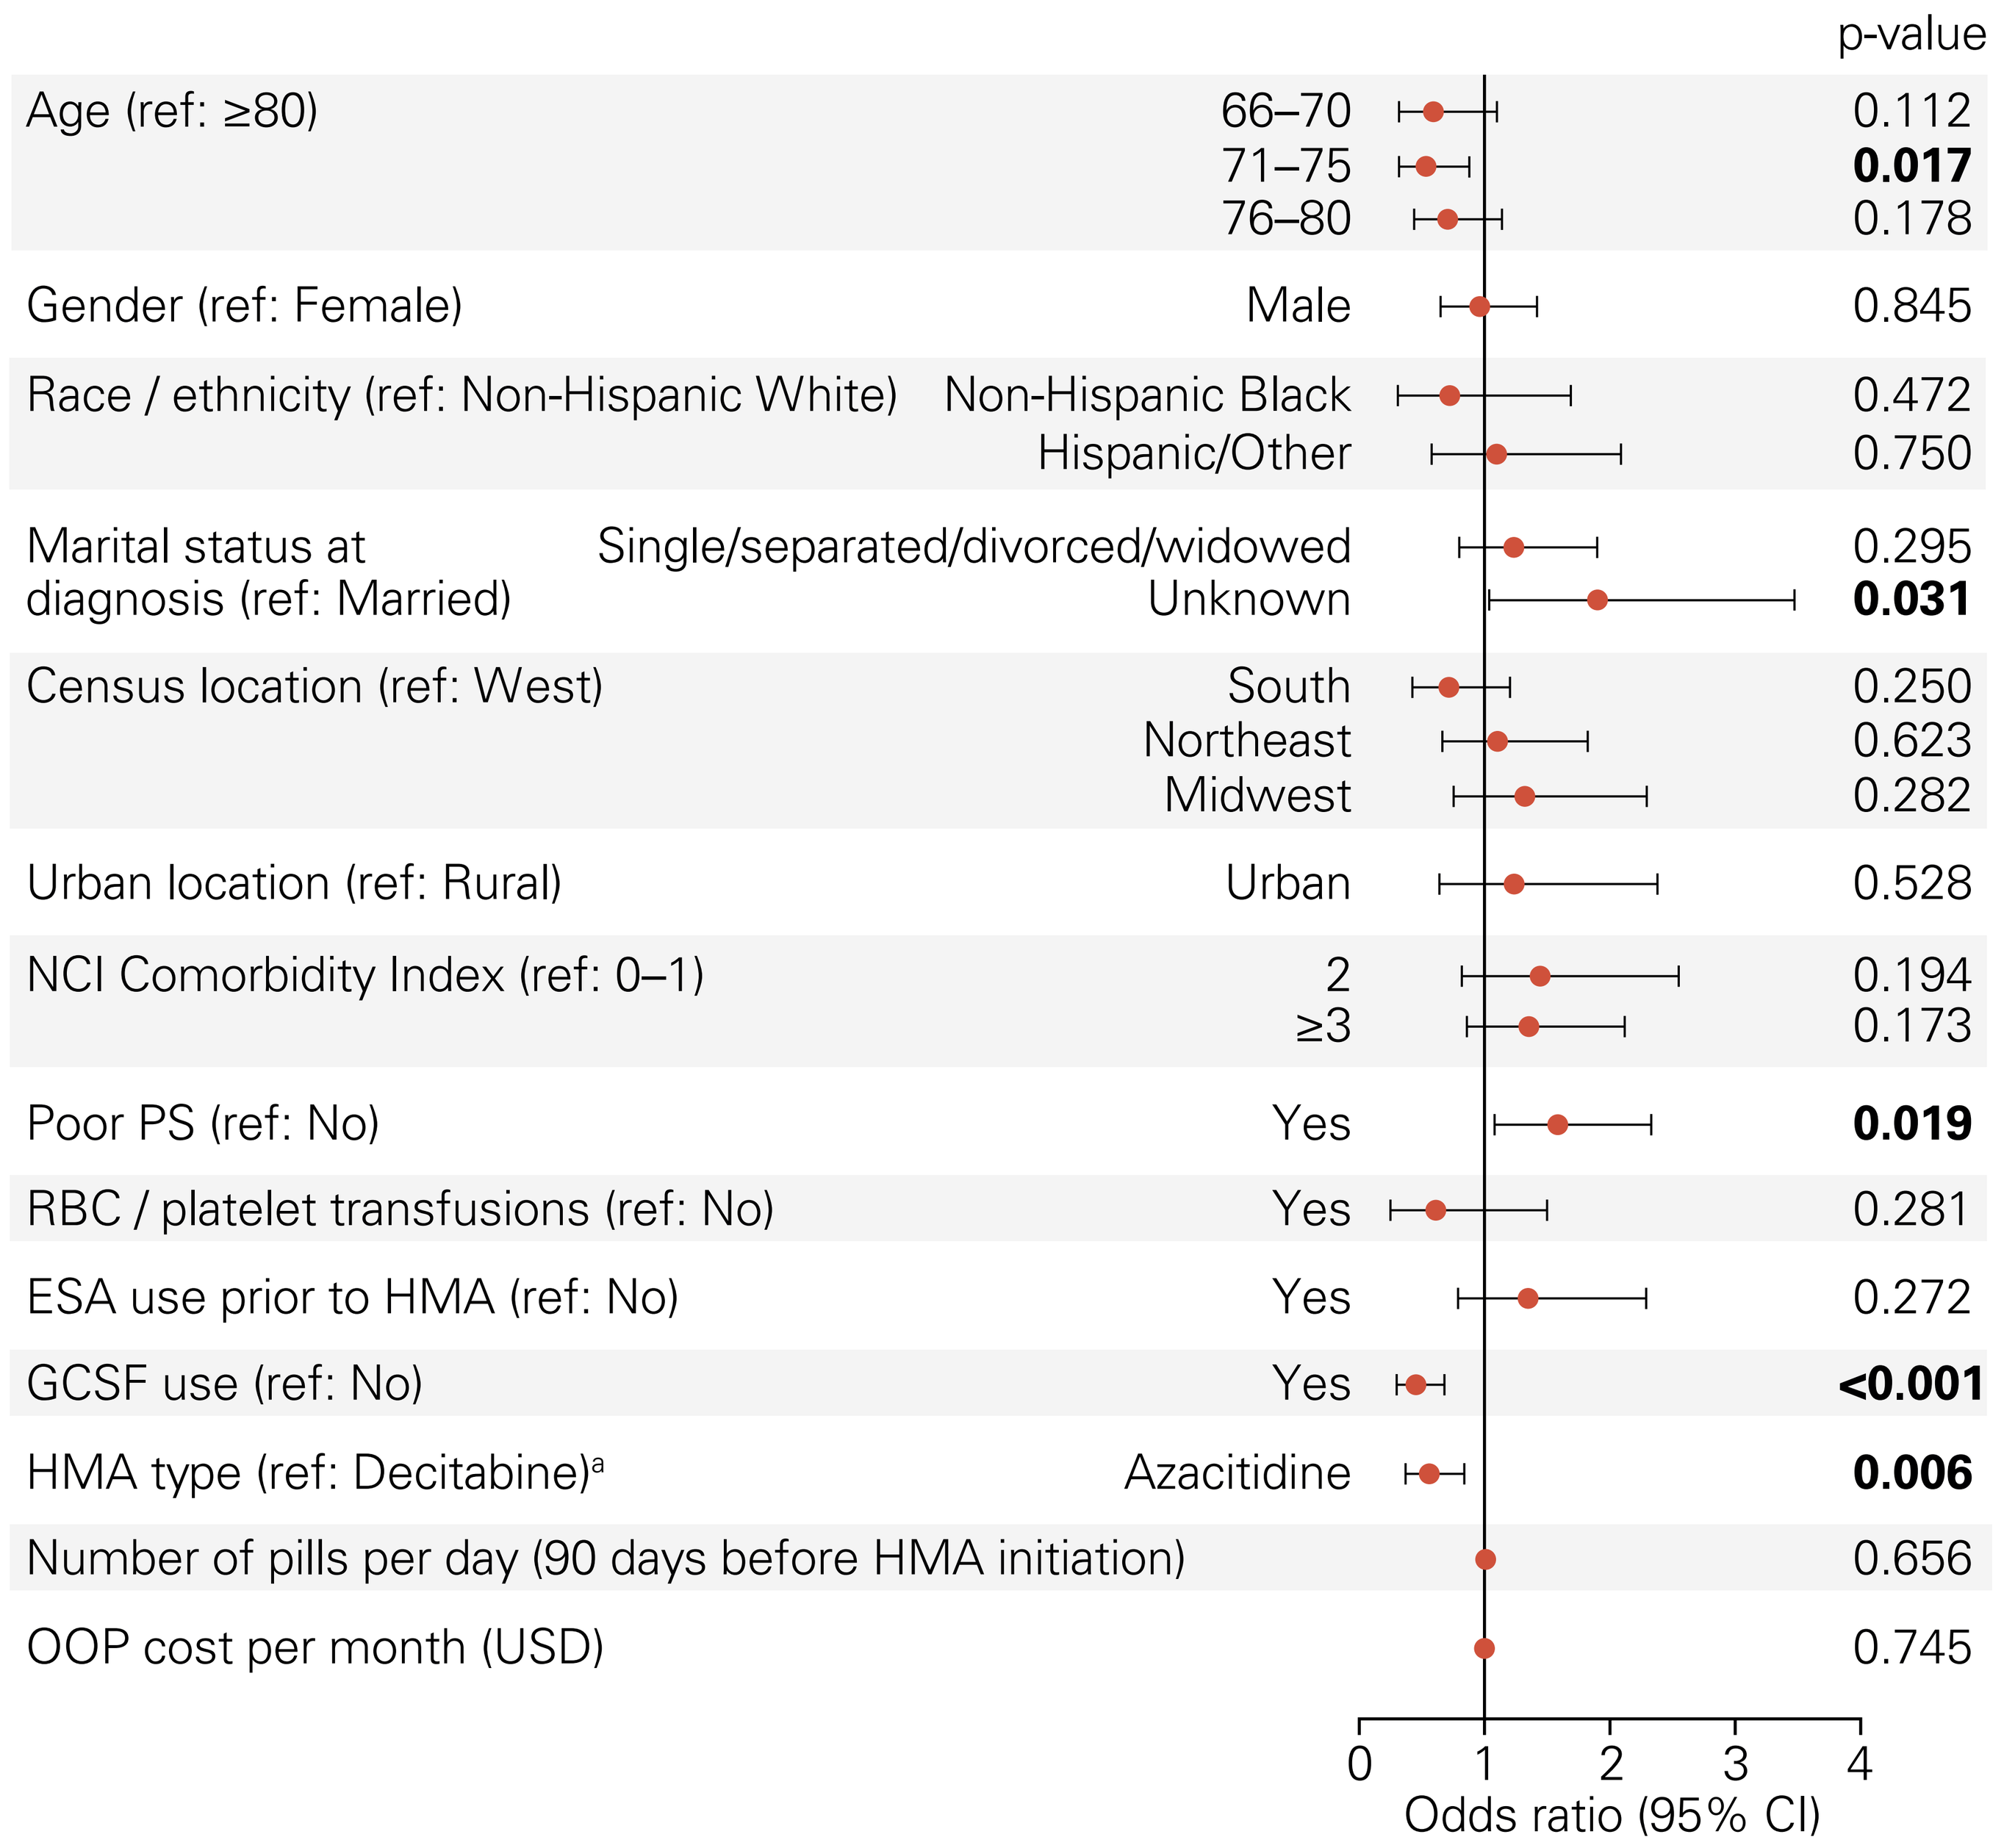
p-value
Age (ref: ≥80) 66–70 0.112
71–75 **0.017**
76–80 0.178
Gender (ref: Female) Male 0.845
Race / ethnicity (ref: Non-Hispanic White) Non-Hispanic Black 0.472
Hispanic/Other 0.750
Marital status at diagnosis (ref: Married) Single/separated/divorced/widowed 0.295
Unknown **0.031**
Census location (ref: West) South 0.250
Northeast 0.623
Midwest 0.282
Urban location (ref: Rural) Urban 0.528
NCI Comorbidity Index (ref: 0–1) 2 0.194
≥3 0.173
Poor PS (ref: No) Yes **0.019**
RBC / platelet transfusions (ref: No) Yes 0.281
ESA use prior to HMA (ref: No) Yes 0.272
GCSF use (ref: No) Yes **<0.001**
HMA type (ref: Decitabine)[a] Azacitidine **0.006**
Number of pills per day (90 days before HMA initiation) 0.656
OOP cost per month (USD) 0.745
0 1 2 3 4
Odds ratio (95% CI)

