SEVABERTINIB

CLINICAL DRUG–DRUG INTERACTION STUDIES INVESTIGATING THE PHARMACOKINETICS AND SAFETY OF SEVABERTINIB

Alissa Uddin, PharmD[1], on behalf of Barbara J. Brennan, PharmD[1], Michaela Damaske[2], Frank-Thorsten Hafner, PhD[3], Robert Fricke, PhD[2], Isabel Piel, PhD[3], Clemens-Jeremias von Buehler, PhD[3], Michael Niehues, PhD[2,3], Natalia Jungmann, PhD[3], Stefanie Reif, PhD[2], Sue Prendergast, MSc[4], Philip Lienau, PhD[2], Bart Ploeger, PhD[2], Chunlin Chen, MD, PhD[1,a]

[1]Bayer HealthCare Pharmaceuticals, Inc., Whippany, New Jersey, USA; [2]Bayer AG, Berlin, Germany; [3]Bayer AG, Wuppertal, Germany; [4]Bayer plc, Reading, UK

[a]Affiliation at the time the study was performed

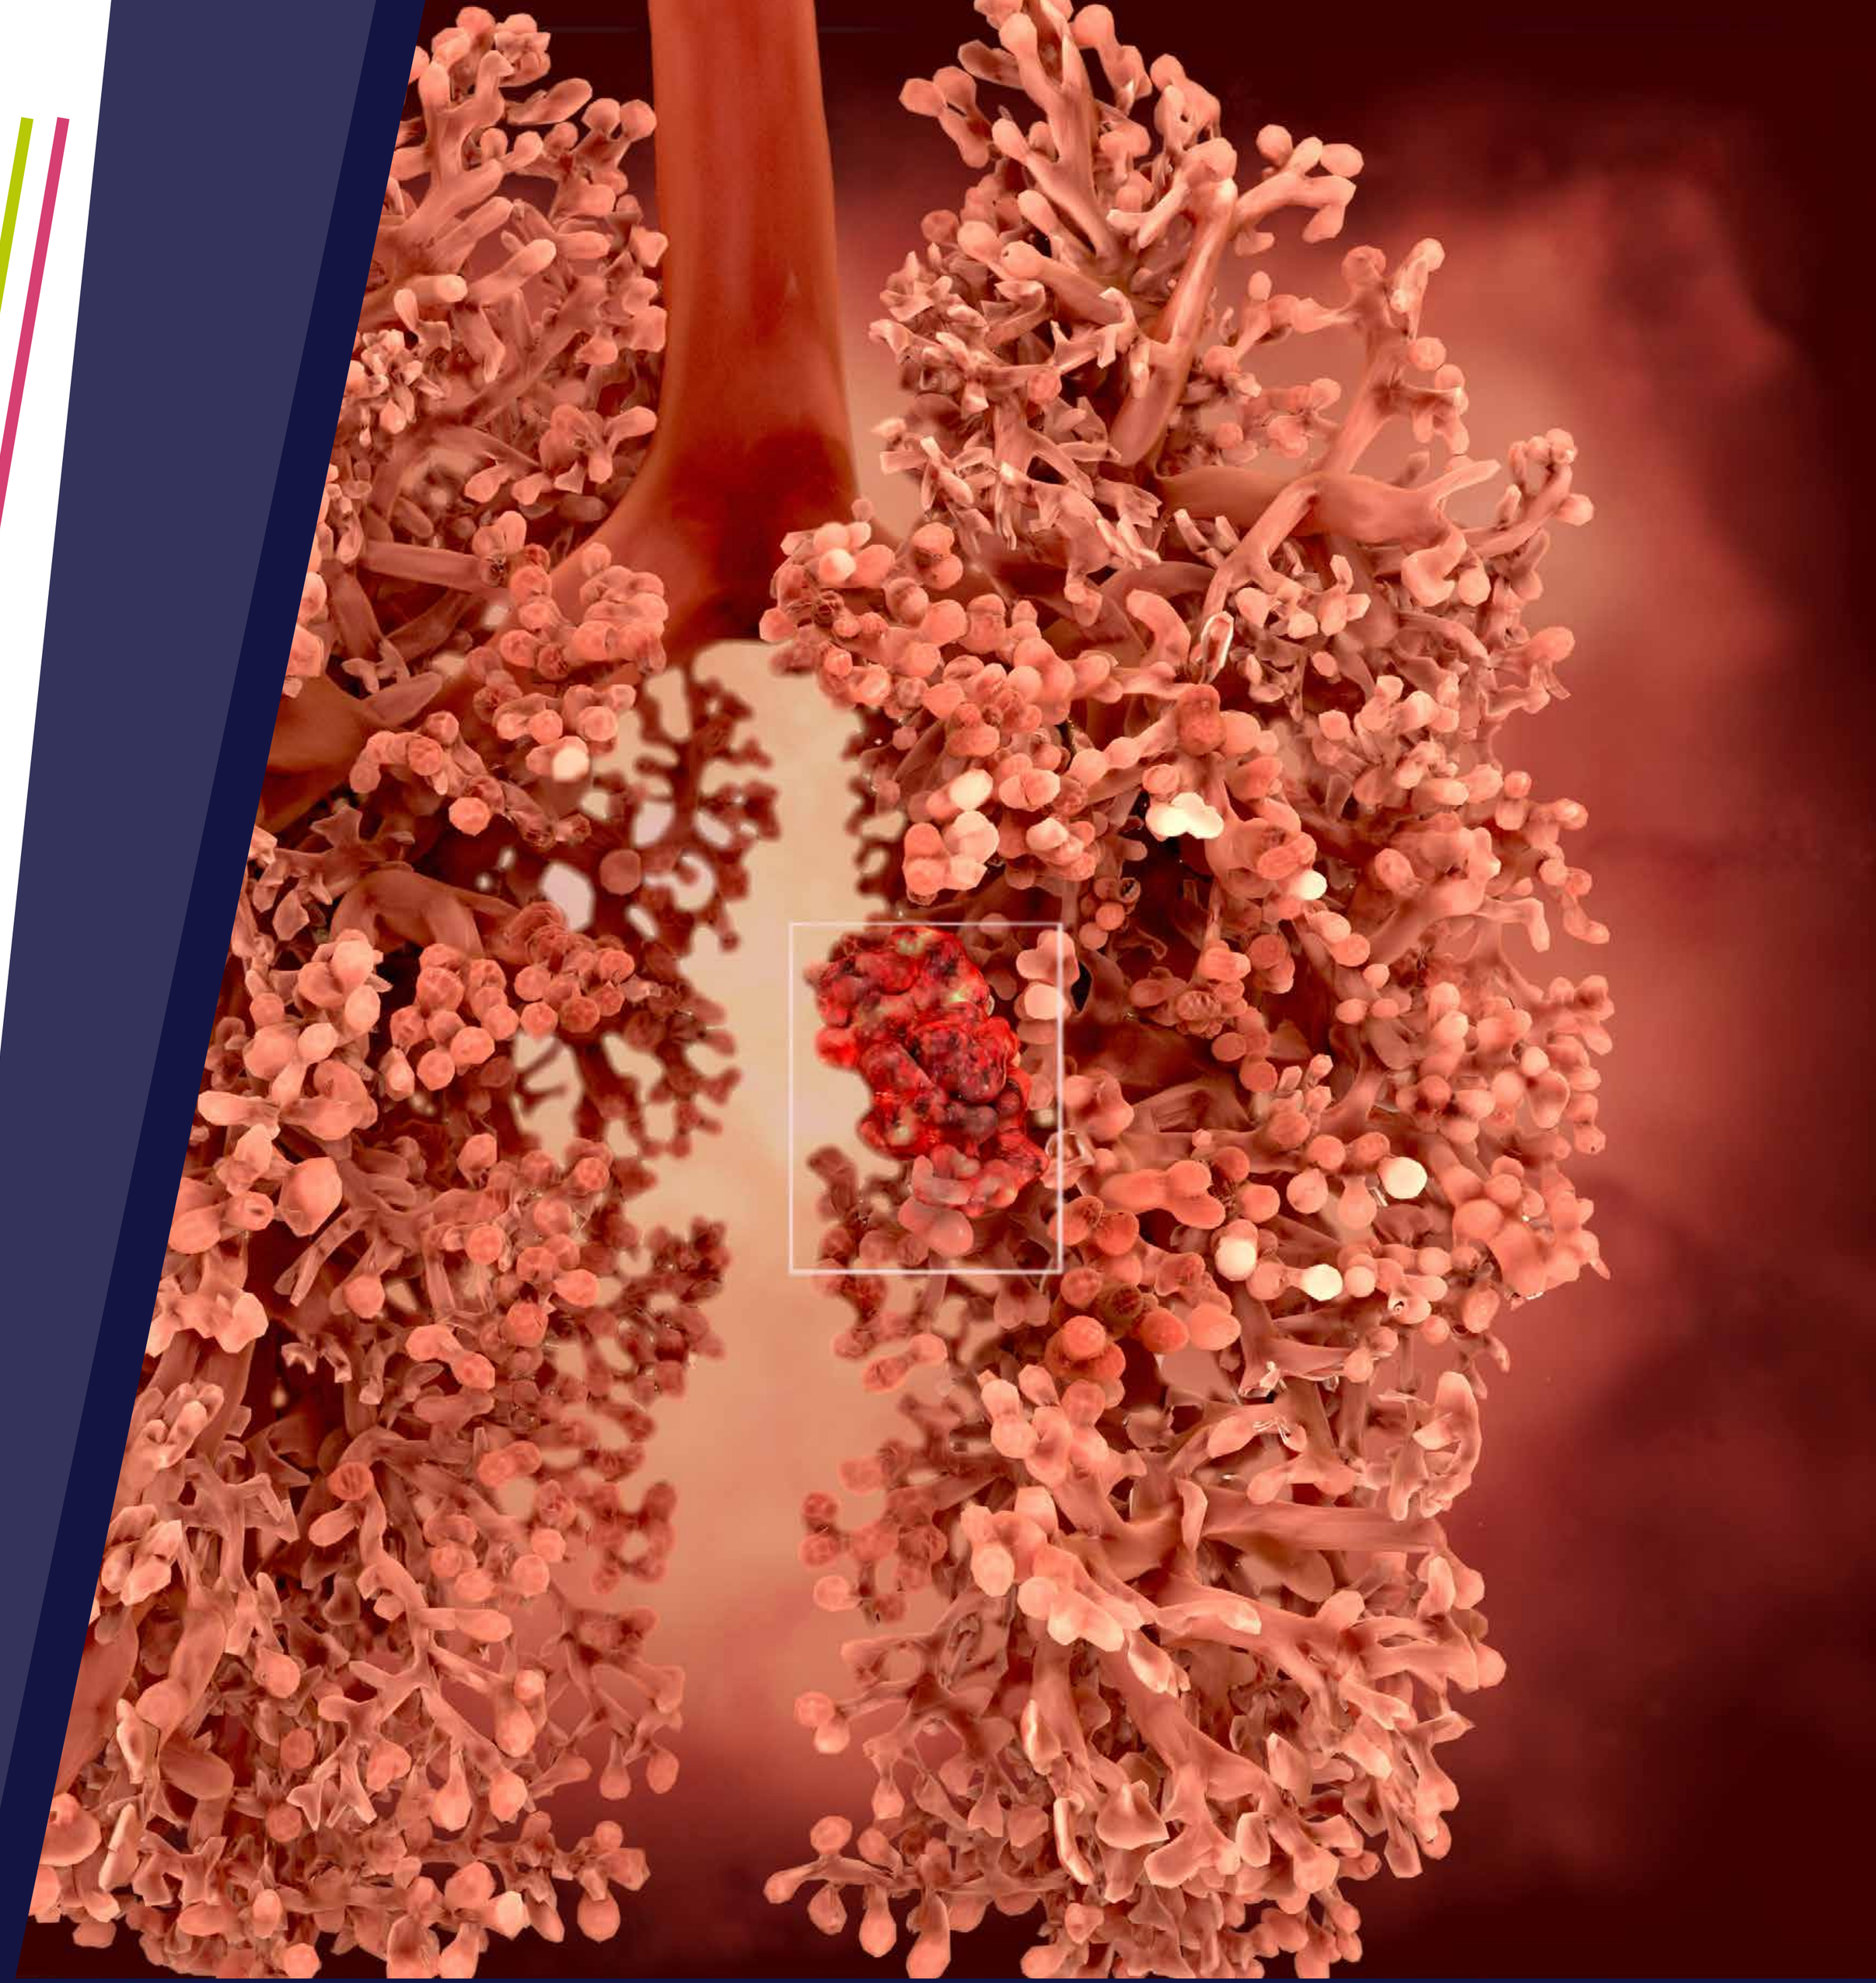

INTRODUCTION

Sevabertinib (BAY 2927088) is an oral tyrosine kinase inhibitor being developed for treatment of adults with advanced NSCLC harboring HER2-activating mutations[1,2]

OBJECTIVE

Primary objectives were to investigate the effect of:

- The CYP3A4/P-gp inhibitor itraconazole and CYP3A4/P-gp inducer carbamazepine on the PK of sevabertinib (NCT06348888)
- Sevabertinib on the PK of the CYP3A4 substrate midazolam (NCT06360211)
- Sevabertinib on P-gp using dabigatran etexilate, and on BCRP using rosuvastatin (NCT06329895)

