Hospitalization and Emergency Room Visits Among Patients With Higher-Risk Myelodysplastic Syndromes by Hypomethylating Agent Use

Amer M Zeidan,[1] Namita Joshi,[2] Hrishikesh Kale,[2] Wei-Jhih Wang,[2] Shelby Corman,[2] Kala Hill,[3] Tehseen Salimi,[3] Robert S Epstein[4]

[1]Yale University and Yale Cancer Center, New Haven, CT, USA; [2]OPEN Health, Bethesda, MD, USA; [3]Taiho Oncology, Inc., Princeton, NJ, USA; [4]Epstein Health LLC, Woodcliff Lake, NJ, USA

Background

- Hypomethylating agents (HMAs) are the standard-of-care, guideline-recommended treatment for patients with higher-risk myelodysplastic syndromes (MDS).[1]
- In the United States (US), the HMAs azacitidine, decitabine, and more recently the oral combination of decitabine and cedazuridine, are approved by the US Food and Drug Administration for the treatment of adult patients with MDS.[2-4]
- While HMAs provide clear therapeutic benefit,[5-7] previous analyses have shown underutilization of intravenously or subcutaneously administered HMAs in clinical practice.[8,9]
 - Our prior real-world analyses using the Surveillance, Epidemiology, and End Results (SEER)-Medicare linked database found that 526 of 1,190 higher-risk MDS patients (44%) did not use HMA therapy, and of those who received treatment, 295 (25%) were non-persistent HMA users.[10]
 - Factors significantly associated with HMA non-use were older age at diagnosis, unmarried status, a greater number of comorbidities, and poor performance status.[10]
 - Non-use of HMA therapy was associated with impaired survival; median overall survival was 3.8 months in non-users and 12.3 months in HMA users.[10]

Objective

- To better understand the impact of HMA non-use on healthcare resource utilization, this study compared the rate of hospitalizations and emergency room (ER) visits among higher-risk MDS patients according to use or non-use of HMA therapy.

Methods

Study design and data source

- This was an observational, retrospective cohort study using data from the 2010–2016 SEER-Medicare linked database.[11]
 - The SEER dataset covers patients from 18 cancer registries and includes data on clinical and demographic information, and cause of death for people with cancer.
 - The Medicare component provides information recorded on Medicare claims for covered healthcare services from the time of Medicare eligibility until death.
- The study design is summarized in **Figure 1**.

Eligibility criteria

- Patients with a new diagnosis of MDS and an International Classification of Diseases for Oncology (3rd edition) code 9983/3 for refractory anemia with excess blasts (RAEB; a surrogate for higher-risk MDS) between January 1, 2011 and December 31, 2015.
- Age ≥66 years at diagnosis.
- Continuously enrolled in Medicare Part A (inpatient/hospital coverage) and Part B (outpatient/medical coverage) for ≥12 months before initial MDS diagnosis and until death or end of the study period (December 31, 2016), whichever occurred first.
- Exclusion criteria included:
 - Any other cancer diagnosis in the 12 months before MDS diagnosis.
 - Enrollment in a health maintenance organization (HMO) in the 12 months before and including the month of diagnosis.
 - Allogeneic hematopoietic stem cell transplant or lenalidomide at any time during the follow-up period.

Outcomes

- Hospitalizations and ER visits were defined as the number of new hospitalizations/ER visits from MDS diagnosis to the 12 months after MDS diagnosis.
- Frequency and percentage distributions were reported for hospitalizations and ER visits among higher-risk MDS patients at 1 year, including the number of inpatient cases and the number of hospitalizations and ER visits.

Statistical analyses

- Descriptive statistics were used to summarize patient characteristics.
- Multivariable zero-inflated negative binomial regression models were used to assess the relationship between HMA use and the number of hospitalizations and ER visits.
- Separate models were used to study the number of hospitalizations (count variable), and any hospitalization (dichotomous variable [yes/no]). Similar models were used for ER visits.
- All multivariable models controlled for patient demographics and clinical characteristics as additional covariates.

Results

Patient population

- Overall, 2,869 patients had a diagnosis of RAEB during the study period, and 1,190 patients were included in the study cohort. Of these, 664 (55.8%) were HMA users and 526 (44.2%) were non-users (**Figure 2**).
- Among all patients (n=1,190), the mean (standard deviation) age at diagnosis was 79.4 (6.9) years, 60.1% of patients were male, and 84.4% of patients were non-Hispanic White (**Table 1**).

Poster presented at the National Community Oncology Dispensing Association (NCODA) 2021 Fall Summit, October 20–22, 2021, Scottsdale, AZ.

