Retrospective Analysis of the Relationship between Transfusion Independence and Bone Marrow Fibrosis Reduction in Patients with Myelofibrosis Treated with Pacritinib versus Ruxolitinib

Stephen T Oh,[1] Jamila Shammo,[2] Vikas Gupta,[3] Mary Frances McMullin,[4] Prithviraj Bose,[5] Ruben Mesa,[6] Alessandro Lucchesi,[7] Sarah Buckley,[8] Purvi Suthar,[8] Karisse Roman-Torres,[8] John Mascarenhas,[9] Francisca Ferrer-Marín[10]

[1]Washington University School of Medicine, St. Louis, MO; [2]Northwestern University Feinberg School of Medicine, Chicago, IL; [3]Princess Margaret Cancer Centre, University Health Network, Toronto, ON, Canada; [4]Queen's University Belfast, Northern Ireland, United Kingdom; [5]The University of Texas MD Anderson Cancer Center, Houston, TX; [6]Atrium Health Wake Forest Baptist Comprehensive Cancer Center; Winston-Salem, NC; [7]Hematology Unit, IRCCS Istituto Romagnolo per lo Studio dei Tumori (IRST) "Dino Amadori", Meldola, Italy; [8]CTI BioPharma Corp., a Sobi company, Seattle, WA; [9]Tisch Cancer Institute, Icahn School of Medicine at Mount Sinai, New York, NY; Hematology Department, [10]Hospital Morales Meseguer, Centro de Investigación Biomédica en Red de Enfermedades Raras, Universidad Católica San Antonio de Murcia, Murcia, Spain

CONCLUSIONS

- In cytopenic myelofibrosis (MF) patients from PERSIST-2, transfusion independence response (TI-response) for patients in the pacritinib (PAC) group was associated with bone marrow fibrosis improvement.
- Though these results are based on a small sample size, they contrast with recent data suggesting no correlation between bone marrow fibrosis reduction and achieved TI-response on the JAK1/2 inhibitors momelotinib and ruxolitinib (RUX).[1]
- Further studies are warranted to confirm the relationship between bone marrow fibrosis reduction and anemia benefit in patients treated with PAC.

INTRODUCTION

- MF is a life-limiting malignancy characterized by marrow fibrosis, splenomegaly, and progressive cytopenias.
- PAC is a JAK1-sparing inhibitor of JAK2/IRAK1/ACVR1[2,3] that in addition to improving spleen volume is associated with anemia benefit in MF patients.
 - PAC is a potent inhibitor of ACVR1, which regulates hepcidin production, and may thus ameliorate anemia of inflammation that occurs in myelofibrosis.
- Recent *in vivo* studies have shown that dual JAK2/IRAK1 inhibition is associated with improvement in both cytopenias and bone marrow fibrosis in an inflammation-driven murine MF model.[4,5]

AIM

- To retrospectively analyze the relationship between achieving transfusion independence (TI) and reduction in bone marrow fibrosis in MF patients treated with PAC 200 mg twice daily (BID) vs RUX as best available therapy (BAT) on the phase 3 PERSIST-2 study.

METHODS

- This analysis focused on patients who received PAC 200 mg BID and RUX (**Figure 1**).
- TI was assessed using Gale criteria.
 - To be considered TI, they did not receive any red blood cell transfusions over any 12-week period.
- Treatment comparisons (PAC vs. RUX) for efficacy outcomes, TI response and ≥50% reduction in transfusion burden were performed using the Fisher's exact test.
- The proportion of patients with bone marrow fibrosis reduction (≥1 grade decrease in reticulin fibrosis from baseline at week 24) was reported among patients on PAC achieving TI-R vs. non-response (NR).

Figure 1. PERSIST-2 Analysis of Transfusion Independence

Retrospective Analysis Group (PERSIST-2 study, N=311)
- Not TI at baseline
- Enrolled ≥12 weeks prior to study termination
- Platelet counts ≤100 x 10^9/L

Pacritinib 200 mg BID (n=41)

BAT (n=43) including RUX (n=18)

Objective:
- Determine the percentage of patients who became TI over any 12-week interval through week 24 via Gale criteria

Transfusion independence (Gale criteria): no RBC transfusion over 12 weeks

BAT=best available therapy; BID=twice daily; JAK=Janus associated kinase; TI=transfusion independent.

RESULTS

- Baseline characteristics were similar between the PAC and RUX groups, including median platelet count (41 vs 38 x 10^9/L) and median hemoglobin (8.7 vs 8.6 g/dL; **Table 1**)
- All patients required RBC transfusion at baseline

Table 1. Patient Characteristics

Baseline characteristics	PAC 200 mg BID N= 41	RUX N=18
Age, median	67	70
DIPSS high risk, n (%)	18 (44)	8 (44)
PLT count (x10^9/L), median	41	38
Hemoglobin (g/dL), median	8.7	8.6
Median RBC transfusions* over past 90 days, n	1.51	2.49
Prior JAK2 inhibitor, n (%)	23 (56)	14 (78)
Spleen volume (cm^3), median	2225.9	2451.6
Palpable spleen length (cm), median	15.00	13.50

*Packs transfused
DIPSS, Dynamic International Prognostic Scoring System; PLT, platelet; RBC, red blood count.

Pacritinib maintains full dose intensity

- The analysis included 41 patients on PAC (median dose intensity 100% through week 24) and 18 on RUX (median daily dose 10 mg at week 24, **Figure 2**).