METHODS

Three Phase I, open-label, fixed sequence, crossover trials were conducted in healthy participants

Dosing schema for the three clinical trials are presented in the online supplement

AUC and C_{max} were assessed, and safety was monitored throughout the studies

NCT06348888

Co-administration with itraconazole increased mean sevabertinib AUC by 125% and C_{max} by 60%

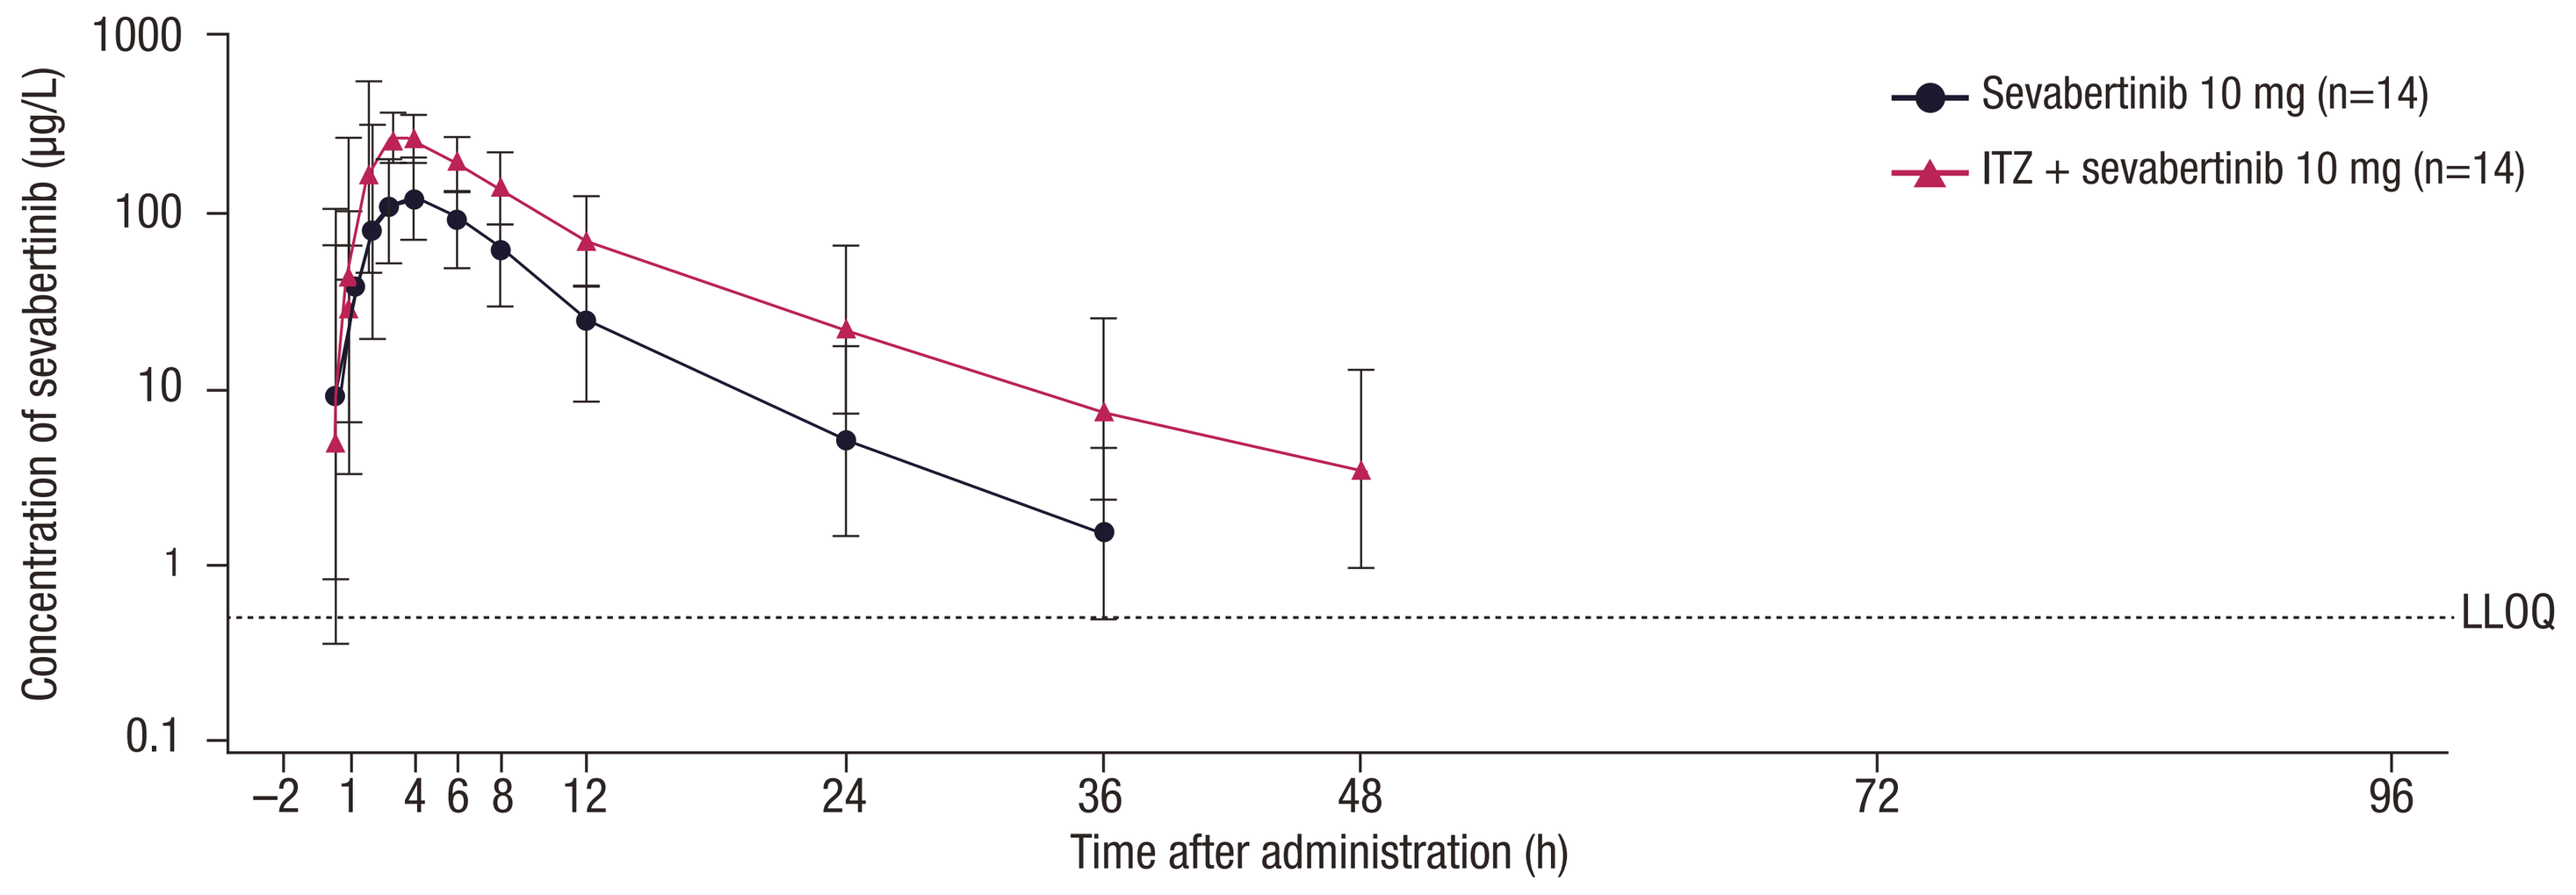
Concentration of sevabertinib (µg/L)
1000
100
10
1
0.1
–2
1
4
6
8
12
24
36
48
72
96
Time after administration (h)
Sevabertinib 10 mg (n=14)
ITZ + sevabertinib 10 mg (n=14)
LLOQ