© Copyright 2021

Figure 1. Study Design

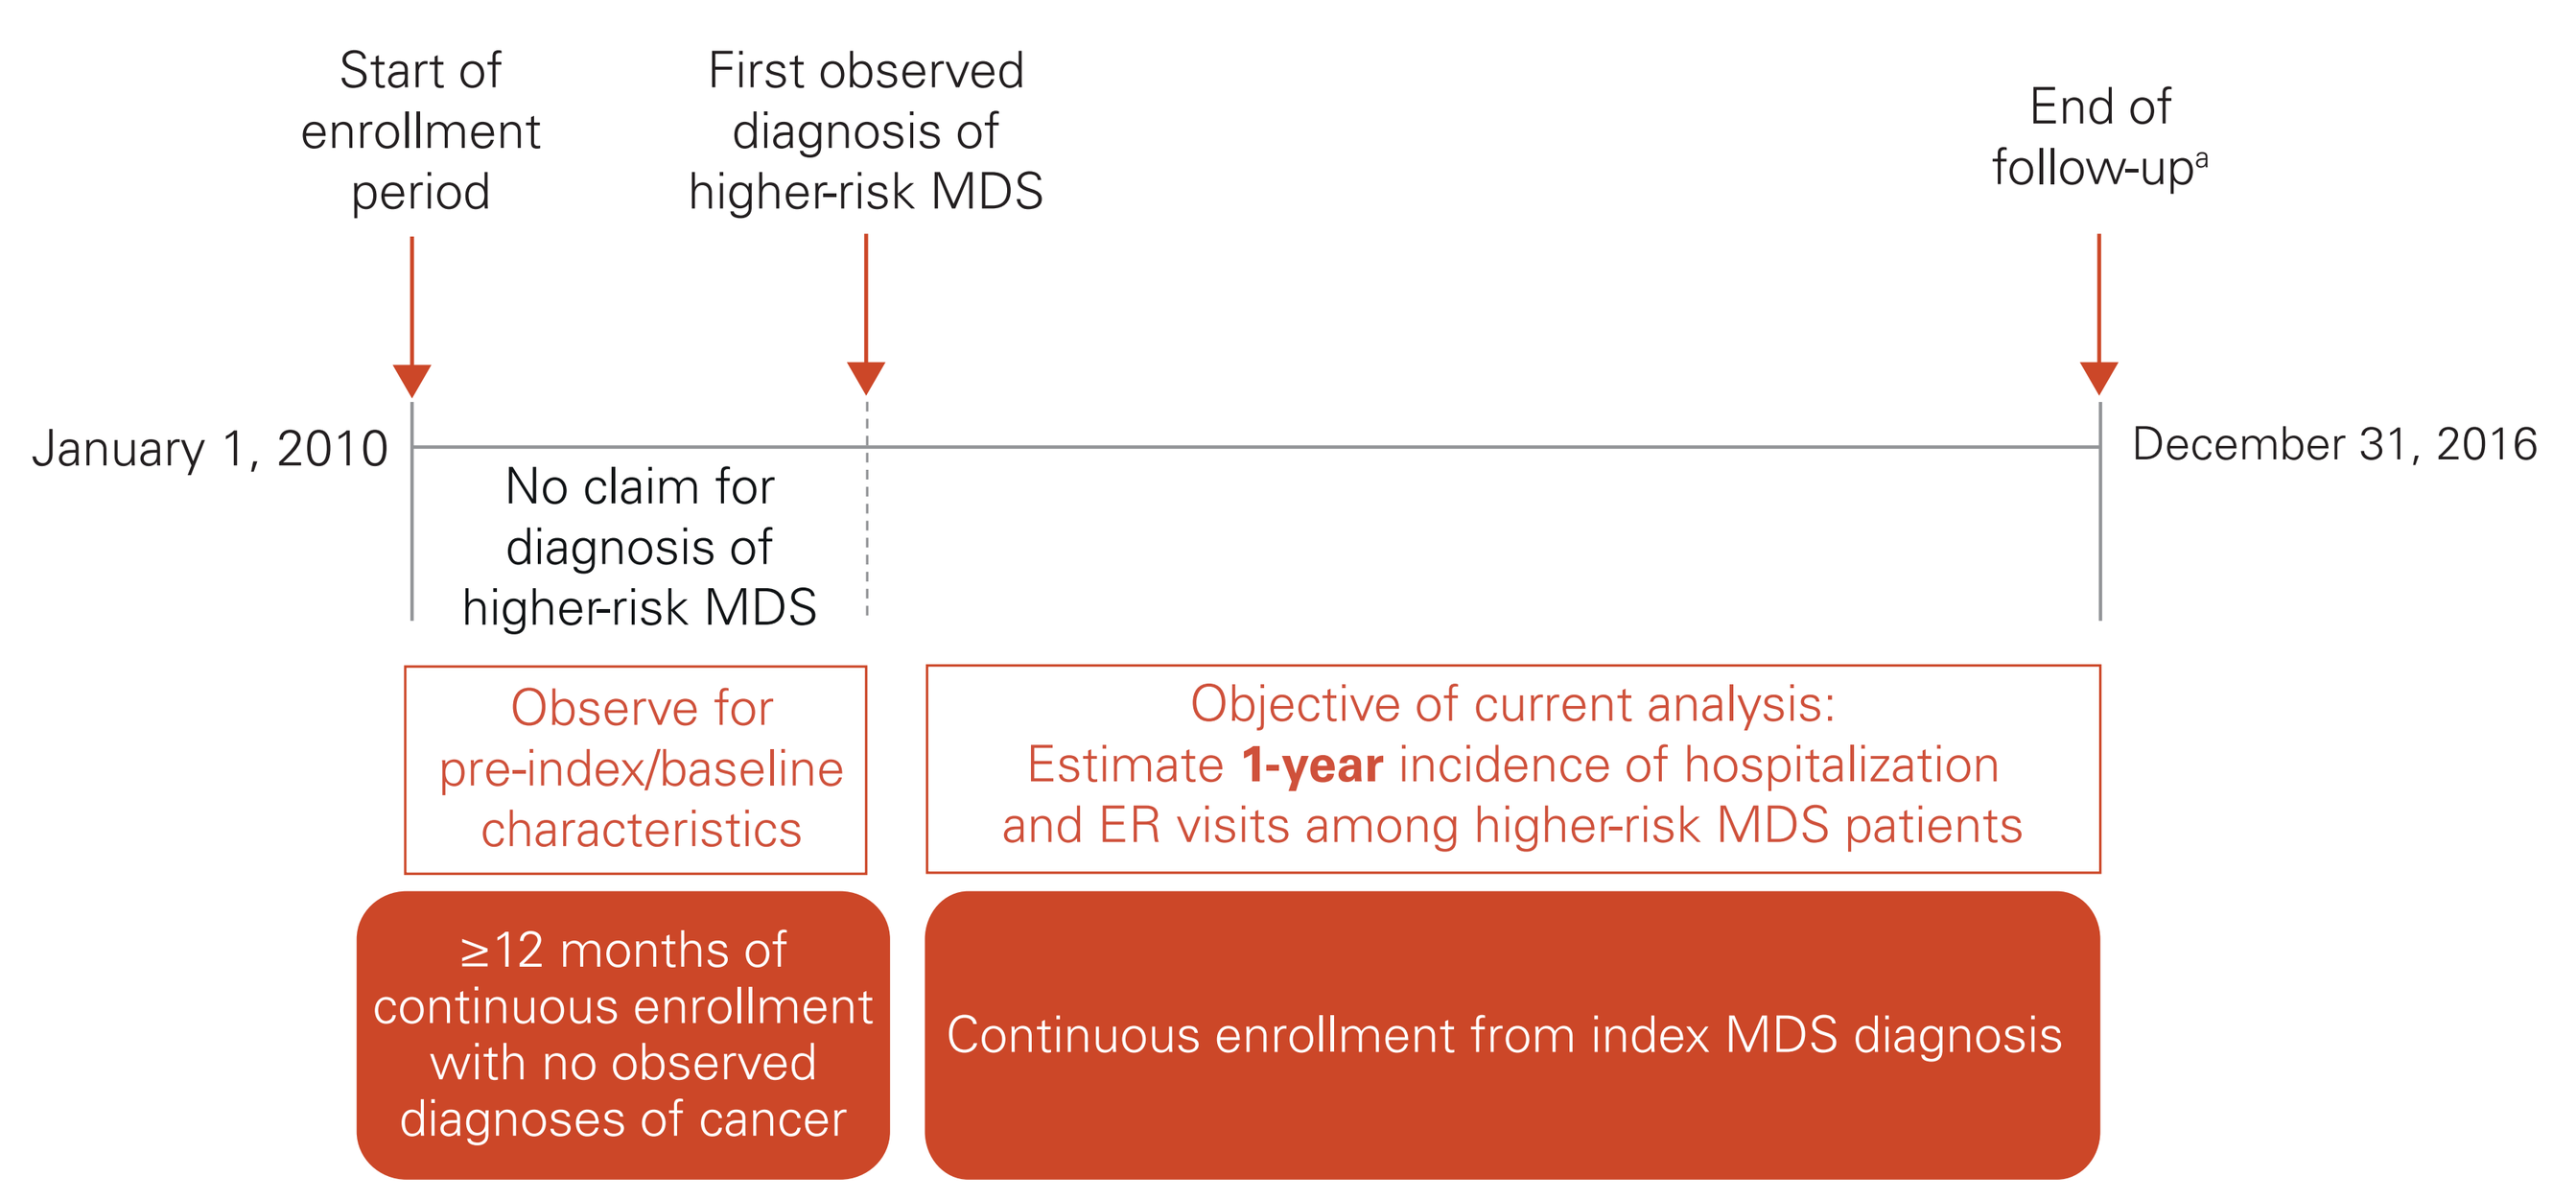

Start of enrollment period
First observed diagnosis of higher-risk MDS
End of follow-up[a]
January 1, 2010
December 31, 2016
No claim for diagnosis of higher-risk MDS
Observe for pre-index/baseline characteristics
Objective of current analysis: Estimate 1-year incidence of hospitalization and ER visits among higher-risk MDS patients
≥12 months of continuous enrollment with no observed diagnoses of cancer
Continuous enrollment from index MDS diagnosis

[a]Patients were censored based on occurrence of an outcome, end of data cut (administrative censoring), drop-out of enrollment, death, HMO enrollment, Medicare disenrollment, or loss to follow-up.
ER, emergency room; HMO, health maintenance organization; MDS, myelodysplastic syndromes.