Figure 2. Dose intensity of patients on PAC and RUX

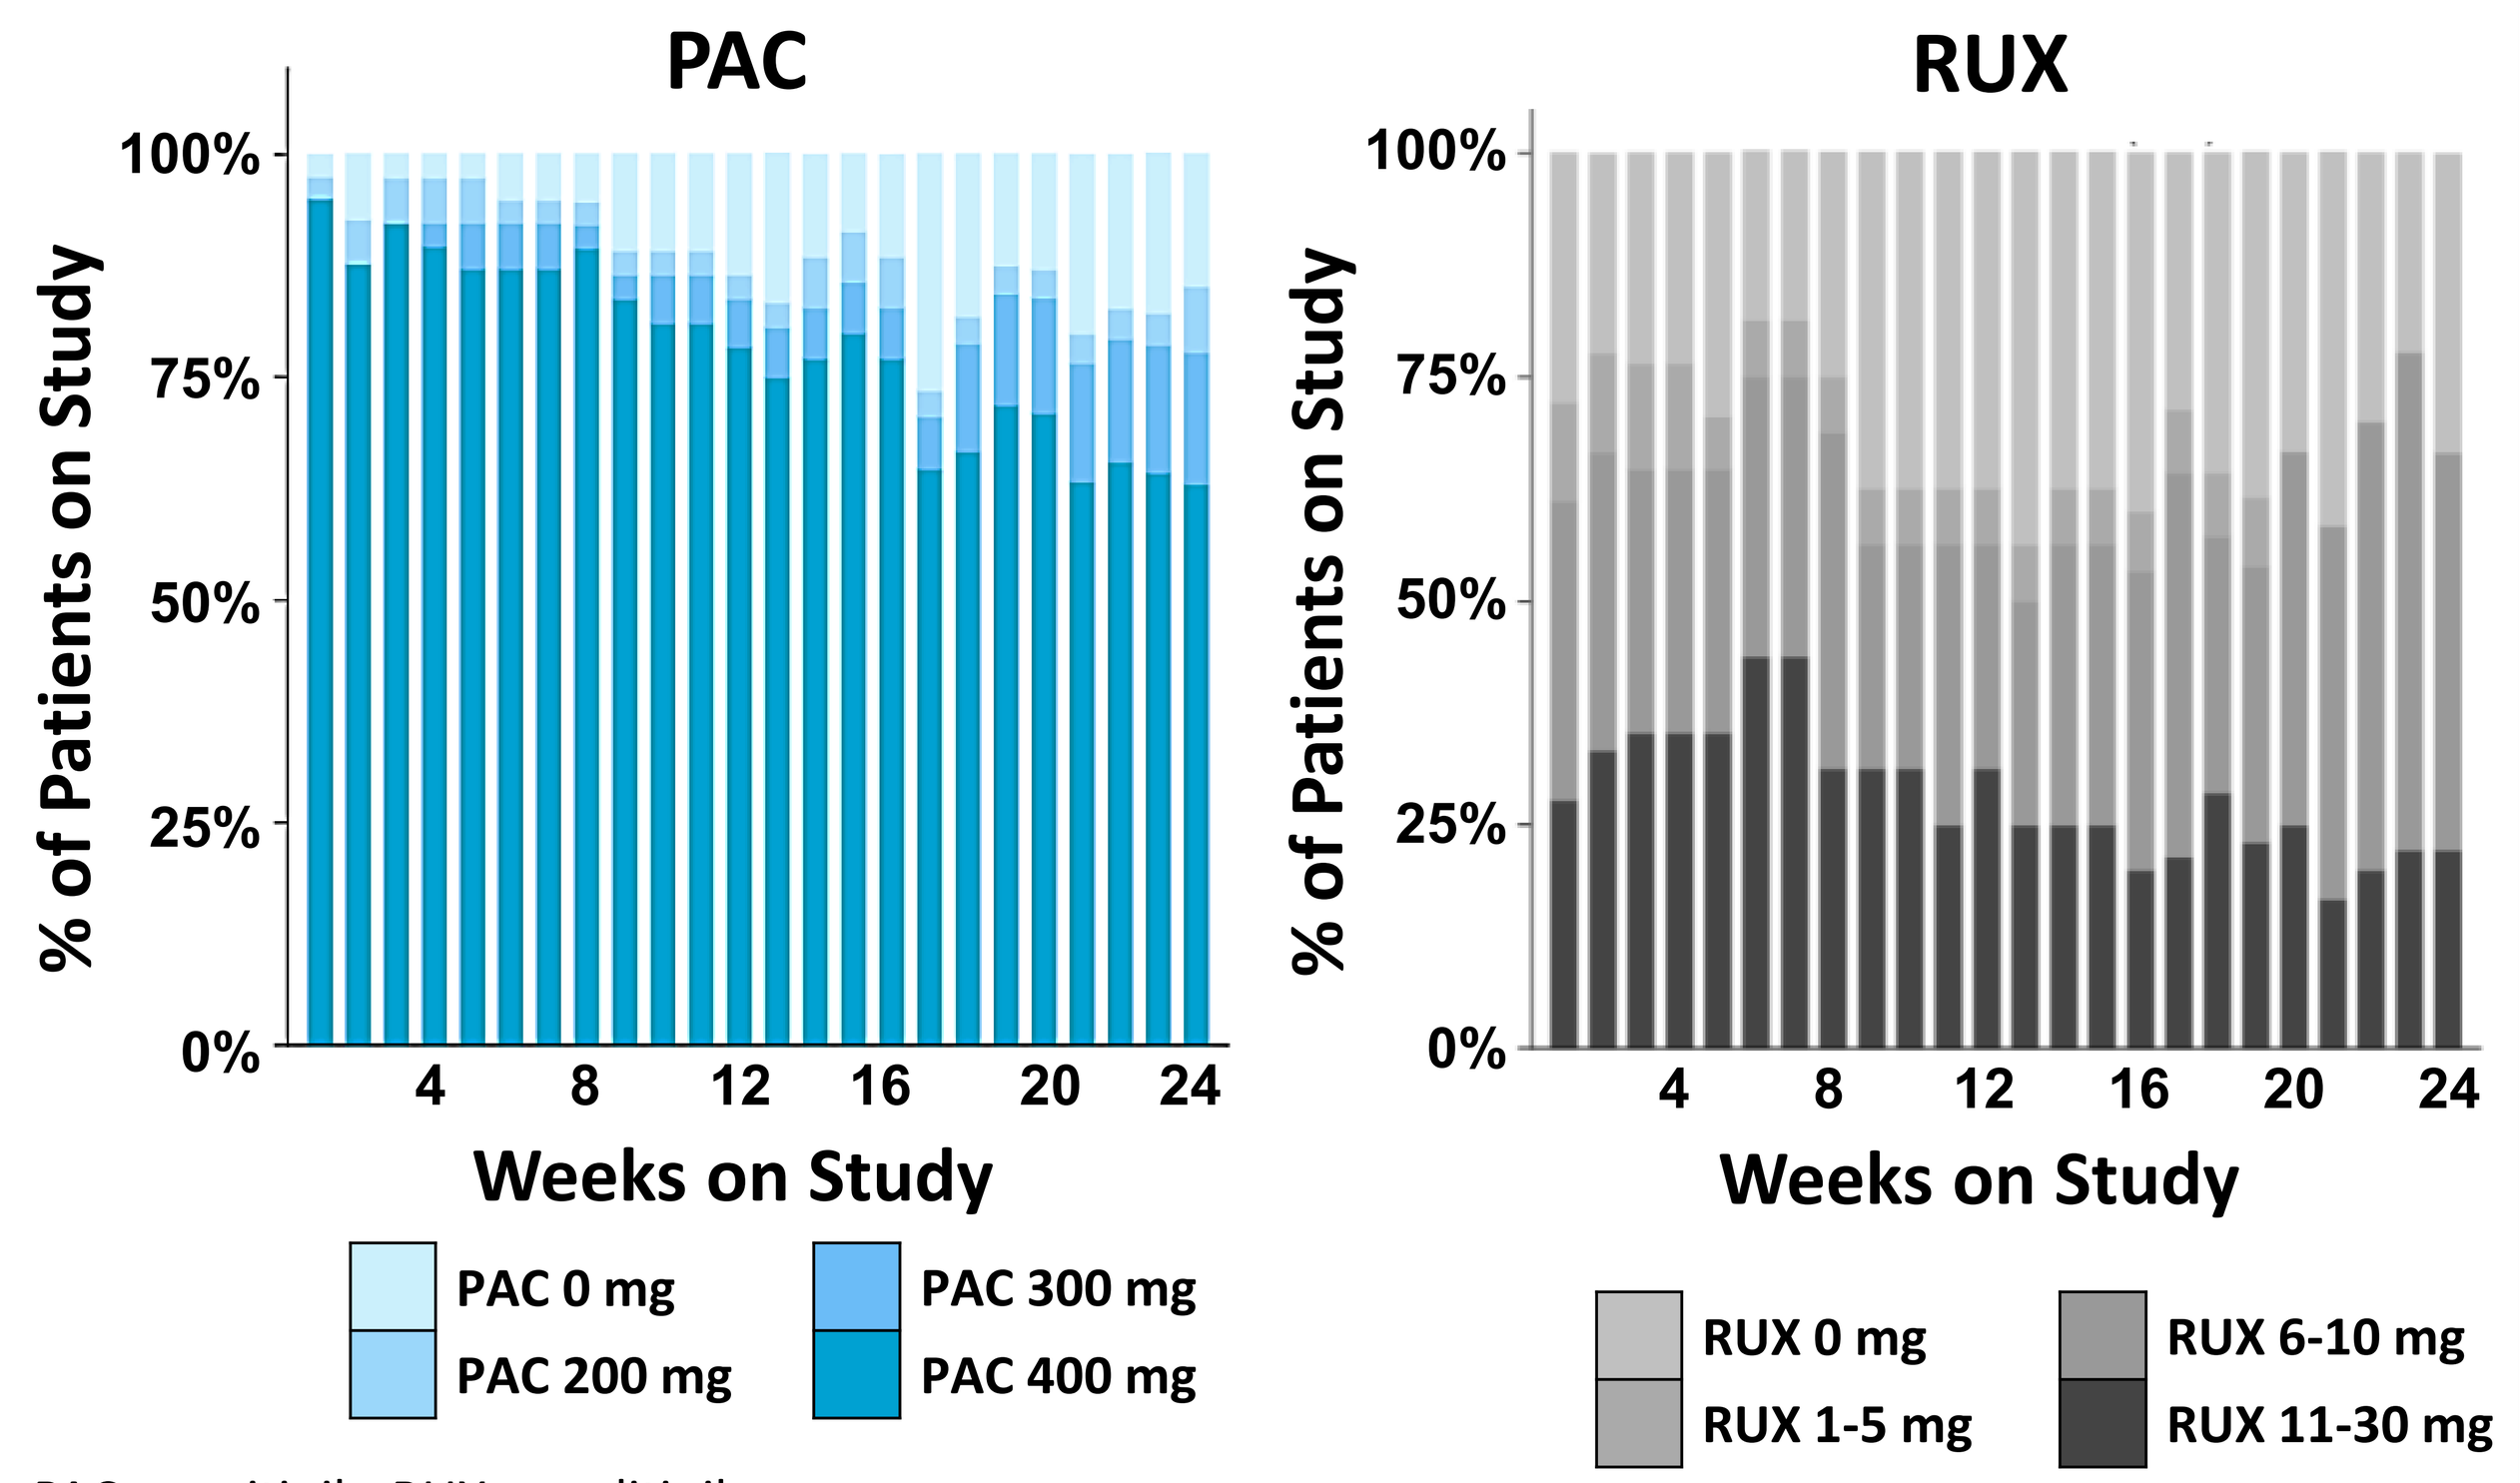
PAC
RUX
100%
75%
50%
25%
0%
% of Patients on Study
4 8 12 16 20 24
Weeks on Study
PAC 0 mg
PAC 200 mg
PAC 300 mg
PAC 400 mg
RUX 0 mg
RUX 1-5 mg
RUX 6-10 mg
RUX 11-30 mg