AUC, area under the curve; C_{max}, maximum observed plasma drug concentration; h, hours; ITZ, itraconazole; LLOQ, lower limit of quantification.

Co-administration with carbamezapine reduced mean sevabertinib AUC by 79% and C_{max} by 57%

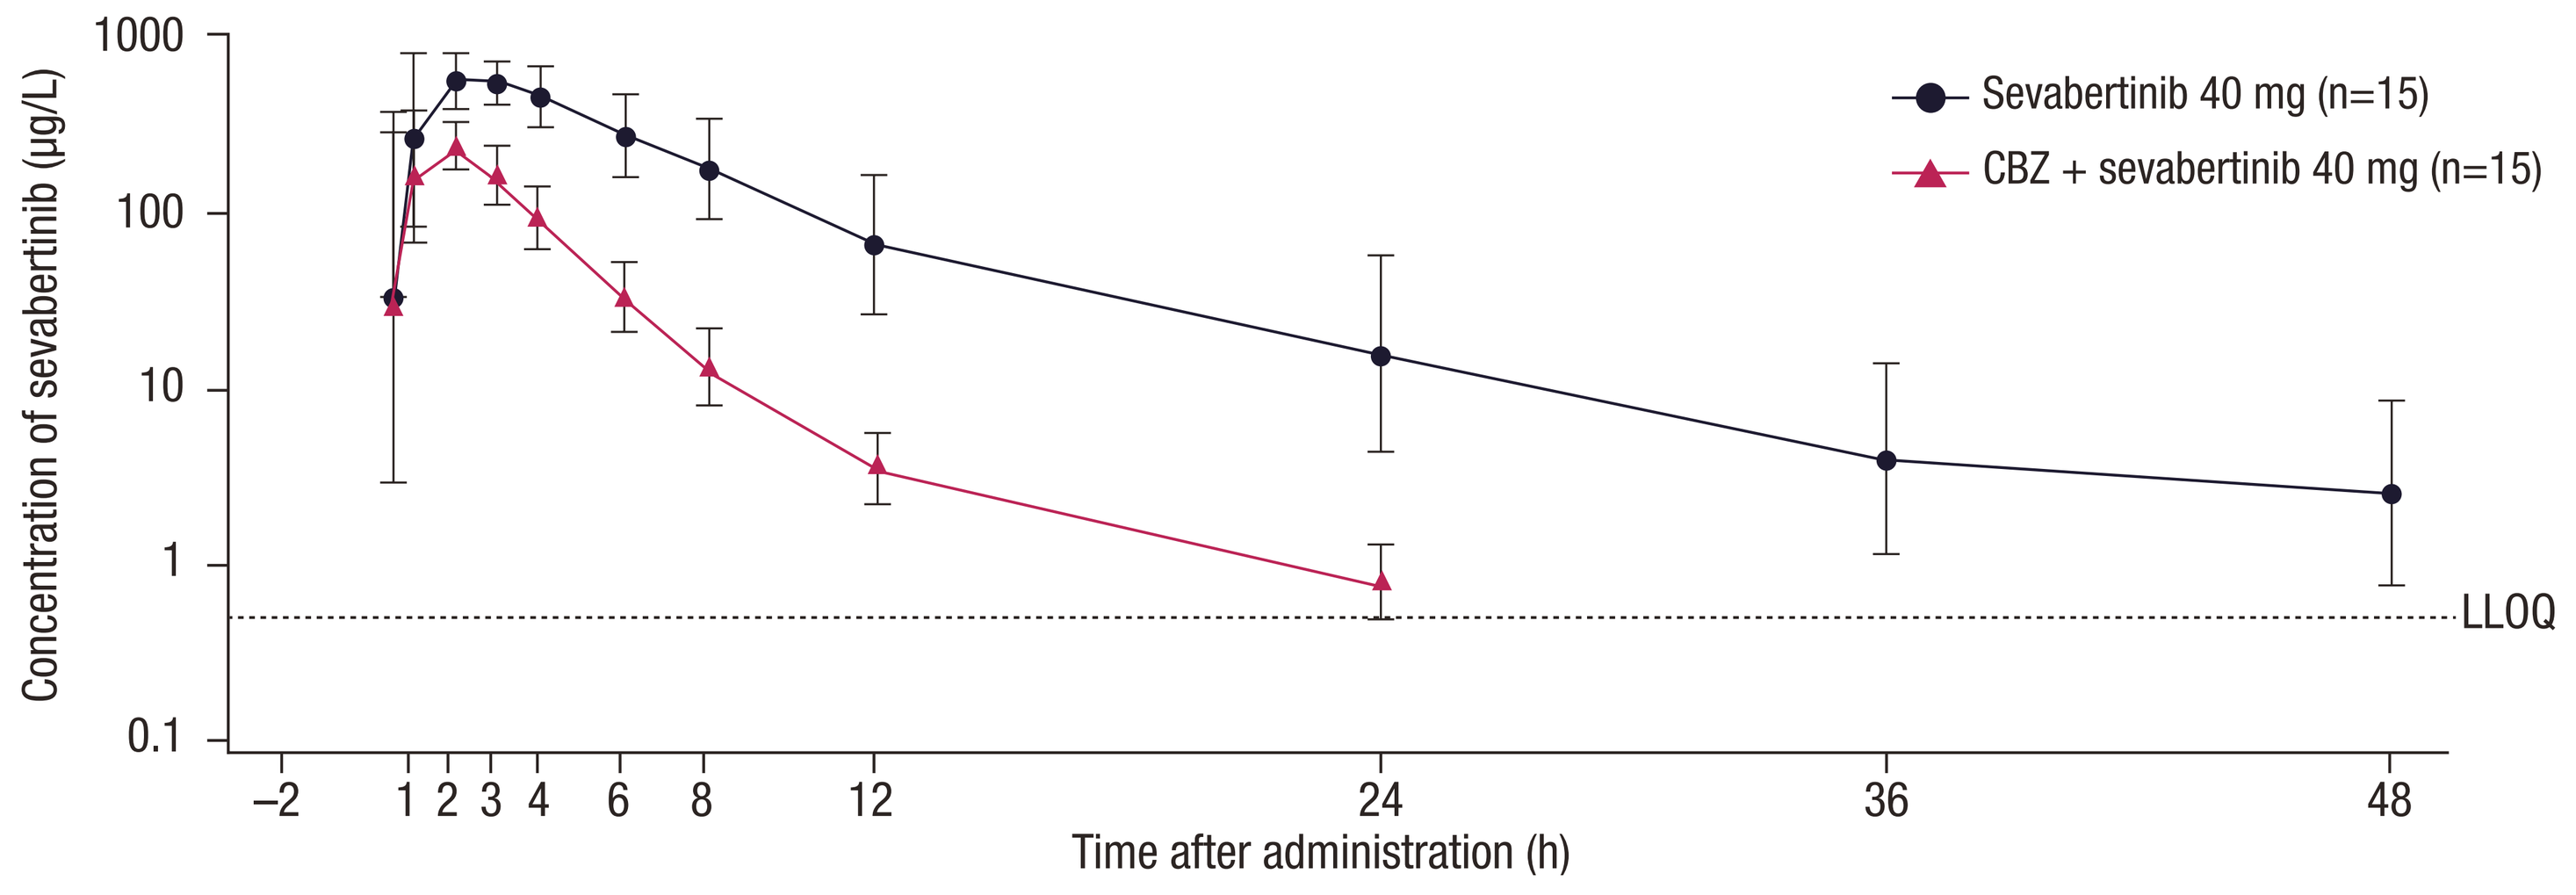
Concentration of sevabertinib (µg/L)
1000
100
10
1
0.1
–2
1
2
3
4
6
8
12
24
36
48
Time after administration (h)
Sevabertinib 40 mg (n=15)
CBZ + sevabertinib 40 mg (n=15)
LLOQ