Figure 2. Cohort Selection for Higher-Risk MDS Patients

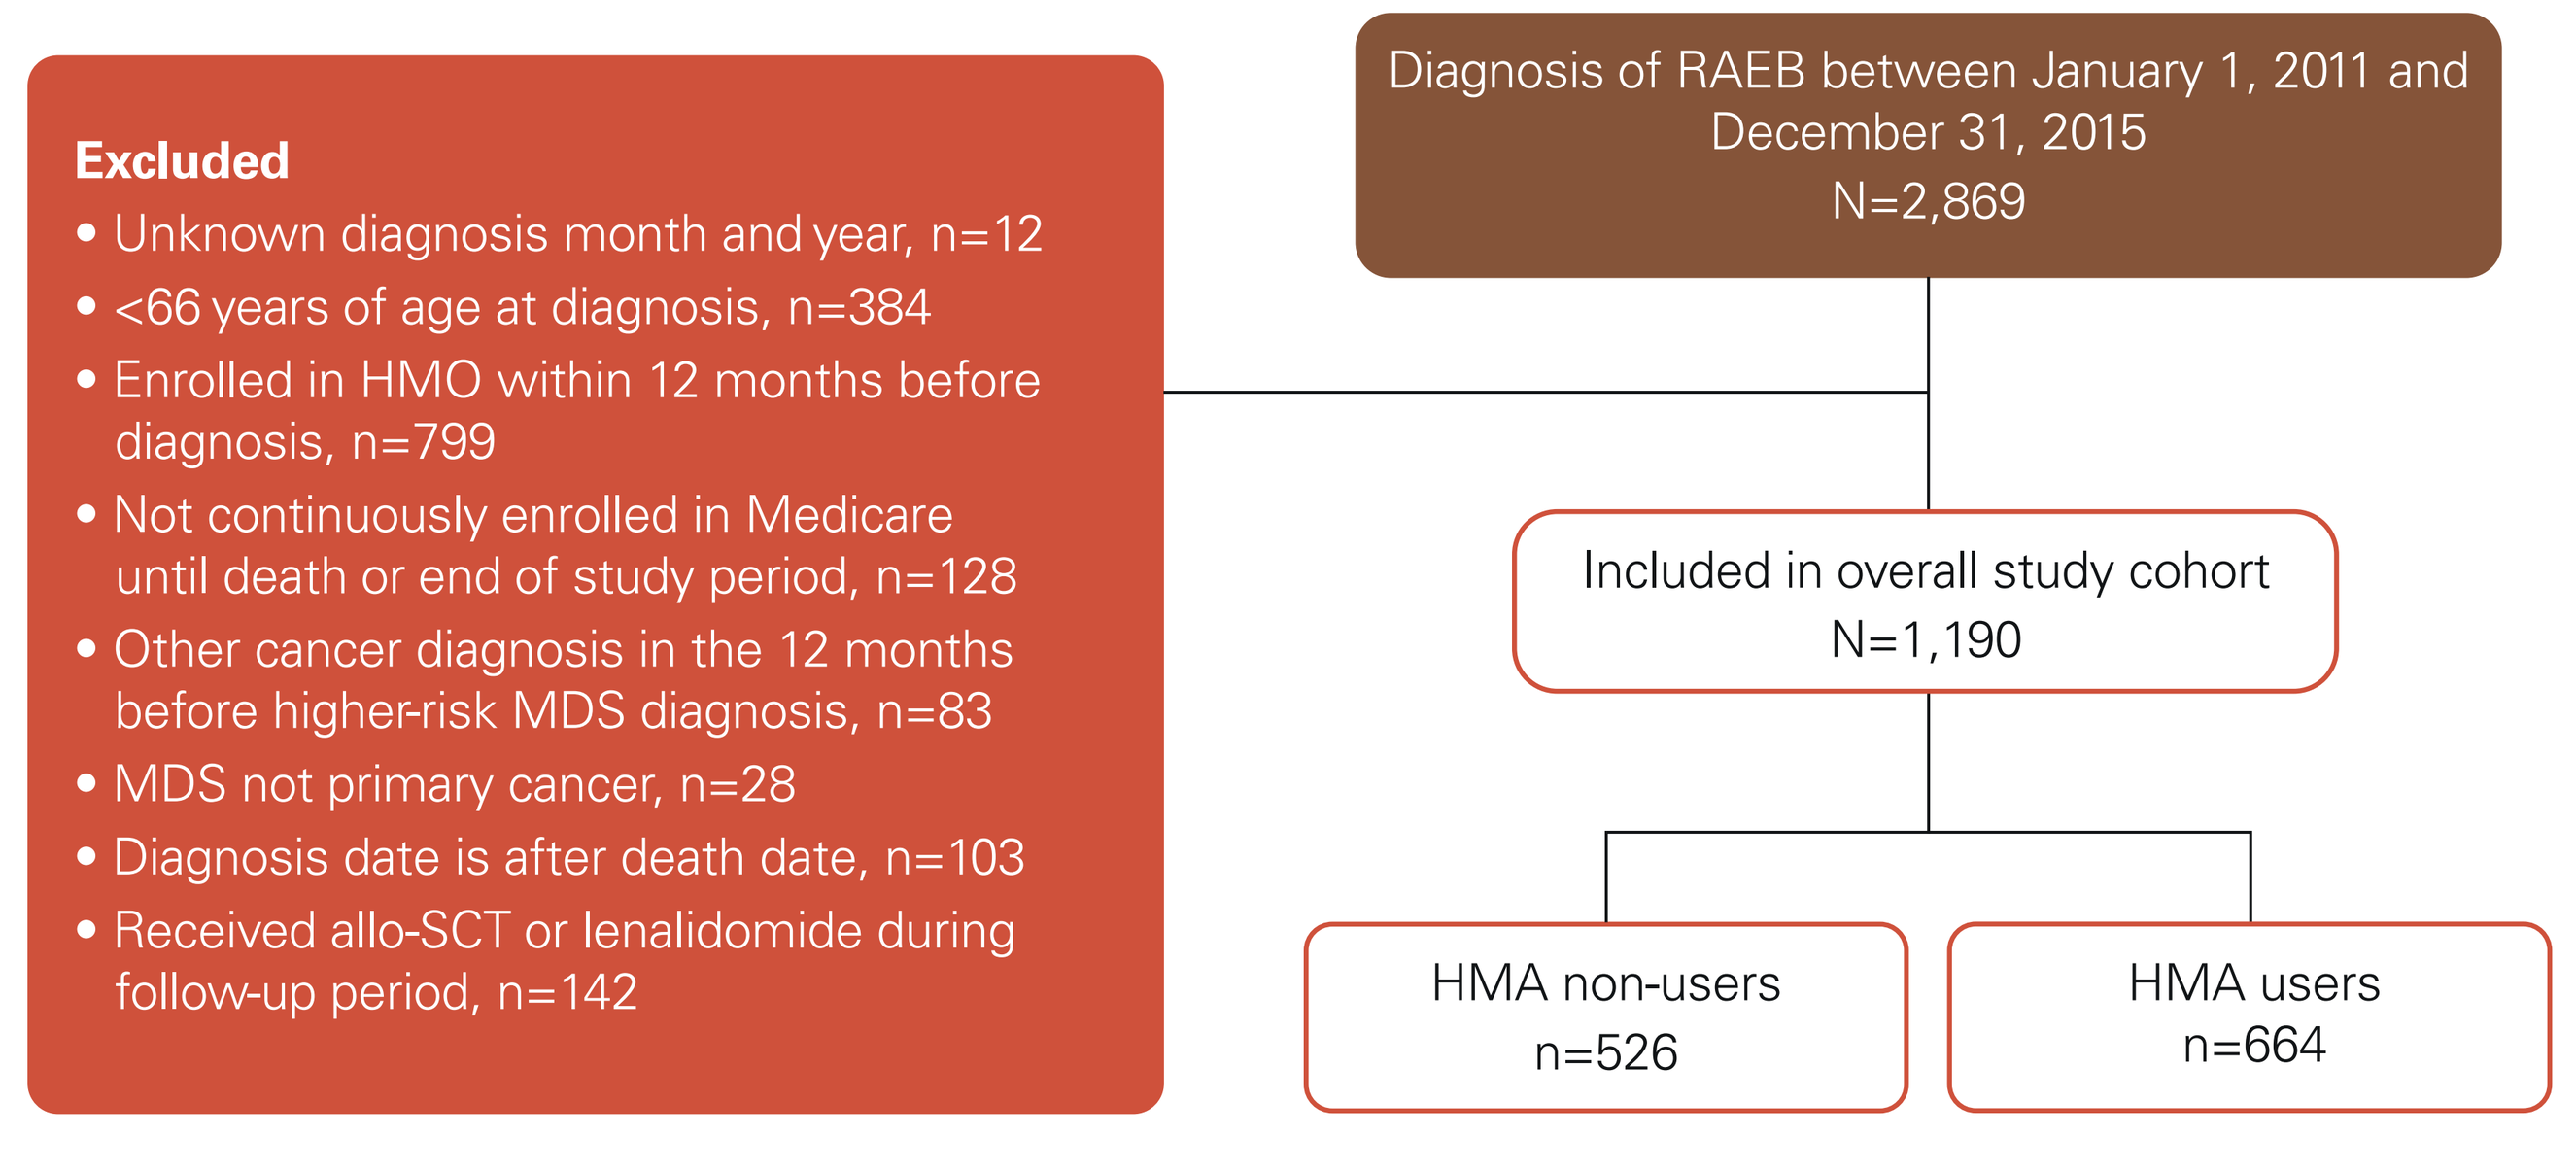

Diagnosis of RAEB between January 1, 2011 and December 31, 2015
N=2,869
Excluded
• Unknown diagnosis month and year, n=12
• <66 years of age at diagnosis, n=384
• Enrolled in HMO within 12 months before diagnosis, n=799
• Not continuously enrolled in Medicare until death or end of study period, n=128
• Other cancer diagnosis in the 12 months before higher-risk MDS diagnosis, n=83
• MDS not primary cancer, n=28
• Diagnosis date is after death date, n=103
• Received allo-SCT or lenalidomide during follow-up period, n=142
Included in overall study cohort
N=1,190
HMA non-users
n=526
HMA users
n=664

allo-SCT, allogeneic stem cell transplant; HMA, hypomethylating agent; HMO, health maintenance organization; MDS, myelodysplastic syndromes; RAEB, refractory anemia with excess blasts.

Figure 3. Mean Number of Hospitalizations and ER Visits per Month (Count/Follow-up Time), by HMA Use