PAC, pacritinib; RUX, ruxolitinib.

Efficacy outcomes

- In this subgroup of patients (PAC=41 and RUX=18), higher percentage of patients in the PAC arm achieved spleen volume reduction ≥35% than those in RUX arm (19% vs 0%; *P*=0.0025).
- Total symptom score ≥50% improvement was experienced by a higher percentage of patients on PAC than those on RUX (28% vs. 11%; *P*=0.0111).
- Patient global impression change response of "much improved" and "very much improved" was higher for the patients in the PAC arm than the RUX arm (30% vs. 6%; *P*=0.0057).

Greater proportion of patients on pacritinib achieve TI

- A significantly greater proportion of patients treated with PAC vs RUX achieved TI through week 24: 37% (n=15/41) vs 6% (n=1/18), *P*=0.023.
 - Nominally, this trend held for those with baseline platelets <50 x 10^9/L: 28% vs 8%, *P*=0.222 (**Figure 3**).

Figure 3. Rate of transfusion independence over 12 weeks

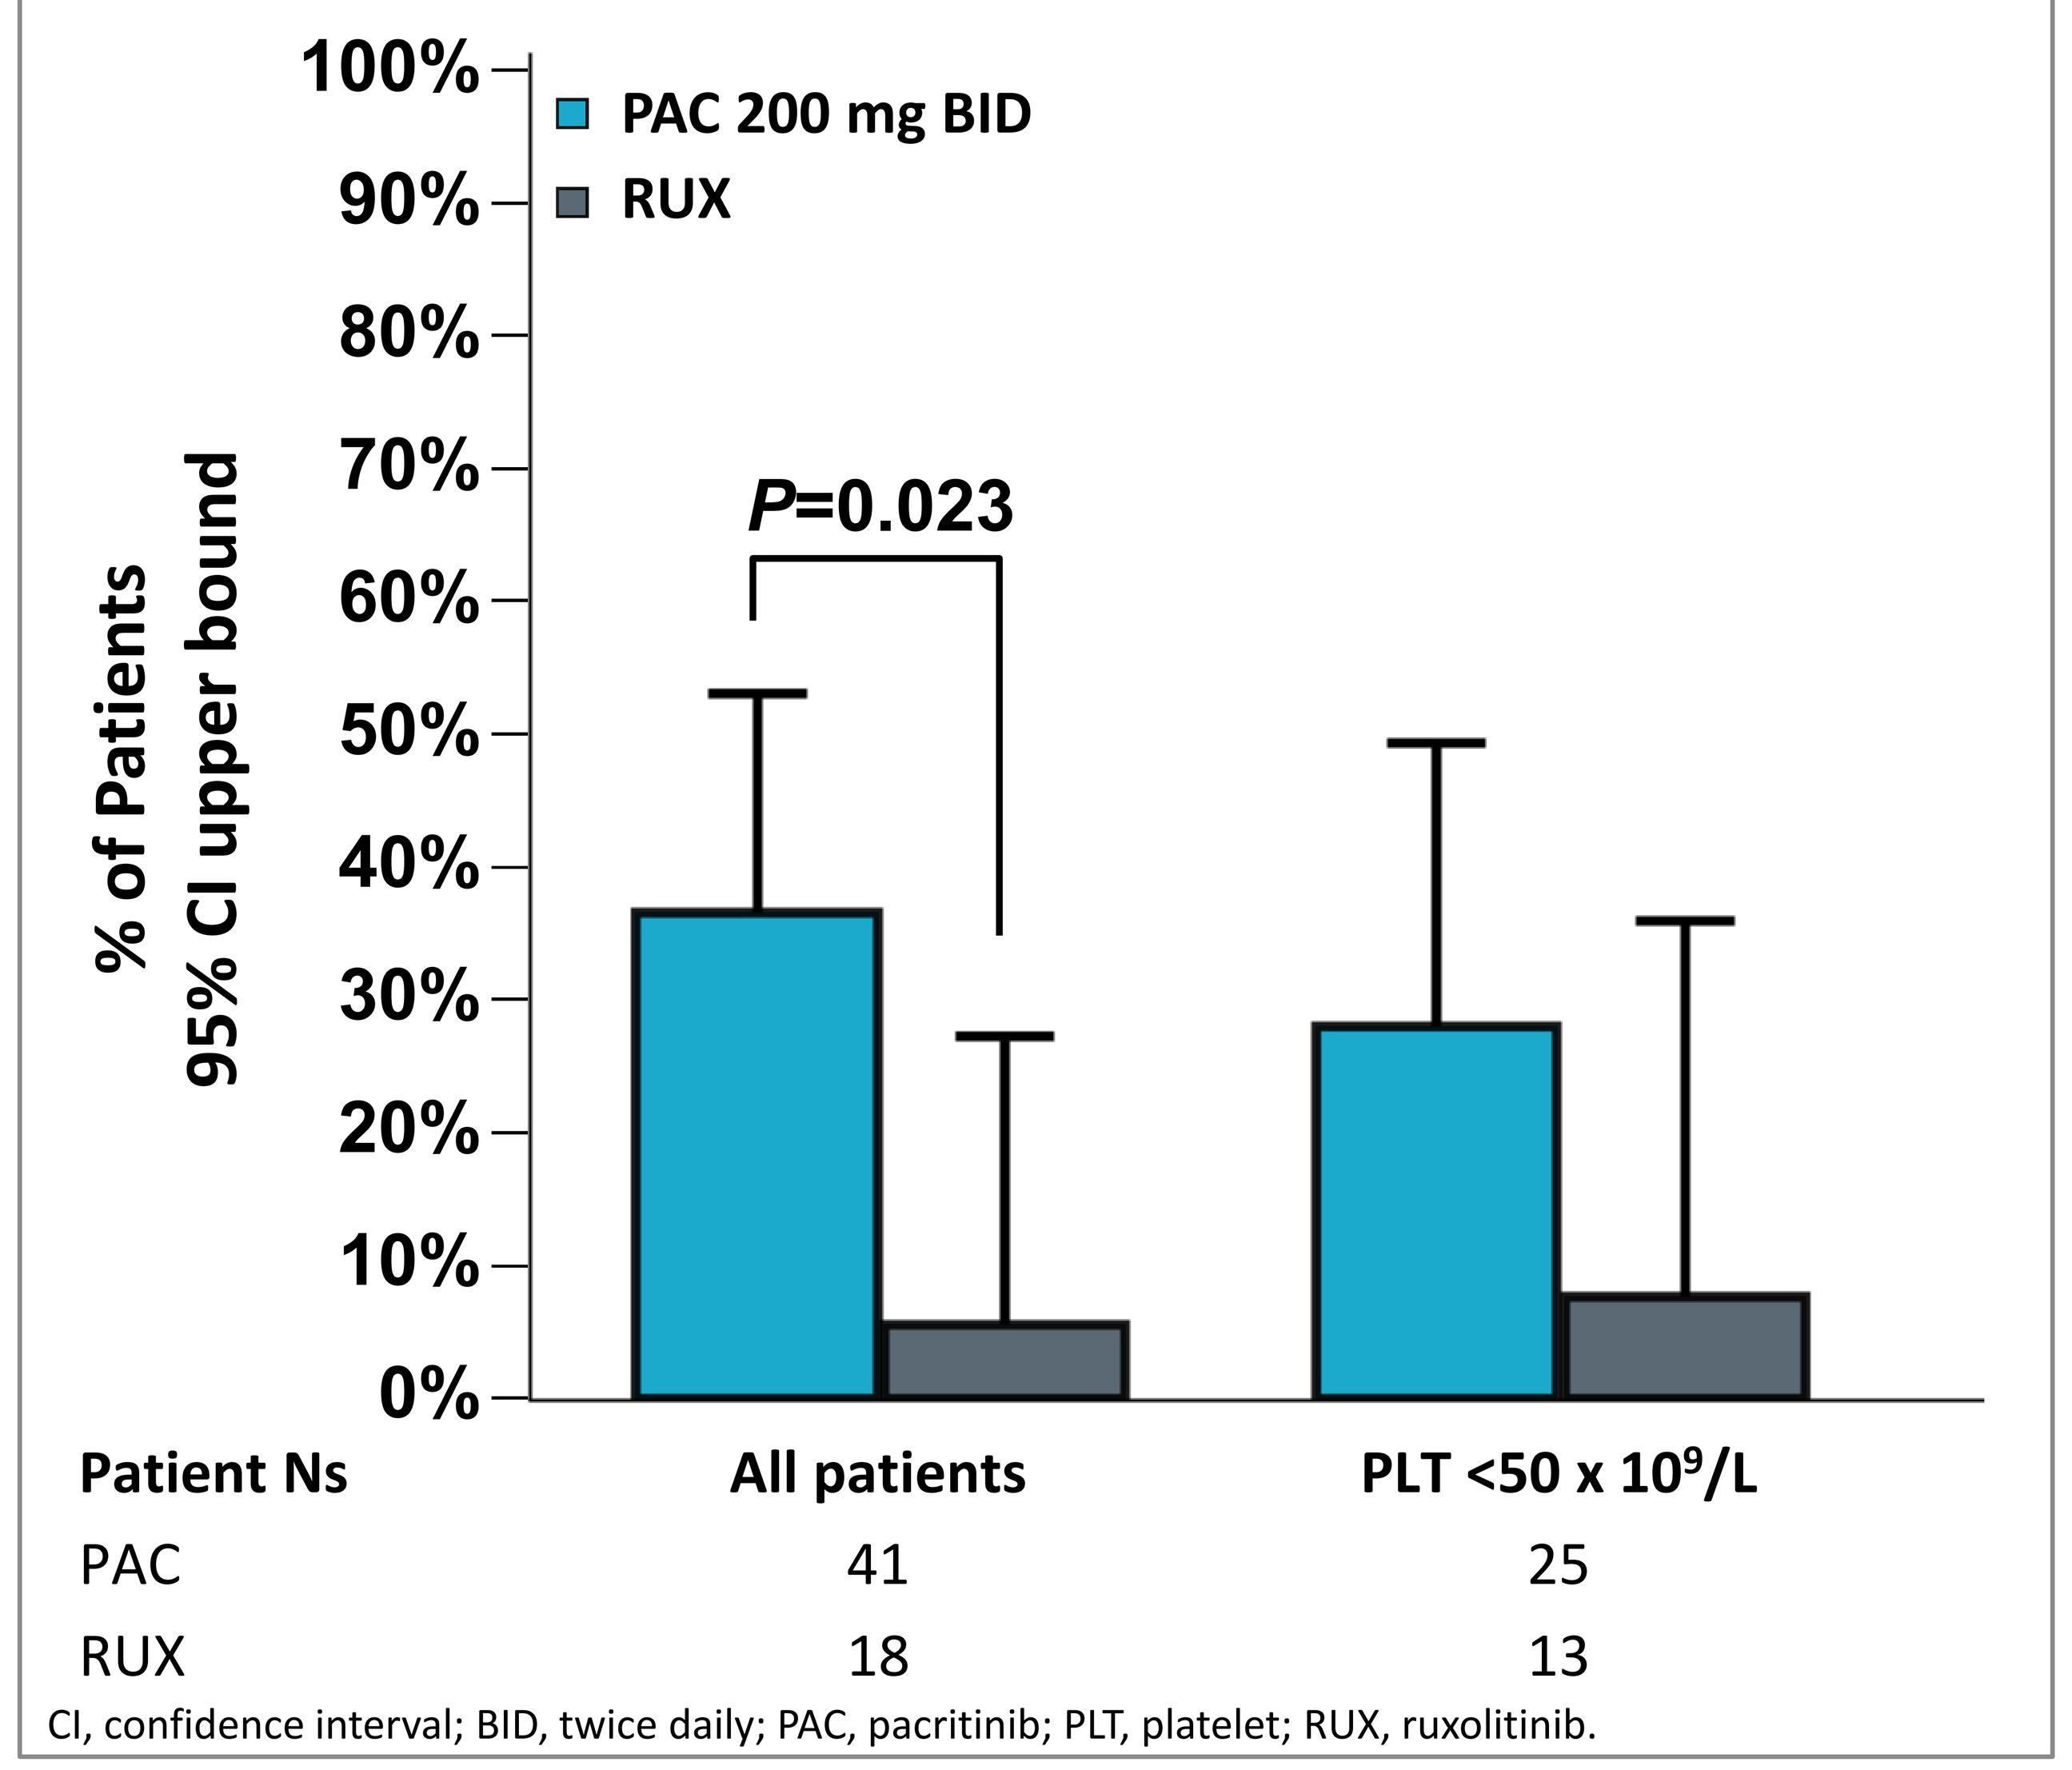
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
% of Patients
95% CI upper bound
PAC 200 mg BID
RUX
P=0.023
Patient Ns
All patients
PLT <50 x 10⁹/L
PAC 41 25
RUX 18 13

CI, confidence interval; BID, twice daily; PAC, pacritinib; PLT, platelet; RUX, ruxolitinib.

