SEVABERTINIB

THE EFFECTS OF FOOD AND ESOMEPRAZOLE ON THE PHARMACOKINETICS OF SEVABERTINIB

Ahmed Elsayed, PharmD[1], on behalf of Barbara J. Brennan, PharmD[1], Michaela Damaske[2], Frank-Thorsten Hafner, PhD[3], Uwe Muenster, PhD[3], Andreas Lender, PhD[3], Jan Joseph, PhD[3], Stefanie Reif, PhD[2], Sue Prendergast, MSc[4], Philip Lienau, PhD[2], Bart Ploeger, PhD[2], Chunlin Chen, MD, PhD[1,a]

[1]Bayer HealthCare Pharmaceuticals, Inc., Whippany, New Jersey, USA; [2]Bayer AG, Berlin, Germany; [3]Bayer AG, Wuppertal, Germany; [4]Bayer plc, Reading, UK
[a]Affiliation at the time the study was performed

INTRODUCTION

Sevabertinib (BAY 2927088) is an oral tyrosine kinase inhibitor that potently inhibits mutant *HER2* and is being developed for the treatment of adult patients with advanced NSCLC harboring *HER2*-activating mutations[1,2]

OBJECTIVE

To evaluate the effect of food or the acid-reducing agent esomeprazole on the PK of sevabertinib in healthy participants

METHODS

An open-label, randomized, crossover study in healthy participants was conducted from March 29 to June 11, 2024

Over 14 days, participants received single 20 mg doses of sevabertinib under one of the following interventions: A) a fasted state, B) following a light, low-fat meal, or C) following a high-fat, high-calorie meal

Esomeprazole 40 mg was administered for 5 days with a single dose of sevabertinib on Day 4 following a light meal. Participants therefore received four 20 mg doses of sevabertinib

Safety and tolerability were closely monitored throughout the study

Abbreviations: HER2, human epidermal growth factor receptor 2; NSCLC, non-small cell lung cancer; PK, pharmacokinetics.

Mean AUC and C_{max} were reduced by 16% and 28%, respectively, when administered with a low-fat meal compared with a fasted state

Linear (A) and semi-logarithmic (B) plasma concentration–time profiles of sevabertinib after a 20 mg single-dose administration in fasted and fed conditions