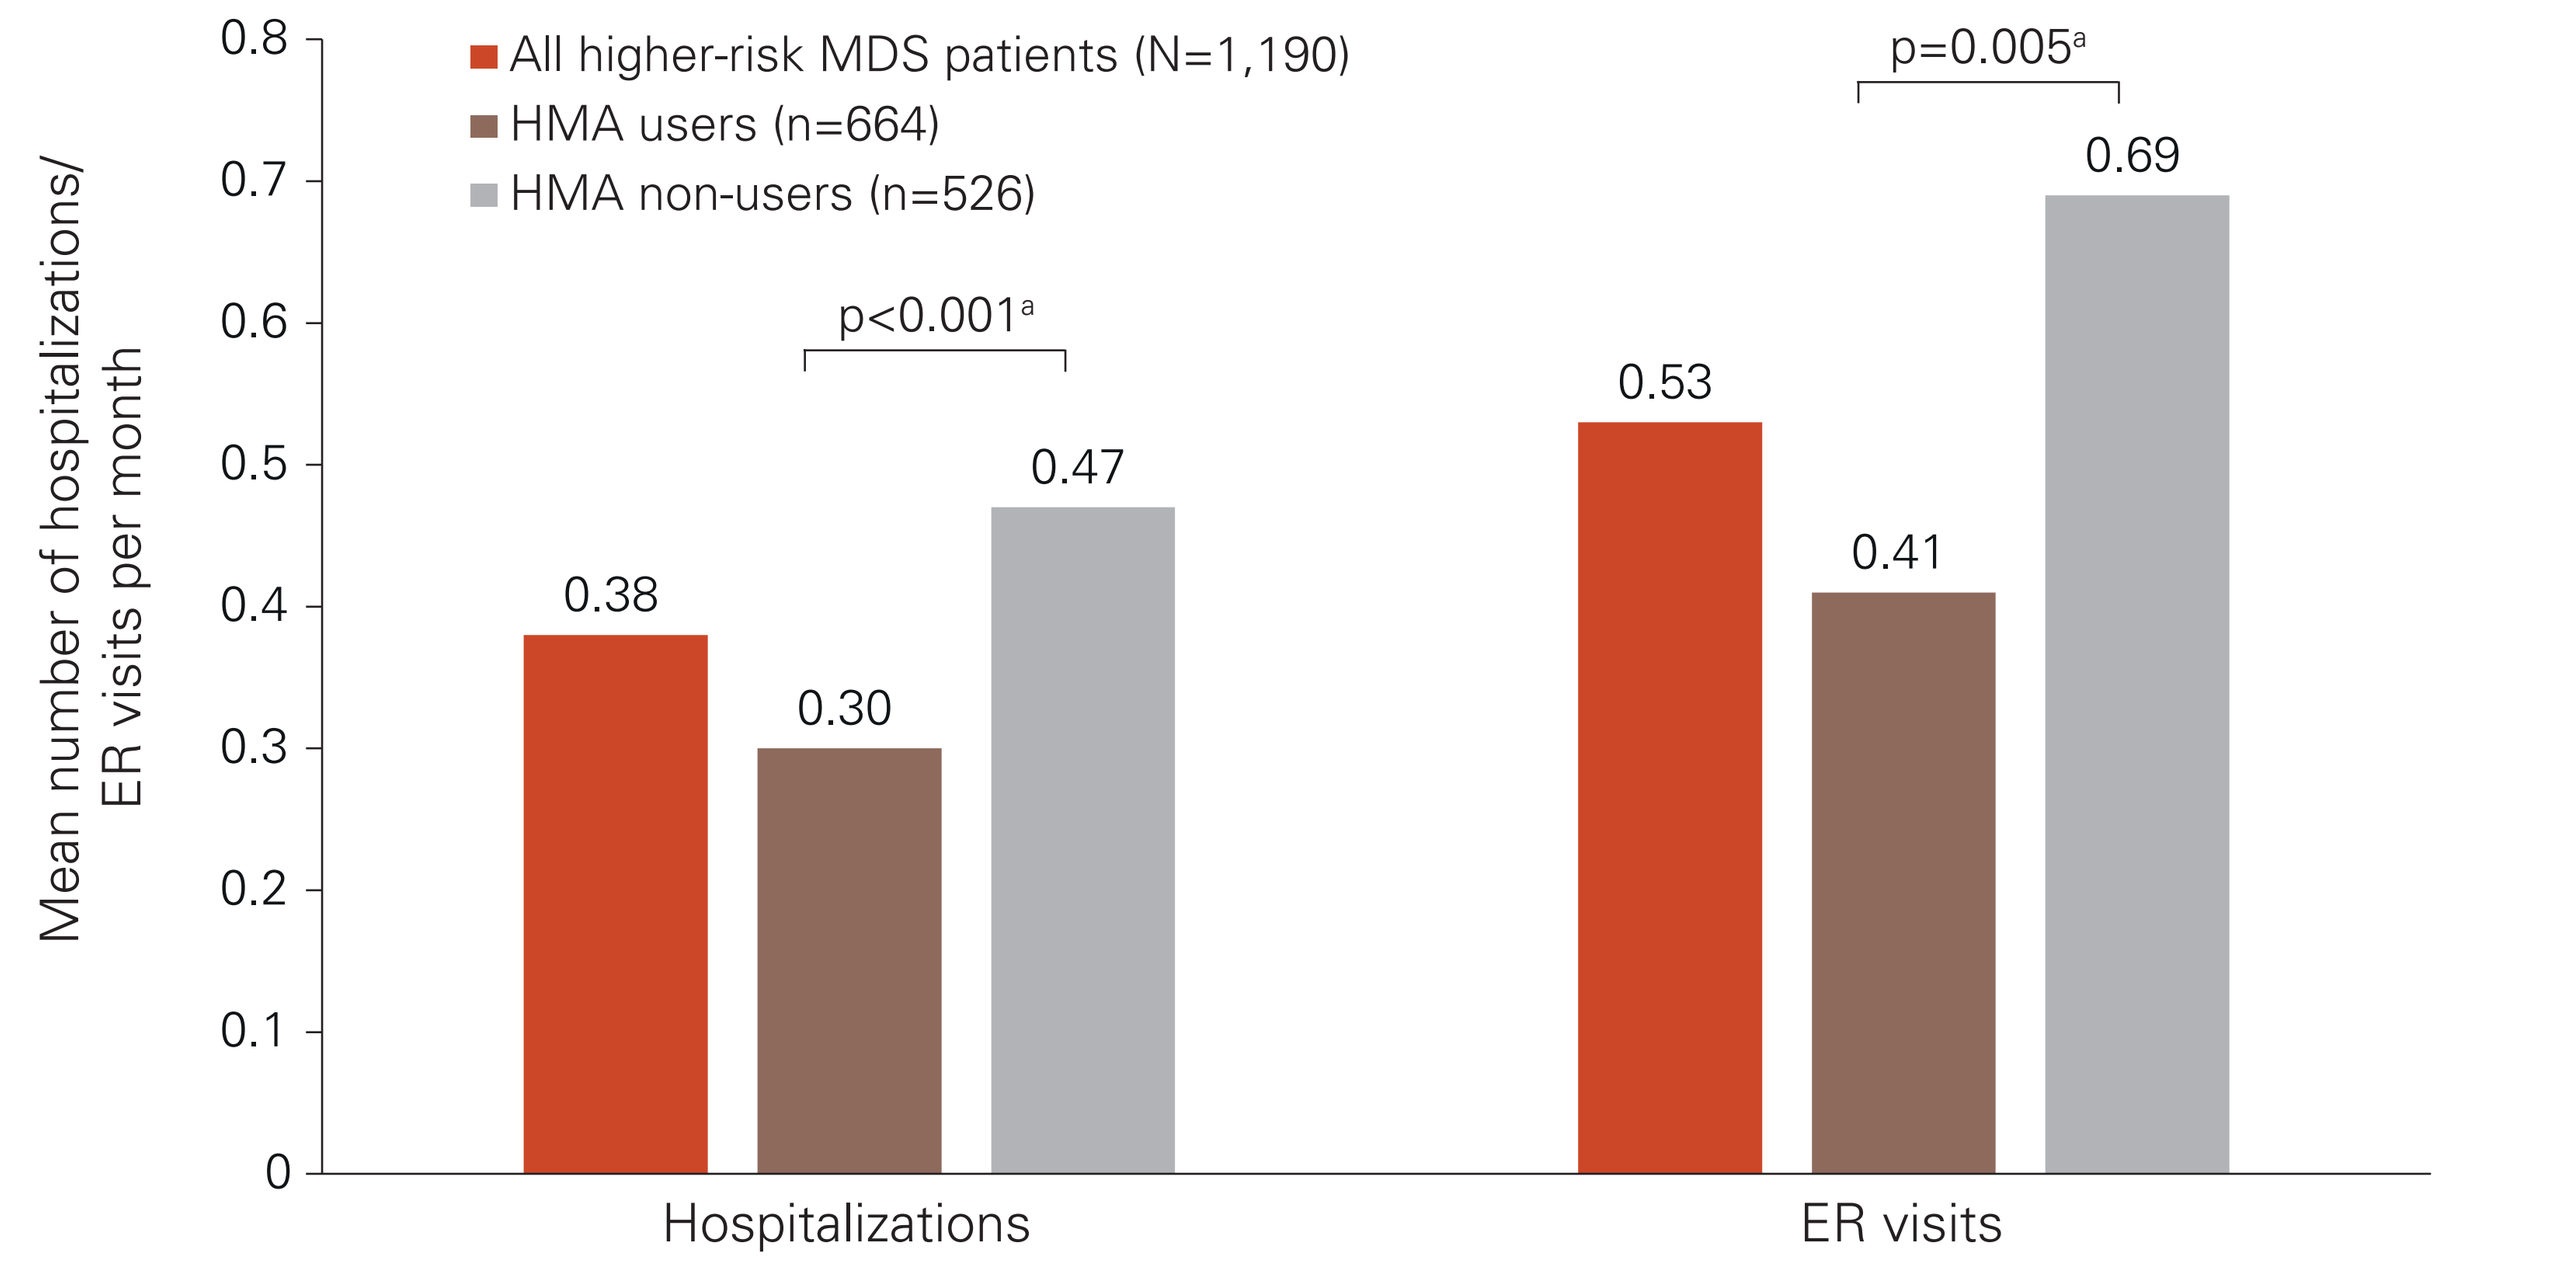

All higher-risk MDS patients (N=1,190)
HMA users (n=664)
HMA non-users (n=526)
Mean number of hospitalizations/ ER visits per month
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
p<0.001[a]
0.38
0.30
0.47
p=0.005[a]
0.53
0.41
0.69
Hospitalizations
ER visits

[a]Wilcoxon rank sum test.
ER, emergency room; HMA, hypomethylating agent; MDS, myelodysplastic syndromes.

Table 1. Baseline Demographic and Clinical Characteristics of Higher-Risk MDS Patients, by HMA Use

Variable	All patients (N=1,190)	HMA non-users (n=526)	HMA users (n=664)	p-value
Age at diagnosis, mean (SD)	79.4 (6.9)	81.4 (7.1)	77.8 (6.34)	<0.001[a]
Male gender, n (%)	715 (60.1)	302 (57.4)	413 (62.2)	0.094[b]
Race / Ethnicity, n (%)				0.457[b]
Non-Hispanic White	1,004 (84.4)	436 (82.9)	568 (85.5)	
Non-Hispanic Black	62 (5.2)	30 (5.7)	32 (4.8)	
Hispanic / Other	124 (10.4)	60 (11.4)	64 (9.6)	
Marital status at diagnosis, n (%)				<0.001[b]
Single / Separated / Divorced / Widowed	440 (37.0)	233 (44.3)	207 (31.2)	
Married	633 (53.2)	239 (45.4)	394 (59.3)	
Unknown	117 (9.8)	54 (10.3)	63 (9.5)	
Census location, n (%)				0.004[b]
West	444 (37.3)	223 (42.4)	221 (33.3)	
South	299 (25.1)	117 (22.2)	182 (27.4)	
Northeast	244 (20.5)	93 (17.7)	151 (22.7)	
Midwest	203 (17.1)	93 (17.7)	110 (16.6)	
NCI comorbidity index, n (%)				<0.001[b]
0–1	631 (53.0)	236 (44.9)	395 (59.5)	
2	139 (11.7)	61 (11.6)	78 (11.7)	
3+	420 (35.3)	229 (43.5)	191 (28.8)	
Poor performance status, n (%)	603 (50.7)	310 (58.9)	293 (44.1)	<0.001[b]

[a]Wilcoxon rank sum test.
[b]Chi-square test.
HMA, hypomethylating agent; MDS, myelodysplastic syndromes; NCI, National Cancer Institute; SD, standard deviation.