- A greater percentage achieved a ≥50% reduction in RBC transfusions over any 12 weeks for all patients (49% vs 6%, *P*=0.001) and those with baseline platelets <50 x 10^9/L (40% vs 8%, *P*=0.06; **Figure 4**).

Figure 4. Rate of ≥50% transfusion reduction over 12 weeks

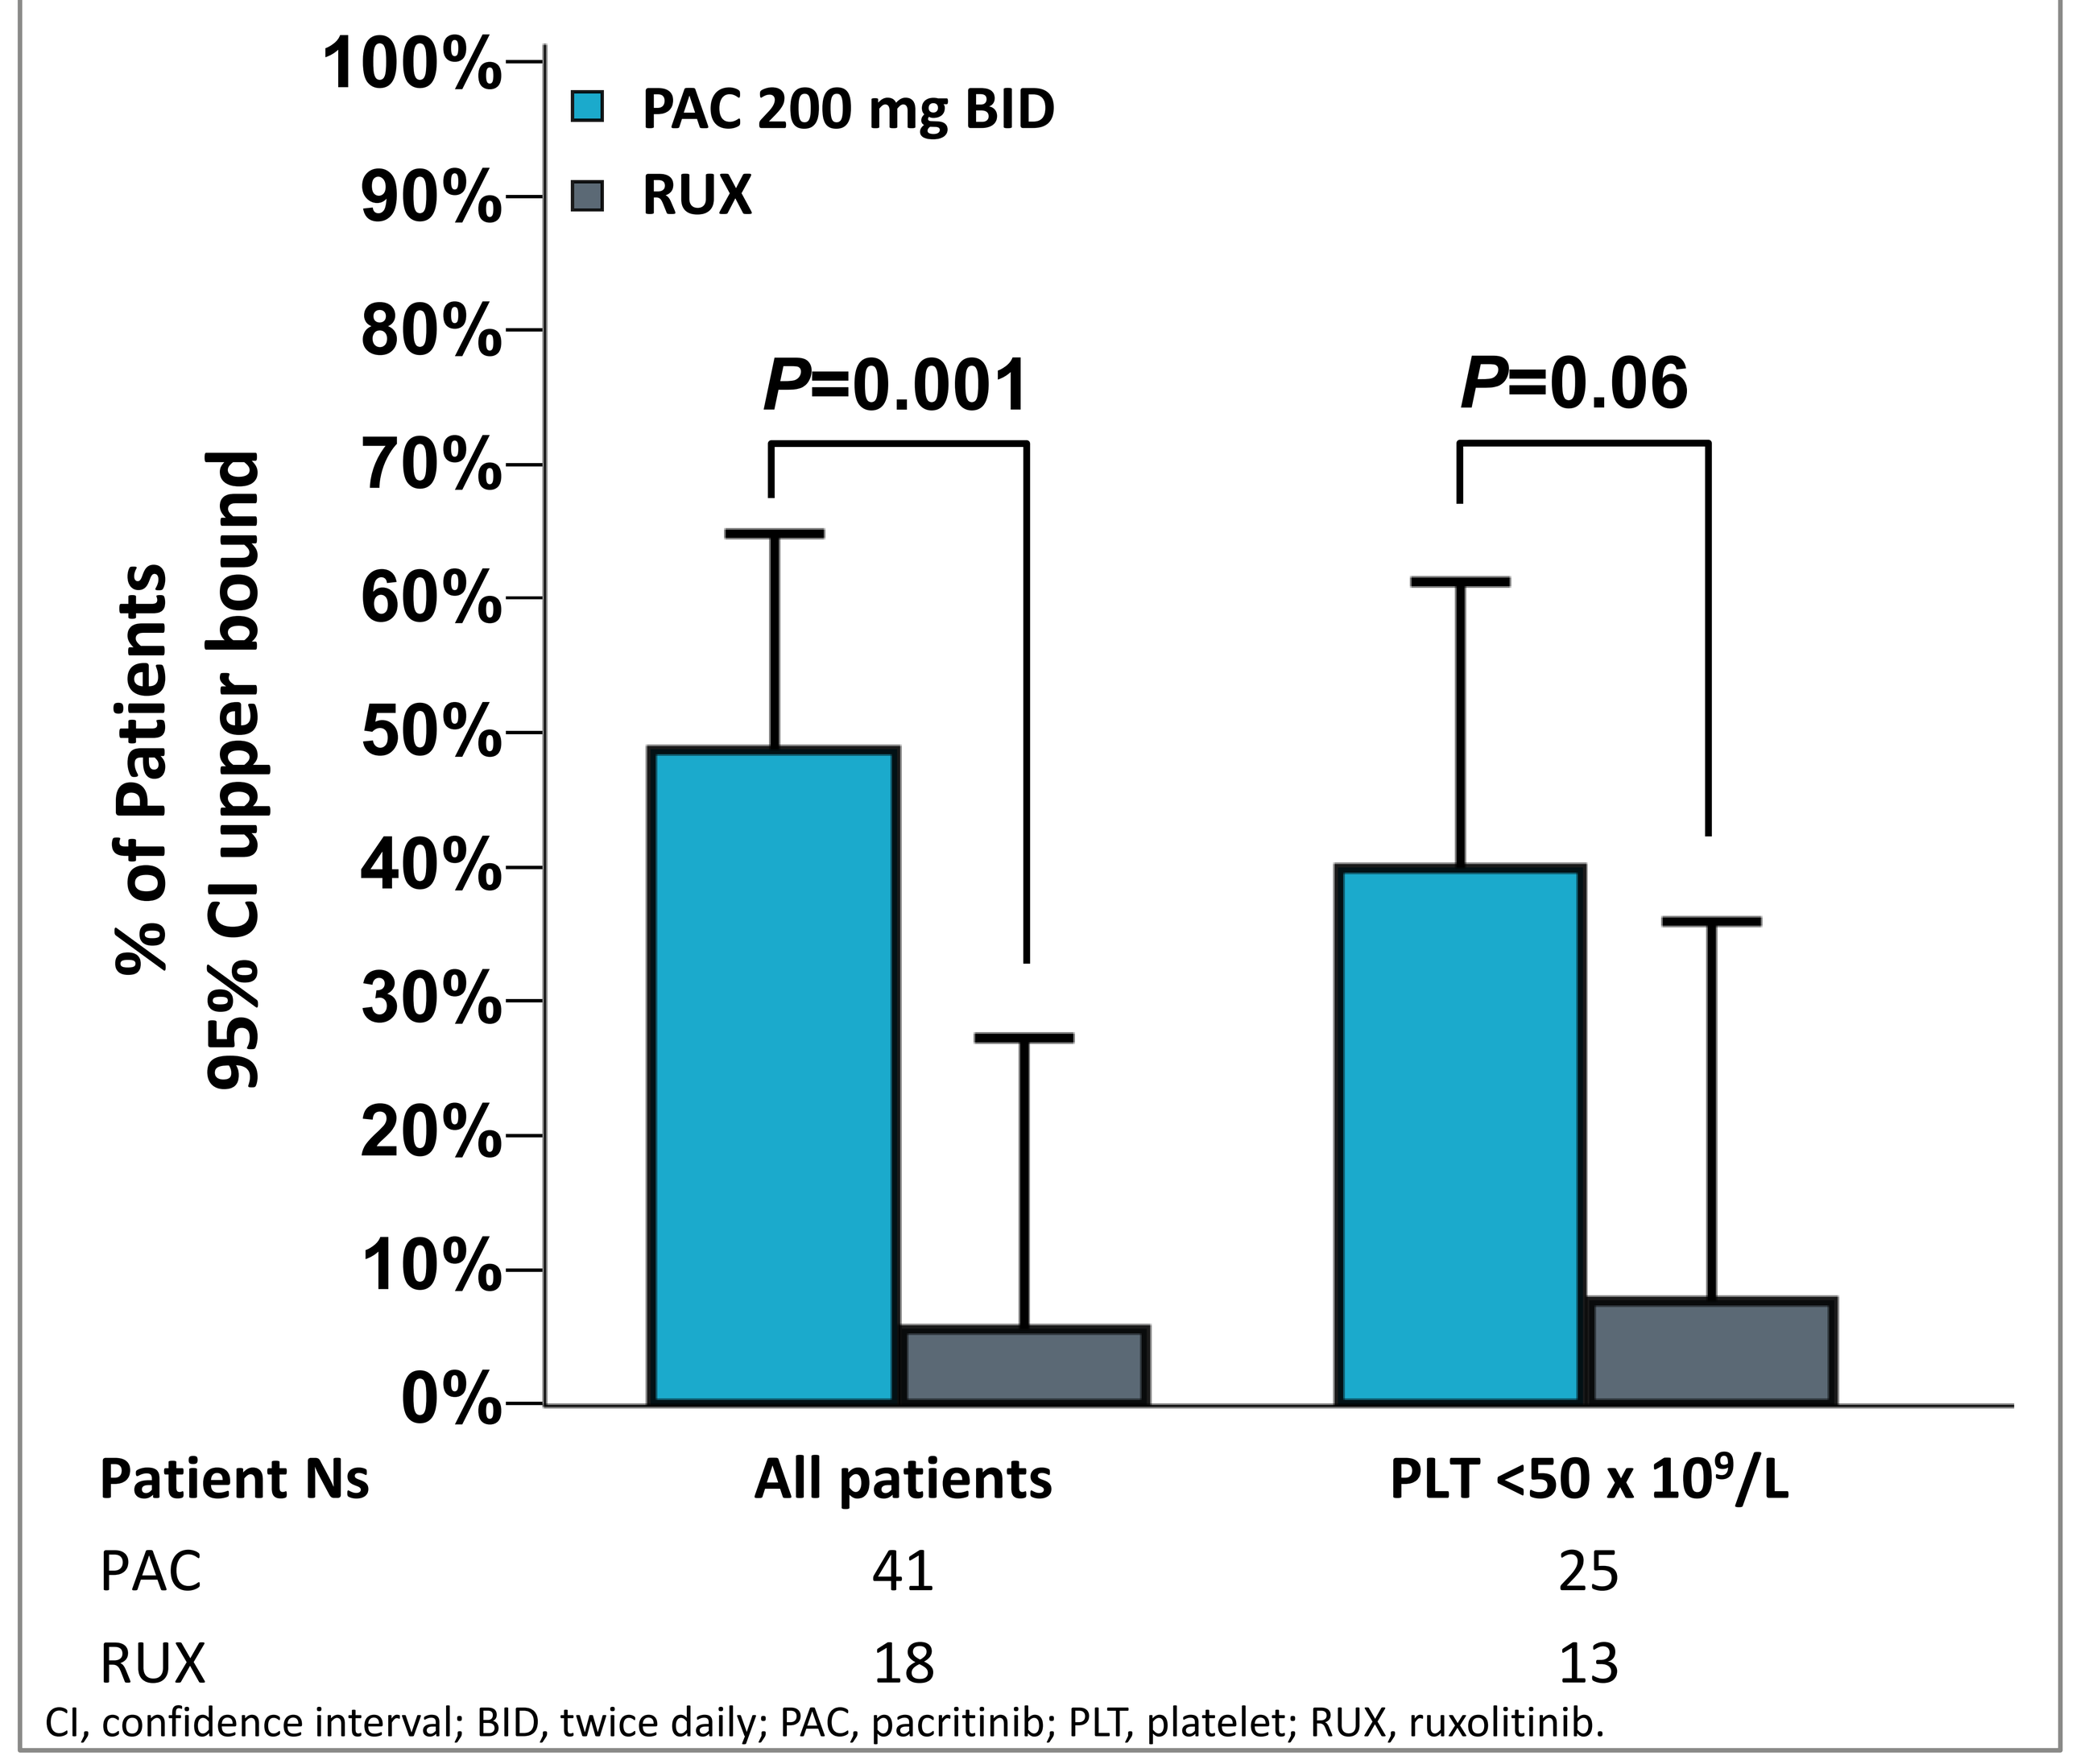
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
% of Patients
95% CI upper bound
PAC 200 mg BID
RUX
P=0.001
P=0.06
Patient Ns
All patients
PLT <50 x 10⁹/L
PAC 41 25
RUX 18 13

CI, confidence interval; BID, twice daily; PAC, pacritinib; PLT, platelet; RUX, ruxolitinib.

Transfusion response associated with bone marrow fibrosis improvement

- Paired bone marrow assessments at baseline and week 24 were available for 18/41 of patients on PAC, of whom 44% (8/18) achieved TI-response on study.