CI, confidence interval; ESA, erythropoiesis-stimulating agent; GCSF, granulocyte colony stimulating factor; HMA, hypomethylating agent; NCI, National Cancer Institute; OOP, out-of-pocket; PS, performance status; RBC, red blood cell; USD, US dollars.
p-values in bold font denote statistical significance (p<0.05).
[a]The HMA-type variable was created based on the first HMA use. Patients may have switched to the other HMA during the follow-up period (time period for which claims were available).

Table 1. Characteristics of Patients With Higher-Risk MDS Stratified by Number of HMA Cycles Completed Before Discontinuation

Variable	HMA discontinuation After 1 cycle (n=91)	HMA discontinuation After 2 cycles (n=57)	HMA discontinuation After 3 cycles (n=45)	HMA discontinuation Total <4 cycles (n=193)	No HMA discontinuation After ≥4 cycles (n=471)	p-value (<4 vs ≥4)
Age at diagnosis, mean (SD)	79.2 (6.0)	79.8 (7.2)	78.0 (6.5)	79.1 (6.5)	77.3 (6.2)	**<0.001[W]**
Marital status at diagnosis, n (%)						**0.024[C]**
Single/separated/divorced/widowed	35 (38.5)	19 (33.3)	14 (31.1)	68 (35.2)	139 (29.5)	
Married	43 (47.3)	31 (54.4)	26 (57.8)	100 (51.8)	294 (62.4)	
Unknown	13 (14.3)	7 (12.3)	5 (11.1)	25 (13.0)	38 (8.1)	
Charlson comorbidity index, n (%)						**0.004[C]**
0–1	38 (41.8)	29 (50.9)	29 (64.4)	96 (49.7)	299 (63.5)	
2	16 (17.6)	6 (10.5)	5 (11.1)	27 (14.0)	51 (10.8)	
≥3	37 (40.7)	22 (38.6)	11 (24.4)	70 (36.3)	121 (25.7)	
Poor performance status,[a] n (%)						**<0.001[C]**
Yes	57 (62.6)	25 (43.9)	24 (53.3)	106 (54.9)	187 (39.7)	
No	34 (37.4)	32 (56.1)	21 (46.7)	87 (45.1)	284 (60.3)	