AUC, area under the curve; CBZ, carbamazepine; C_{max}, maximum observed plasma drug concentration; h, hours; LLOQ, lower limit of quantification.

NCT06360211

Co-administration with a single dose of sevabertinib increased mean midazolam AUC by 10% and C_{max} by 10%
Pretreatment and co-administration with multiple doses of sevabertinib increased mean midazolam AUC by 95% and C_{max} by 79%

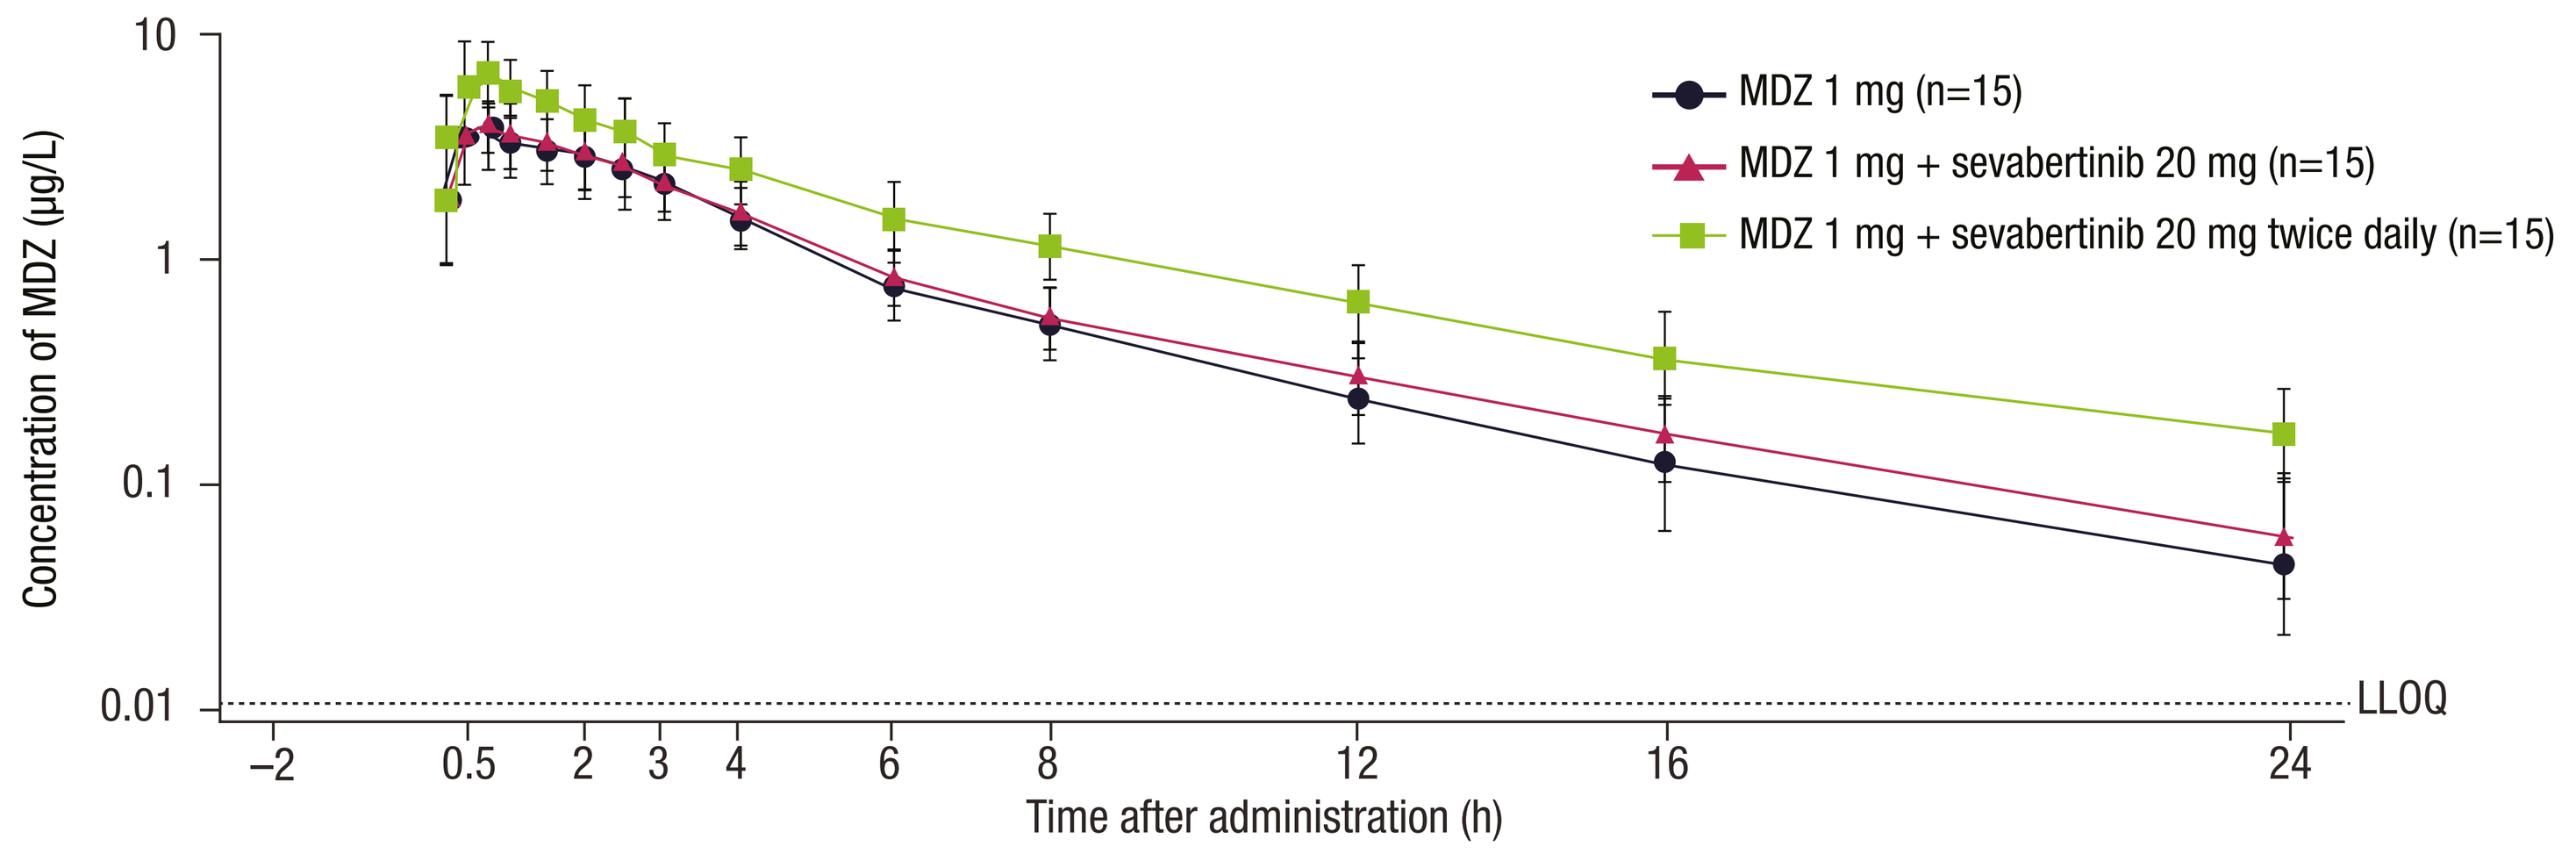
Concentration of MDZ (µg/L)
10
1
0.1
0.01
–2
0.5
2
3
4
6
8
12
16
24
Time after administration (h)
MDZ 1 mg (n=15)
MDZ 1 mg + sevabertinib 20 mg (n=15)
MDZ 1 mg + sevabertinib 20 mg twice daily (n=15)
LLOQ