Figure 5. Bone marrow fibrosis in TI-responders vs non-responders PERSIST-2, PAC 200 mg BID

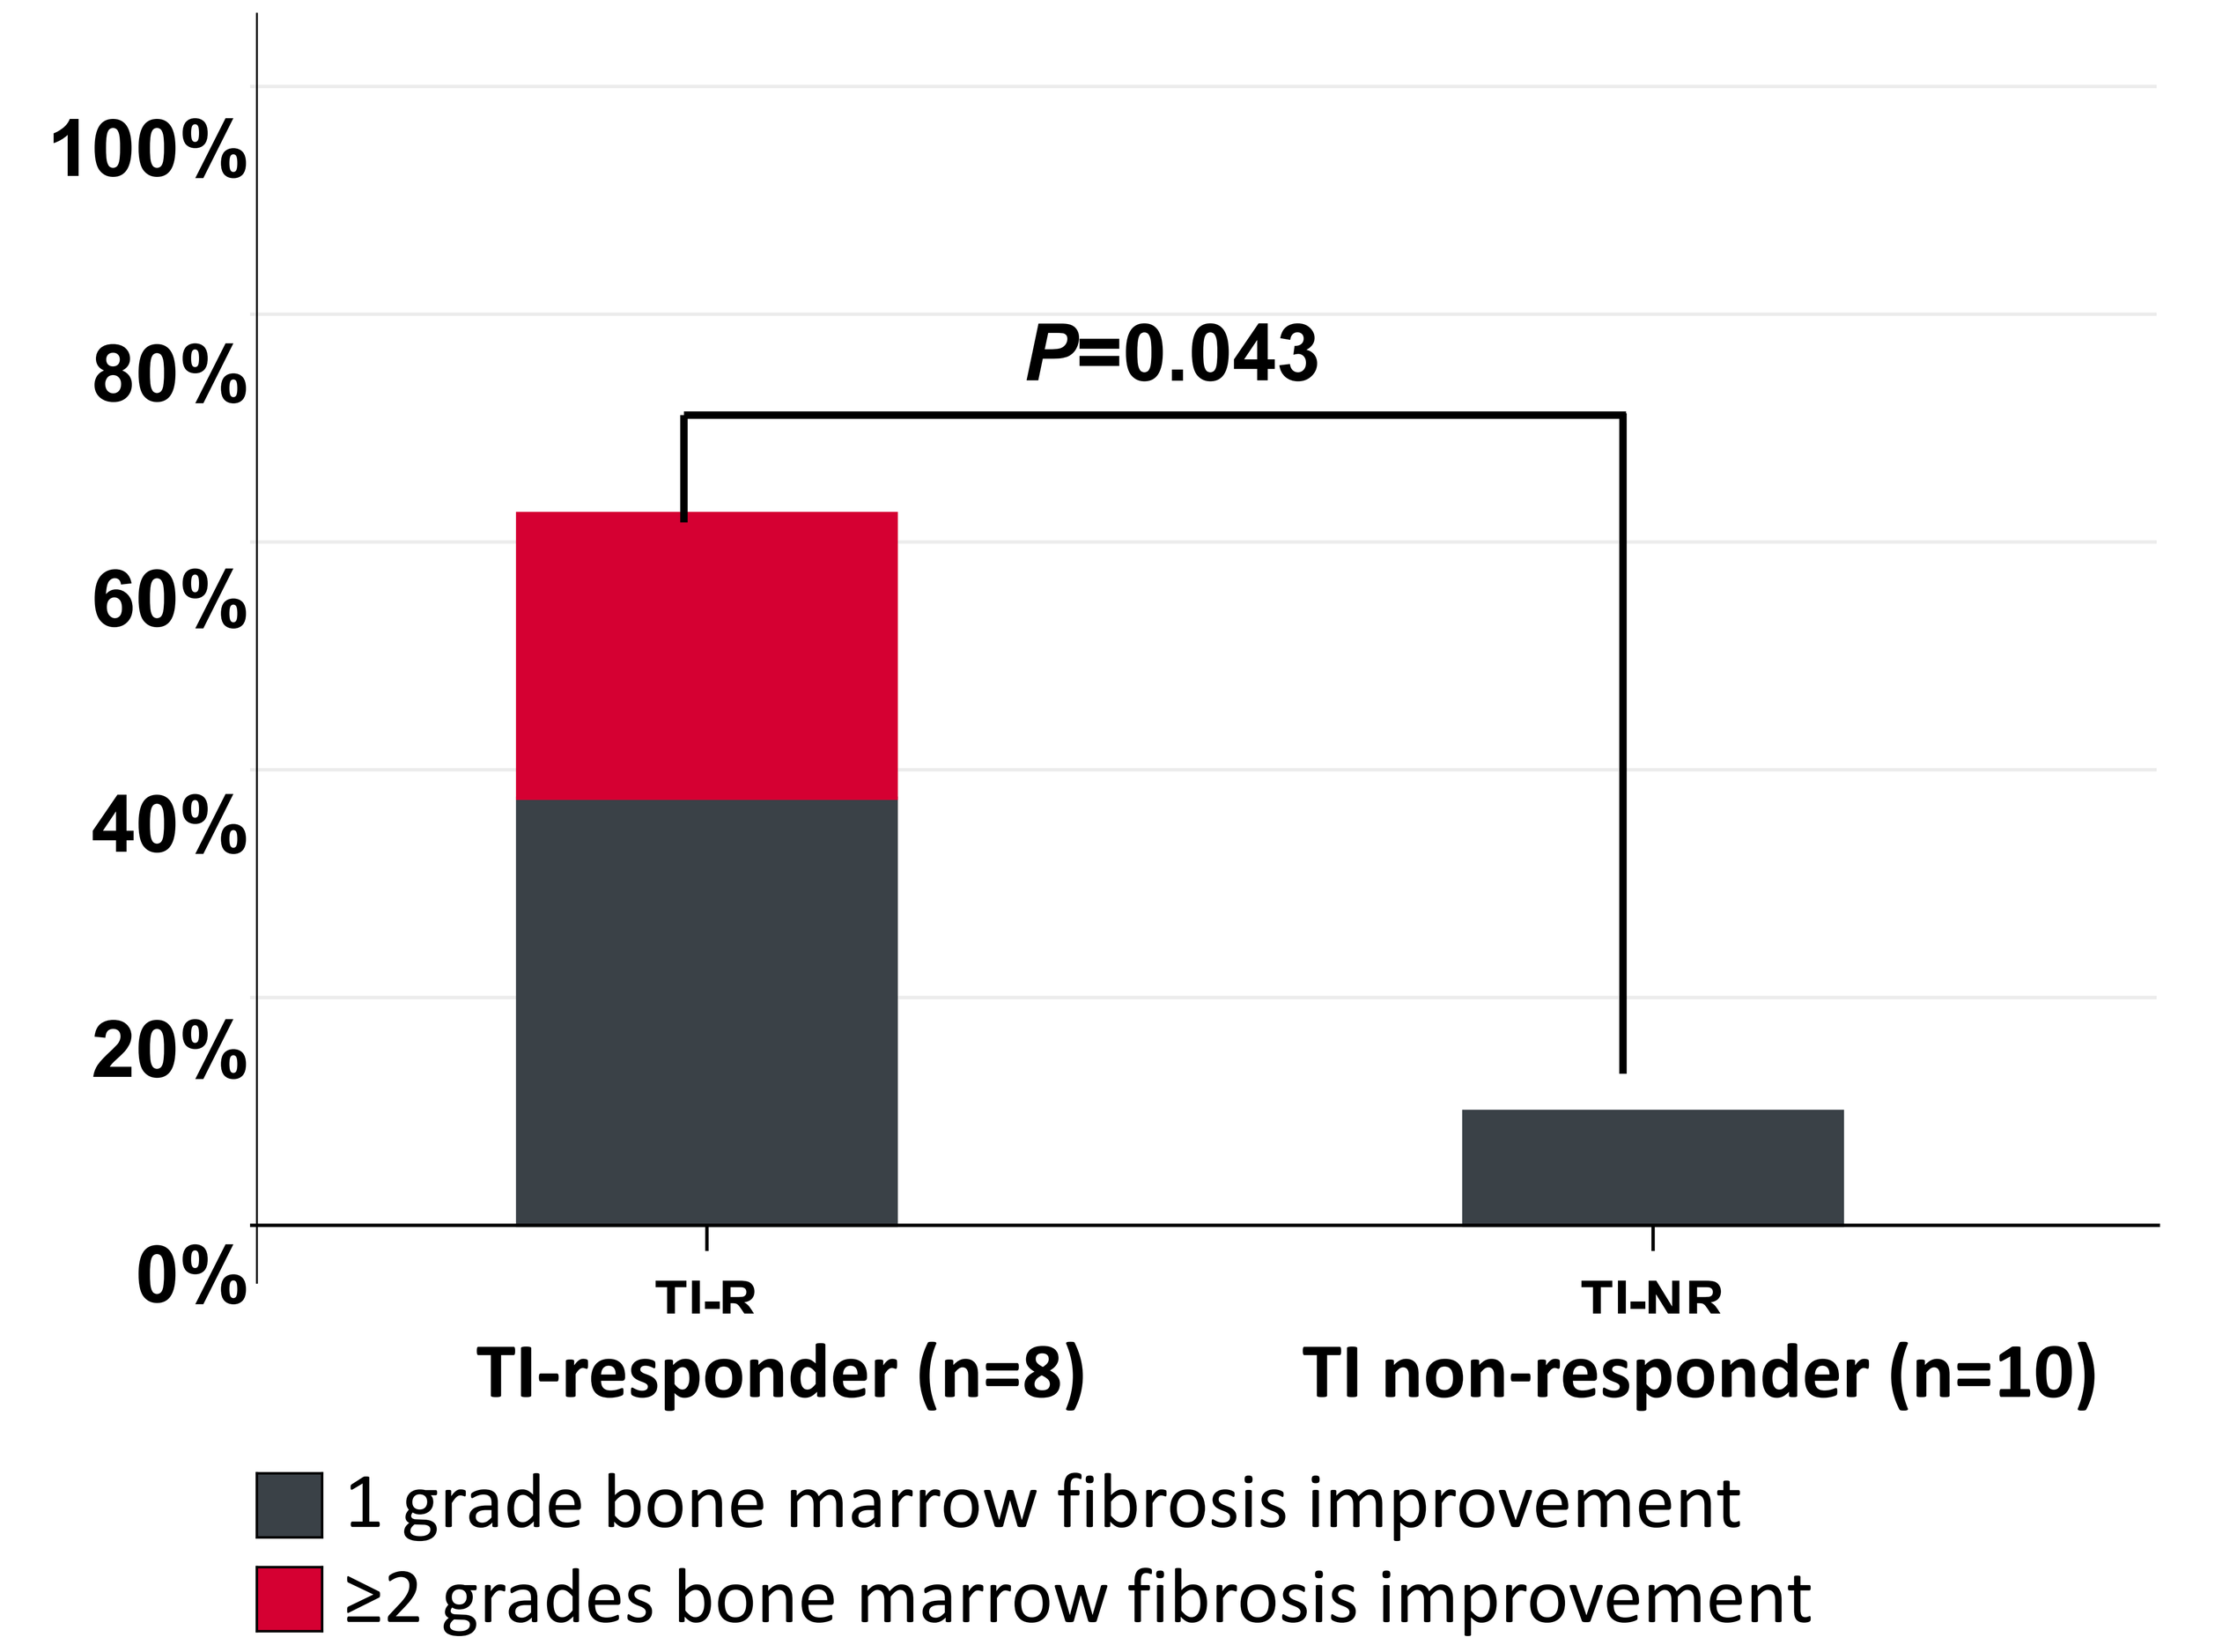
100%
80%
60%
40%
20%
0%
P=0.043
TI-R
TI-NR
TI-responder (n=8)
TI non-responder (n=10)
1 grade bone marrow fibrosis improvement
≥2 grades bone marrow fibrosis improvement