Table 2. Results From Zero-Inflated Regression Model for the Number of Hospitalizations or ER Visits Among Higher-Risk MDS Patients

Variable	Level	Hospitalizations IRR (95% CI)	Hospitalizations p-value	ER visits IRR (95% CI)	ER visits p-value
HMA use (ref: No)	Yes	0.728 (0.648–0.819)	<0.0001	0.684 (0.605–0.774)	<0.0001
Age at diagnosis (ref: 80+)	66–70	1.099 (0.906–1.332)	0.3386	0.942 (0.767–1.157)	0.5679
	71–75	1.128 (0.975–1.305)	0.1066	1.004 (0.860–1.171)	0.9636
	76–80	1.114 (0.968–1.282)	0.1312	1.038 (0.896–1.203)	0.6157
Gender (ref: Female)	Male	0.991 (0.880–1.115)	0.8805	0.910 (0.802–1.031)	0.1390
Race / Ethnicity (ref: Non-Hispanic White)	Hispanic / Other	1.105 (0.915–1.336)	0.3003	1.009 (0.830–1.228)	0.9254
	Non-Hispanic Black	1.125 (0.900–1.406)	0.3001	1.226 (0.969–1.551)	0.0901
Marital status at diagnosis (ref: Married)	Single / Separated / Divorced / Widowed	1.034 (0.911–1.174)	0.6065	1.061 (0.928–1.213)	0.3876
	Unknown	0.957 (0.793–1.154)	0.6458	1.056 (0.861–1.294)	0.6029
Census location (ref: West)	Midwest	1.155 (0.975–1.368)	0.0958	0.988 (0.827–1.180)	0.8943
	Northeast	1.032 (0.885–1.204)	0.6843	0.891 (0.757–1.049)	0.1645
	South	1.077 (0.924–1.254)	0.3428	1.009 (0.861–1.182)	0.9148
Urban location (ref: Rural)	Urban	1.302 (1.069–1.586)	0.0087	1.322 (1.075–1.626)	0.0082
NCI comorbidity index (ref: 0–1)	2	1.110 (0.936–1.317)	0.2313	1.237 (1.030–1.486)	0.0227
	3+	1.208 (1.061–1.374)	0.0042	1.482 (1.290–1.702)	<0.0001
Poor performance status (ref: No)	Yes	1.149 (1.021–1.293)	0.0208	1.123 (0.989–1.275)	0.0725

CI, confidence interval; ER, emergency room; HMA, hypomethylating agent; IRR, incidence rate ratio; MDS, myelodysplastic syndromes; NCI, National Cancer Institute; ref, reference value.