AUC, area under the curve; C_{max}, maximum observed plasma drug concentration; h, hours; LLOQ, lower limit of quantification; MDZ, midazolam.

NCT06329895

Co-administration with sevabertinib increased unconjugated dabigatran AUC by 39% and decreased C_{max} by 4%

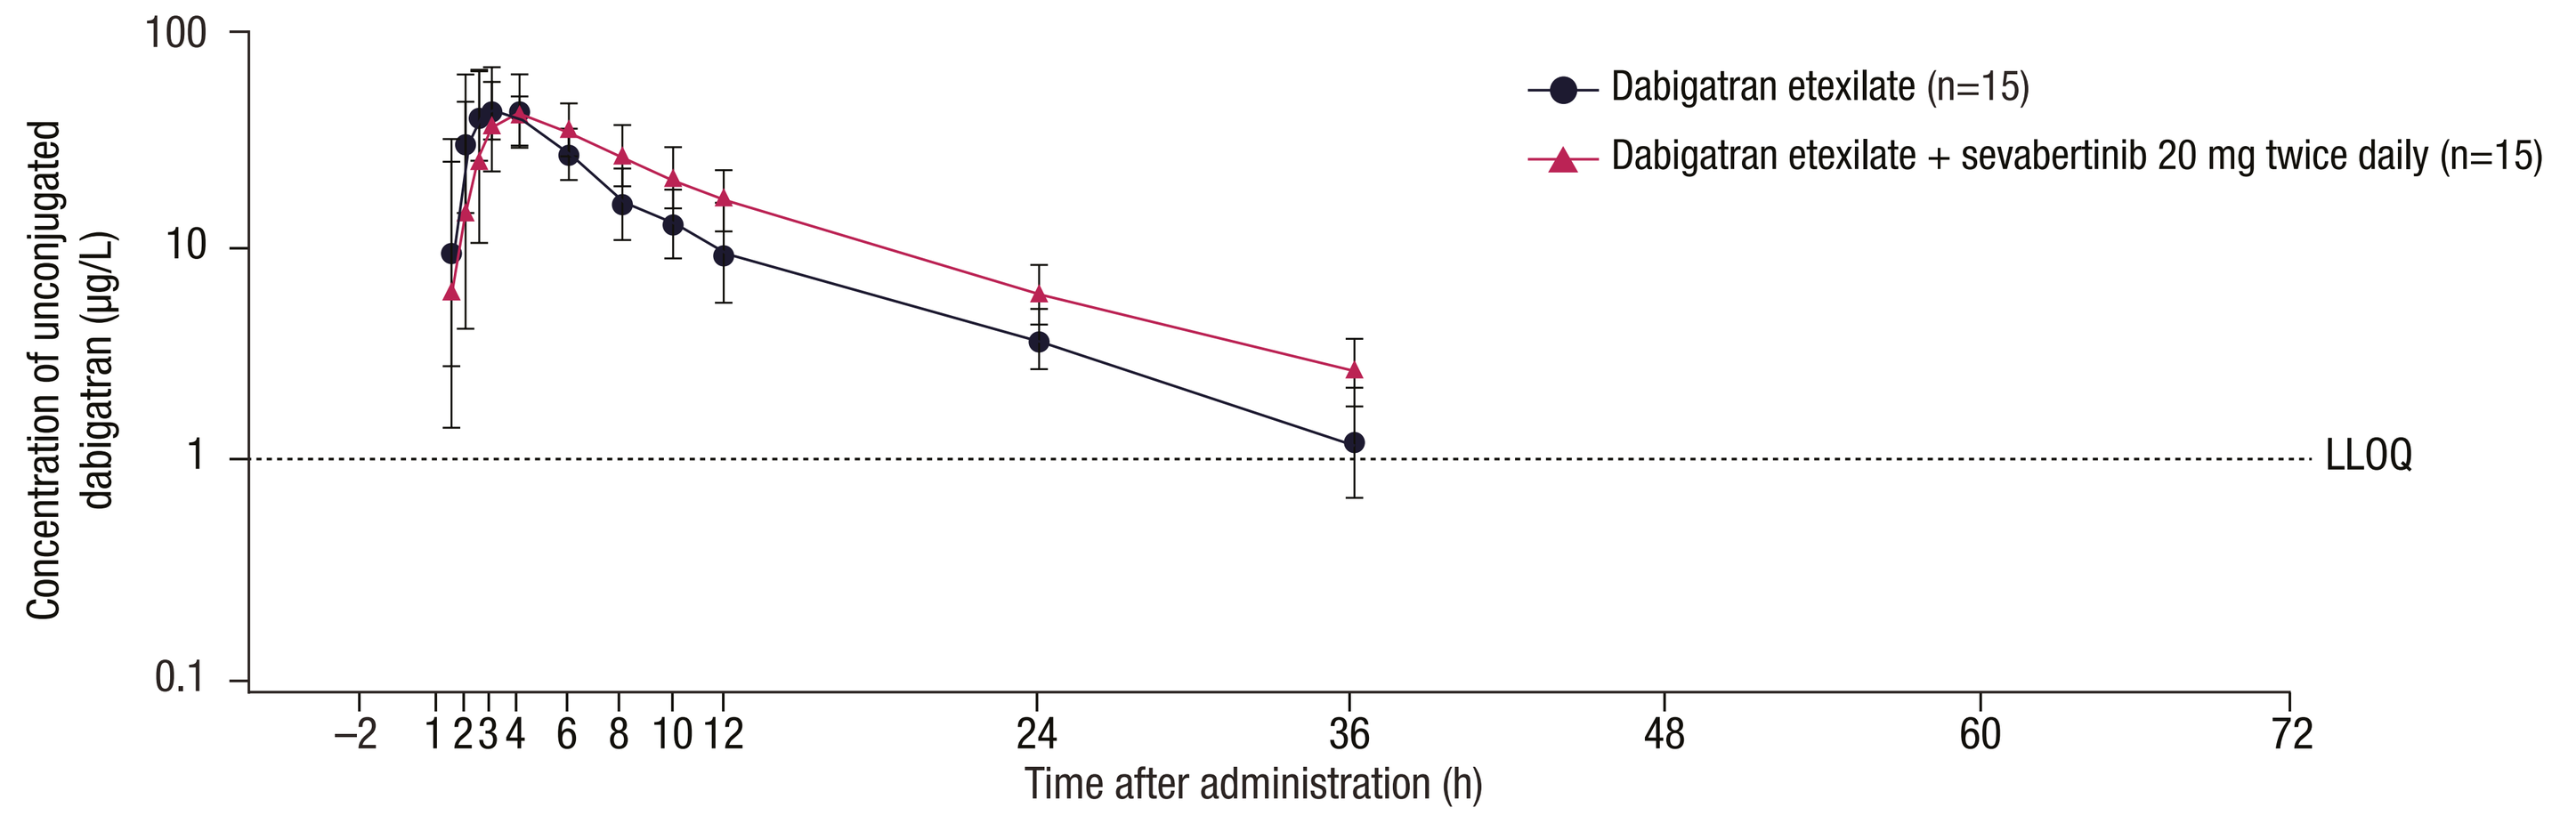
Concentration of unconjugated dabigatran (µg/L)
100
10
1
0.1
–2
1
2
3
4
6
8
10
12
24
36
48
60
72
Time after administration (h)
Dabigatran etexilate (n=15)
Dabigatran etexilate + sevabertinib 20 mg twice daily (n=15)
LLOQ

AUC, area under the curve; C_{max}, maximum observed plasma drug concentration; h, hours; LLOQ, lower limit of quantification.

Co-administration with sevabertinib increased rosuvastatin AUC by 25% and C_{max} by 37%