BID, twice daily; PAC, pacritinib; TI, transfusion independence.

- The proportion of patients on PAC who experienced bone marrow fibrosis reduction (≥1 grade) was significantly greater among TI-responders (62.5%, n=5/8) compared to TI-non-responders (10%, n=1/10) (*P*=0.043; **Figure 5**).
- Of the 5 patients who achieved TI-response and bone marrow fibrosis reduction, all had grade 2-3 fibrosis at baseline, and 2 experienced a reduction from grade 3 to grade 1.
- By contrast, paired bone marrow biopsies were available for 5 patients on RUX (2 patients had baseline bone marrow fibrosis stage 3, 2 patients had stage 2, and 1 patient had stage 1), and there was no association between fibrosis reduction in TI-responders (0%, n=0/1) and TI-non-responders (25%, n=1/4).

ACKNOWLEDGEMENTS: This study is supported by CTI BioPharma Corp., a Sobi company. This poster was previously presented at the American Society of Hematology Annual Meeting, December 9-13, 2023; San Diego, California.

REFERENCES: **1.** Oh et al. *Blood* 2022;140 (Supp 1):821-23. **2.** Singer et al. *J Exp Pharmacol*. 2016;8:11-19. **3.** Oh, et al. *Blood Adv.* 2023;7:5835–5842. **4.** Zamora et al. *HemaSphere*. 2023 Aug;7(Suppl):e08349b7. **5.** Zamora et al. *HemaSphere*. 2023; Aug 7(Suppl) :e3621965.

Disclosures: STO has declared relationships with CTI BioPharma Corp., a Sobi company, Bristol Myers Squibb, Disc Medicine, Blueprint Medicines, PharmaEssentia, Constellation/MorphoSys, Geron, AbbVie, Sierra Oncology/GSK, Cogent, Incyte, Morphic, Protagonist. **JS** has declared relationships with Alexion, Astra Zeneca, BMS, CTI BioPharma Corp., a Sobi company, Incyte, Novartis, Sanofi Aventis, MJH, Apellis, NS Bio, GSK, Protagonist. **VG** has declared relationships with Novartis, BMS Celgene, Sierra Oncology, AbbVie, Constellation, Pfizer, GSK, Roche, and CTI BioPharma Corp., a Sobi company. **MFMcM** has declared relationships with Novartis, BMS, AbbVie, AOP, Incyte, CTI BioPharma Corp., a Sobi company, and GSK. **PB** has declared relationships with Blueprint, BMS, Cogent, CTI BioPharma Corp., a Sobi company, Incyte, MorphSys, GSK, AbbVie, Karyopharm, PharmaEssentia, Novartis, DISC Medicine, Ionis, Janssen, Kartos, Sumitomo, Telios, Geron, Morphic, Jubliant. **RM** has declared relationships with Novartis, Sierra Oncology, LaJolla Pharma, Constellation, Celgene, Incyte, AbbVie, Samus, Genetech, Promedior, CTI BioPharma Corp., a Sobi company, Mays Cancer Center, and NCI. **AL** has declared relationships with MorphoSys, Amgen, Grifols, Novartis, Sanofi, Protagonist, Sobi, Pfizer, Incyte, and BMS. **SB**, **PS**, and **KR-T** are employees of CTI BioPharma Corp., a Sobi company. **JM** has declared relationships with AbbVie, BMS, Celgene, CTI BioPharma Corp., a Sobi company, Constellation, Galacto, Geron, GSK, Imago, Incyte, Kartos, Karyopharm, MorphoSys, Novartis, Prelude Therapeutics, Pfizer, PharmaEssentia, Roche, and Sierra Oncology. **FF-M** has declared relationships with CTI BioPharma Corp., a Sobi company, Incyte, Novartis, BMS, and Celgene.

2024 NCODA International Spring Forum ● April 3-5, 2024 ● Dallas, Texas