Hospitalizations and ER visits among HMA users and non-users

- HMA non-users had more hospitalizations (mean 0.47 vs 0.30; p<0.001) and ER visits (mean 0.69 vs 0.41, p=0.005) per month than HMA users within 1 year after diagnosis (**Figure 3**).
- The rate of hospitalization per month was 27% lower (incidence rate ratio [IRR]: 0.728, 95% confidence interval [CI]: 0.648–0.819; p<0.0001), and the rate of ER visits was 32% lower (IRR: 0.684, 95% CI: 0.605–0.774; p<0.0001) for HMA users than non-users, as calculated by the zero-inflated regression model (**Table 2**).
- The rates of hospitalization per month and ER visits were significantly higher for patients from an urban location and patients with higher comorbidity burden (**Table 2**).

Limitations

- The SEER-Medicare linked database has limitations, including a lack of data on some healthcare services (eg, long-term care), a lack of some clinical data (eg, specific blood cell counts and cytogenetics needed to calculate risk score), and the potential for inaccurate representation of conditions/procedures from claims data.
- In addition, generalizability of the study findings is limited to higher-risk MDS patients aged >65 years who are not enrolled in HMO plans.

Conclusions

- This study found substantial underutilization of HMA therapy, which was associated with considerable economic impact, as indicated by increased rates of hospitalizations and ER visits among higher-risk MDS patients.
- To our knowledge, this is the first study to specifically report the impact of non-use of HMA therapy on healthcare resource utilization among higher-risk MDS patients.
- Reasons for the underutilization of HMA therapy in clinical practice require further investigation.

References

1. Bewersdorf JP, et al. *Ther Adv Hematol.* 2020;11: doi: 10.1177/2040620720955006. **2.** Celgene Corporation. VIDAZA (azacitidine) prescribing information. March 2020. Accessed July 1, 2021. URL: https://packageinserts.bms.com/pi/pi_vidaza.pdf. **3.** Otsuka America Pharmaceutical, Inc. DACOGEN® (decitabine) prescribing information. December 2018. Accessed July 1, 2021. URL: https://www.accessdata.fda.gov/drugsatfda_docs/label/2010/021790s006lbl.pdf. **4.** Otsuka Pharmaceutical Co., Ltd. INQOVI® (decitabine and cedazuridine) prescribing information. July 2020. Accessed July 1, 2021. URL: https://www.accessdata.fda.gov/drugsatfda_docs/label/2020/212576s000lbl.pdf. **5.** Silverman LR, et al. *J Clin Oncol.* 2002;20:2429-40. **6.** Kantarjian H, et al. *Cancer.* 2006;106:1794-803. **7.** Fenaux P, et al. *Lancet Oncol.* 2009;10:223-32. **8.** Davidoff AJ, et al. *Leuk Lymphoma.* 2020;61:1178-87. **9.** Ma X, et al. *BMJ Open.* 2018;8:e019955. **10.** Corman S, et al. *Clin Lymphoma Myeloma Leuk.* 2021;21:e206-11. **11.** National Cancer Institute. Surveillance Epidemiology, and End Results (SEER) Medicare Database. Accessed July 1, 2021. URL: https://healthcaredelivery.cancer.gov/seermedicare/overview/.

Disclosures

This study was funded by Taiho Oncology, Inc.

AM Zeidan has participated in advisory boards, and/or had a consultancy with and received honoraria from AbbVie, Otsuka, Pfizer, Celgene/BMS, Jazz, Incyte, Agios, Boehringer Ingelheim, Novartis, Acceleron, Astellas, Daiichi Sankyo, Cardinal Health, Taiho Oncology, Seattle Genetics, BeyondSpring, Trovagene, Takeda, Ionis, Amgen, Janssen, Epizyme, Syndax, and Tyme; has received research funding (institutional) from Celgene/BMS, AbbVie, Astex, Pfizer, MedImmune/ AstraZeneca, Boehringer Ingelheim, Trovagene, Incyte, Takeda, Novartis, Aprea, and ADC Therapeutics; served on steering and independent data review committees for clinical trials for Novartis, AbbVie, and Janssen; and received travel support for meetings from Pfizer, Novartis, and Trovagene.

N Joshi, H Kale, W-J Wang, and S Corman are employees of OPEN Health, paid consultants to Taiho Oncology, Inc. in connection with this study.

K Hill and T Salimi are employees of Taiho Oncology, Inc. K Hill is a stockholder of Johnson & Johnson and Celldex Therapeutics.

RS Epstein is an employee of Epstein Health LLC; holds leadership positions in Fate Therapeutics, Illumina, and Veracyte; and has received consulting fees from Merck, Radius Health, Halozyme, Intra-Cellular Therapies, Taiho Oncology, and Otsuka.

Acknowledgments

Medical writing support was provided by Ann Cameron of Curo, a division of Envision Pharma Group, and funded by Taiho Oncology, Inc.

Corresponding author:

Amer M. Zeidan (amer.zeidan@yale.edu).

Please scan this Quick Response (QR) code with your smartphone app to view an electronic version of this poster. If you don't have a smartphone, access the poster via the internet at: https://bit.ly/3zJgn8X

Copies of this e-Poster obtained through QR code are for personal use only and may not be reproduced without written permission from the authors of this poster.