C, Chi-square test; HMA, hypomethylating agent; MDS, myelodysplastic syndromes; W, Wilcoxon rank sum.
[a]Poor performance status was defined as a claim in the 12 months before MDS diagnosis for a performance status proxy (wheelchair use, oxygen use, walking aid, hospital bed, hospice, skilled nursing facility).

Limitations

- Generalizability of the study findings is limited to patients with MDS aged >65 years and not enrolled in health maintenance organization plans.
- The SEER-Medicare linked database has some limitations, including lack of clinical data such as specific blood cell counts and cytogenetics needed to calculate risk score, and the potential for inaccurate representation of conditions/procedures from claims data.
- Reasons for discontinuation of HMA therapy and other factors that may influence treatment persistence were not captured in the SEER-Medicare database. Further research is warranted to fully elucidate the reasons for early discontinuation in this population.

Conclusions

- In this real-world study, almost one-third of RAEB patients treated with IV/SC HMAs discontinued treatment before 4 cycles, with almost half of these patients discontinuing after only 1 cycle.
- Predictors of HMA discontinuation included older age and poor PS. Novel approaches are needed to improve persistence with HMA therapy and associated outcomes, particularly among these higher-risk groups.

References

1. Bewersdorf JP, et al. *Ther Adv Hematol*. Nov 2020. doi:10.1177/2040620720955006.
2. Zeidan AM, et al. *Leuk Lymphoma* 2020;61:397–408.
3. Stein EM, et al. *Blood* 2019;134(suppl 1):3495.
4. Steensma DP, et al. *Cancer* 2014;120:1670–6.
5. Corman S, et al. *Clin Lymphoma Myeloma Leuk* 2021;21:e206–e211.
6. Joshi N, et al. *Clin Lymphoma Myeloma Leuk* 2021;21:e248–e254.
7. Greenberg PL, et al. *J Natl Compr Canc Netw* 2017;15:60–87.

Disclosures

This study was funded by Taiho Oncology, Inc. AM Zeidan has participated in advisory boards, and/or had a consultancy with and received honoraria from AbbVie, Otsuka, Pfizer, Celgene/BMS, Jazz, Incyte, Agios, Boehringer Ingelheim, Novartis, Acceleron, Astellas, Daiichi Sankyo, Cardinal Health, Taiho Oncology, Seattle Genetics, BeyondSpring, Trovagene, Takeda, Ionis, Amgen, Janssen, Epizyme, Syndax, and Tyme; has received research funding (institutional) from Celgene/BMS, Abbvie, Astex, Pfizer, MedImmune/AstraZeneca, Boehringer Ingelheim, Trovagene, Incyte, Takeda, Novartis, Aprea, and ADC Therapeutics; served on steering and independent data review committees for clinical trials for Novartis, AbbVie, and Janssen; and received travel support for meetings from Pfizer, Novartis, and Trovagene. N Joshi, H Kale, W-J Wang, and S Corman are employees of OPEN Health, paid consultants to Taiho Oncology, Inc. in connection with this study. K Hill and T Salimi are employees of Taiho Oncology, Inc. K Hill is a stockholder of Johnson & Johnson and Celldex Therapeutics. RS Epstein is an employee of Epstein Health LLC; holds leadership positions in Fate Therapeutics, Illumina, and Veracyte; and has received consulting fees from Merck, Radius Health, Halozyme, Intra-Cellular Therapies, Taiho Oncology, and Otsuka.

Acknowledgments

Medical writing support was provided by Ann Cameron, PhD, of Curo, a division of Envision Pharma Group, and funded by Taiho Oncology, Inc.

Corresponding author:

Amer M. Zeidan (amer.zeidan@yale.edu).

Please scan this Quick Response (QR) code with your smartphone app to view an electronic version of this poster. If you don't have a smartphone, access the poster via the internet at: https://bit.ly/2XMsAfS

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from the authors of this poster.

Poster presented at the National Community Oncology Dispensing Association (NCODA) 2021 Fall Summit, October 20–22, 2021, Scottsdale, AZ

© Copyright 2021