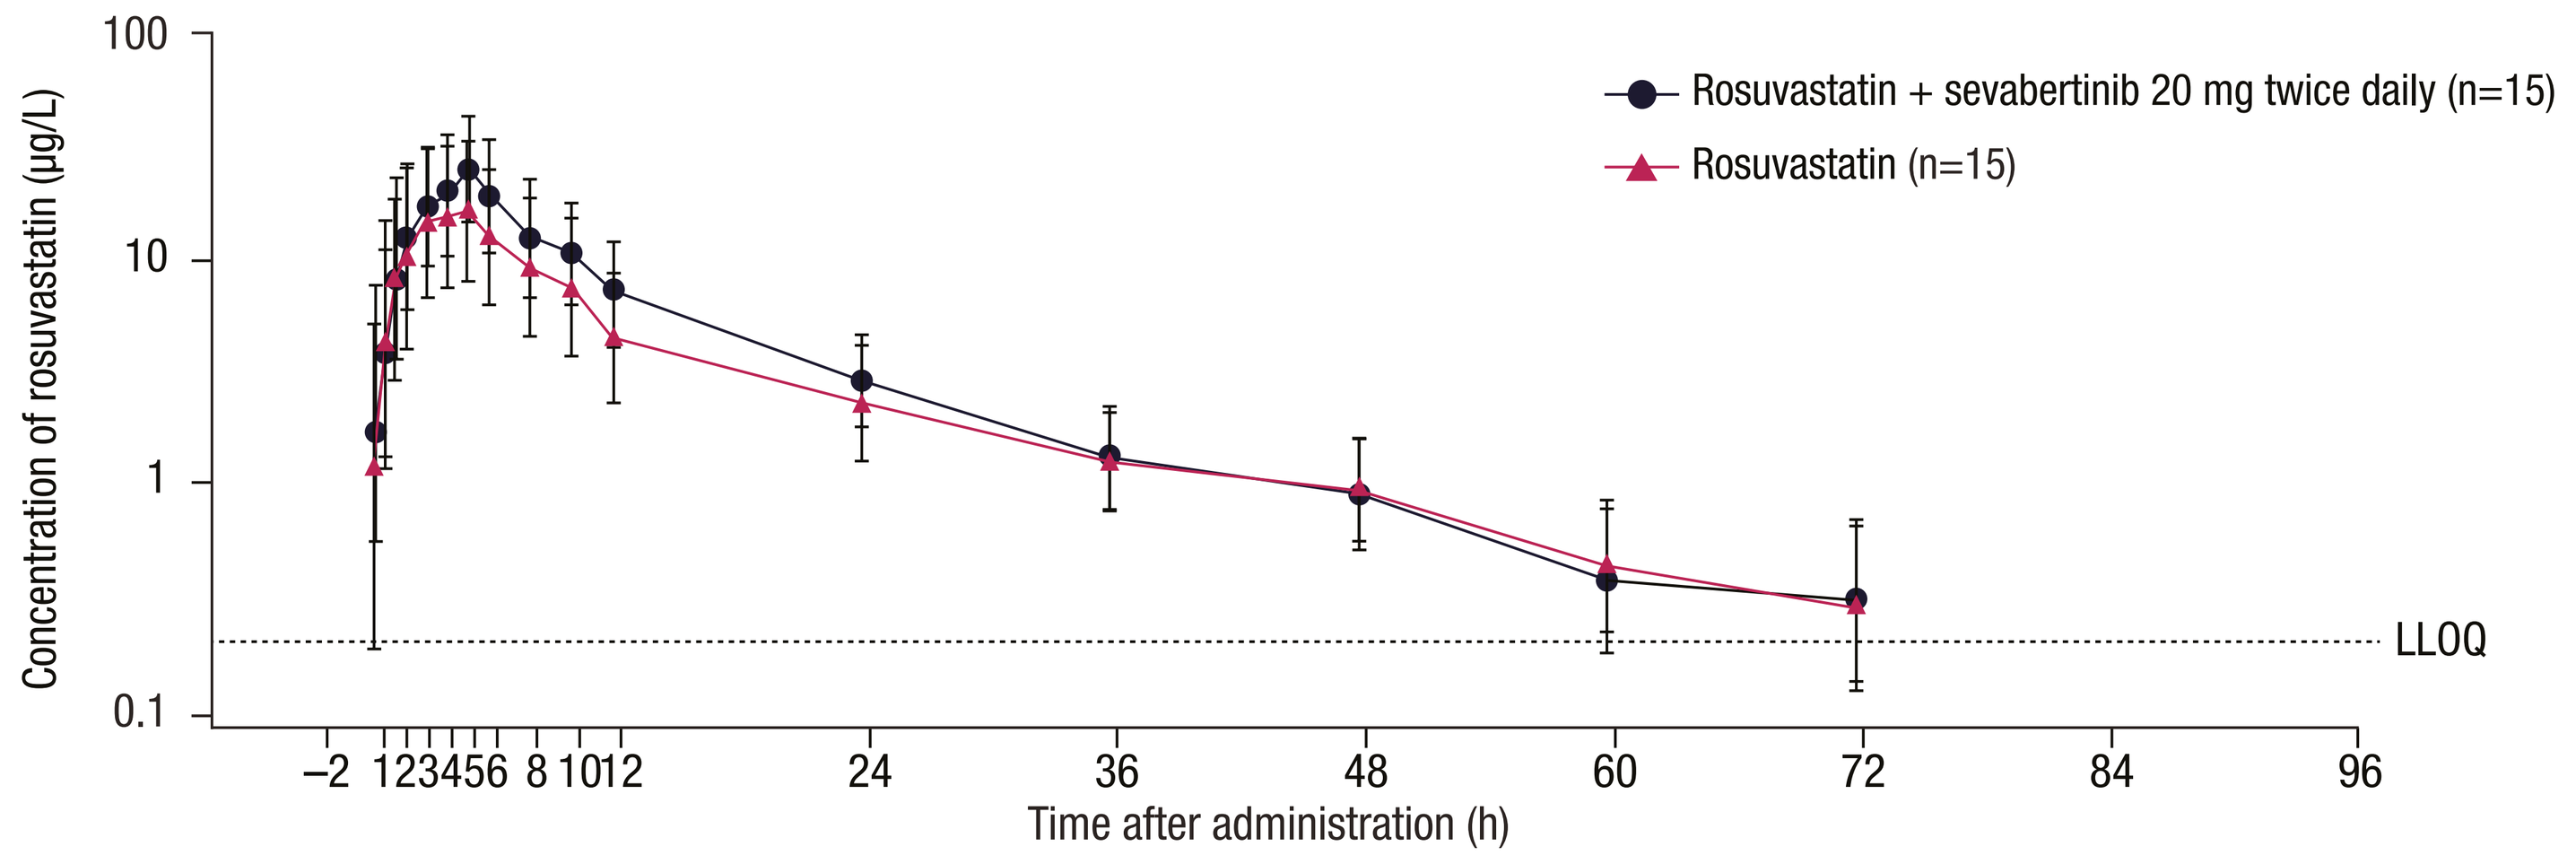
Concentration of rosuvastatin (µg/L)
100
10
1
0.1
–2
1
2
3
4
5
6
8
10
12
24
36
48
60
72
84
96
Time after administration (h)
Rosuvastatin + sevabertinib 20 mg twice daily (n=15)
Rosuvastatin (n=15)
LLOQ

AUC, area under the curve; C_{max}, maximum observed plasma drug concentration; h, hours; LLOQ, lower limit of quantification.

CONCLUSIONS

- Sevabertinib exposure was increased by CYP3A4/P-gp inhibition and reduced by CYP3A4/P-gp induction
- Our findings indicate that sevabertinib is a weak inhibitor of CYP3A4, P-gp, and BCRP
- Sevabertinib was well tolerated in all three studies

SCAN ME FOR MORE INFO

References: 1. Siegel F et al. Cancer Res 2023;83(Suppl 7):1098-1099;
2. Siegel F et al. EJC Supplements 2022;174(Suppl 1):S9-S10.

Abbreviations: AUC, area under the curve; BCRP, breast cancer resistant protein; C_{max}, maximum observed plasma drug concentration; CYP3A4, cytochrome P450 3A4; HER2, human epidermal growth factor receptor 2; NSCLC, non-small cell lung cancer; P-gp, P-glycoprotein; PK, pharmacokinetics.

Presented at the National Community Oncology Dispensing Association (NCODA) International Fall Summit 2025, October 16–17, Orlando, FL, USA

Originally presented at the American Society for Clinical Pharmacology and Therapeutics (ASCPT) Annual Meeting 2025, May 28–31, Washington, D.C USA

This study was supported by Bayer AG. Medical writing services were provided by Laura Chalmers, PhD, of Adelphi Communications Ltd (Bollington, UK), funded by Bayer, in accordance with Good Publication Practice 2022 guidelines.