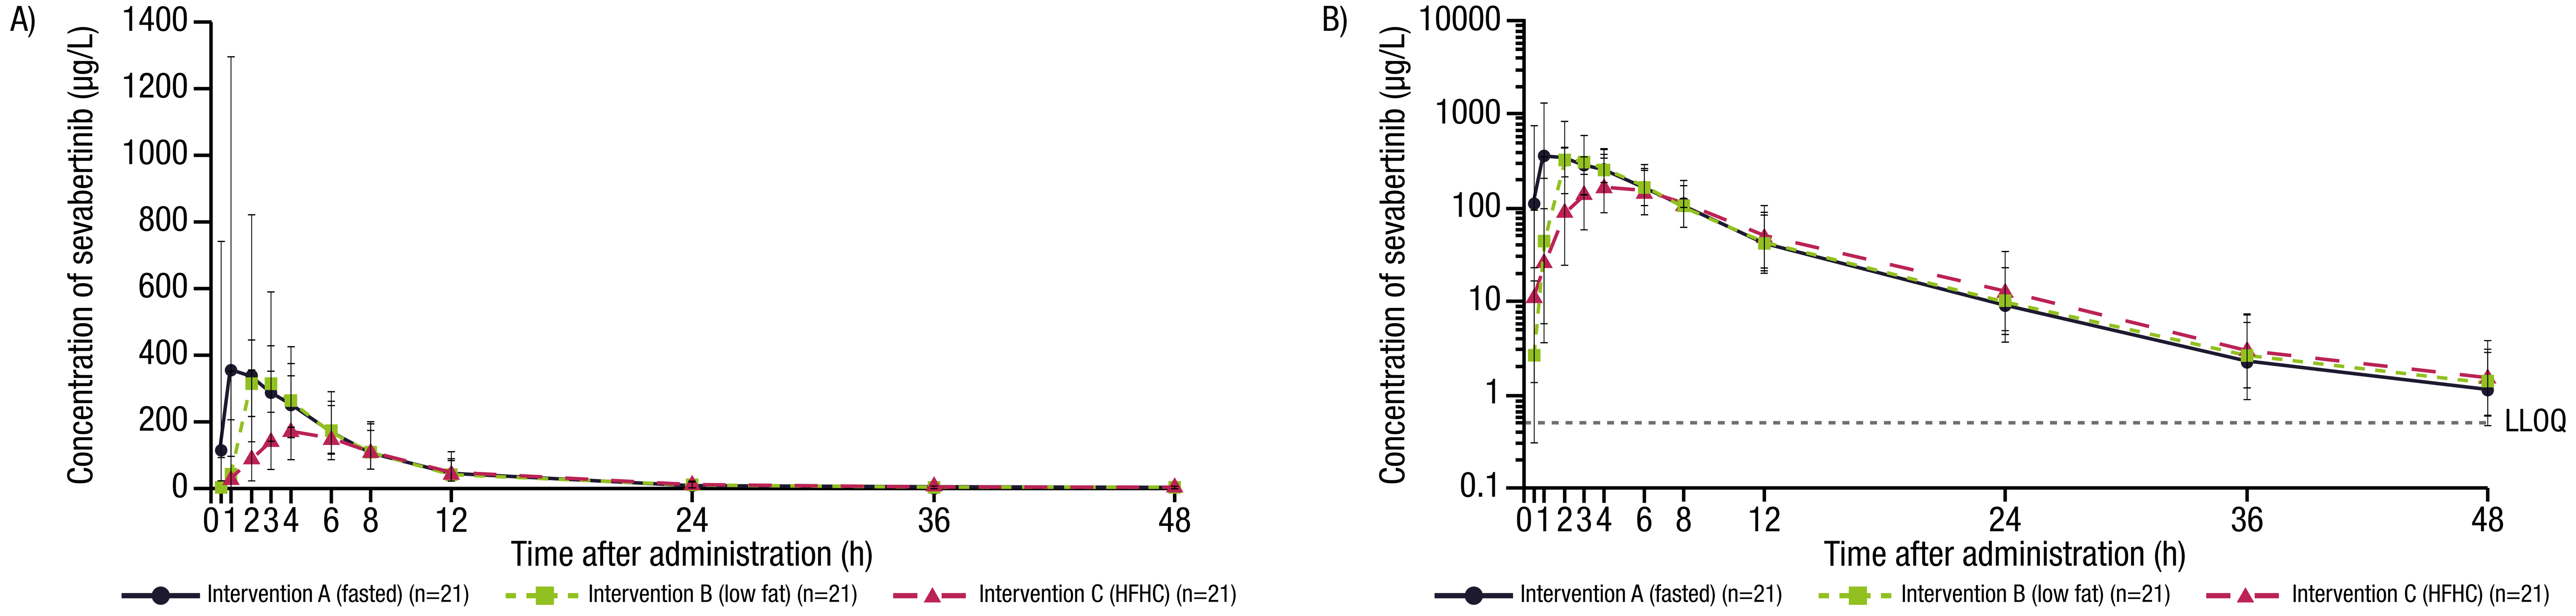
A)
Concentration of sevabertinib (µg/L)
1400
1200
1000
800
600
400
200
0
0 1 2 3 4 6 8 12 24 36 48
Time after administration (h)
Intervention A (fasted) (n=21)
Intervention B (low fat) (n=21)
Intervention C (HFHC) (n=21)
B)
Concentration of sevabertinib (µg/L)
10000
1000
100
10
1
0.1
LLOQ
0 1 2 3 4 6 8 12 24 36 48
Time after administration (h)
Intervention A (fasted) (n=21)
Intervention B (low fat) (n=21)
Intervention C (HFHC) (n=21)

Data shown as geometric mean ± standard deviation.
AUC, area under the curve; C_{max}, maximum observed drug concentration; LLOQ, lower limit of quantification; h, hours; HFHC, high-fat, high-calorie.

Co-administration of esomeprazole did not affect mean AUC of sevabertinib and decreased mean C_{max} of sevabertinib by 29% compared with sevabertinib alone

Linear (A) and semi-logarithmic (B) plasma concentration–time profiles of sevabertinib after a 20 mg single-dose administration alone and co-administered with esomeprazole

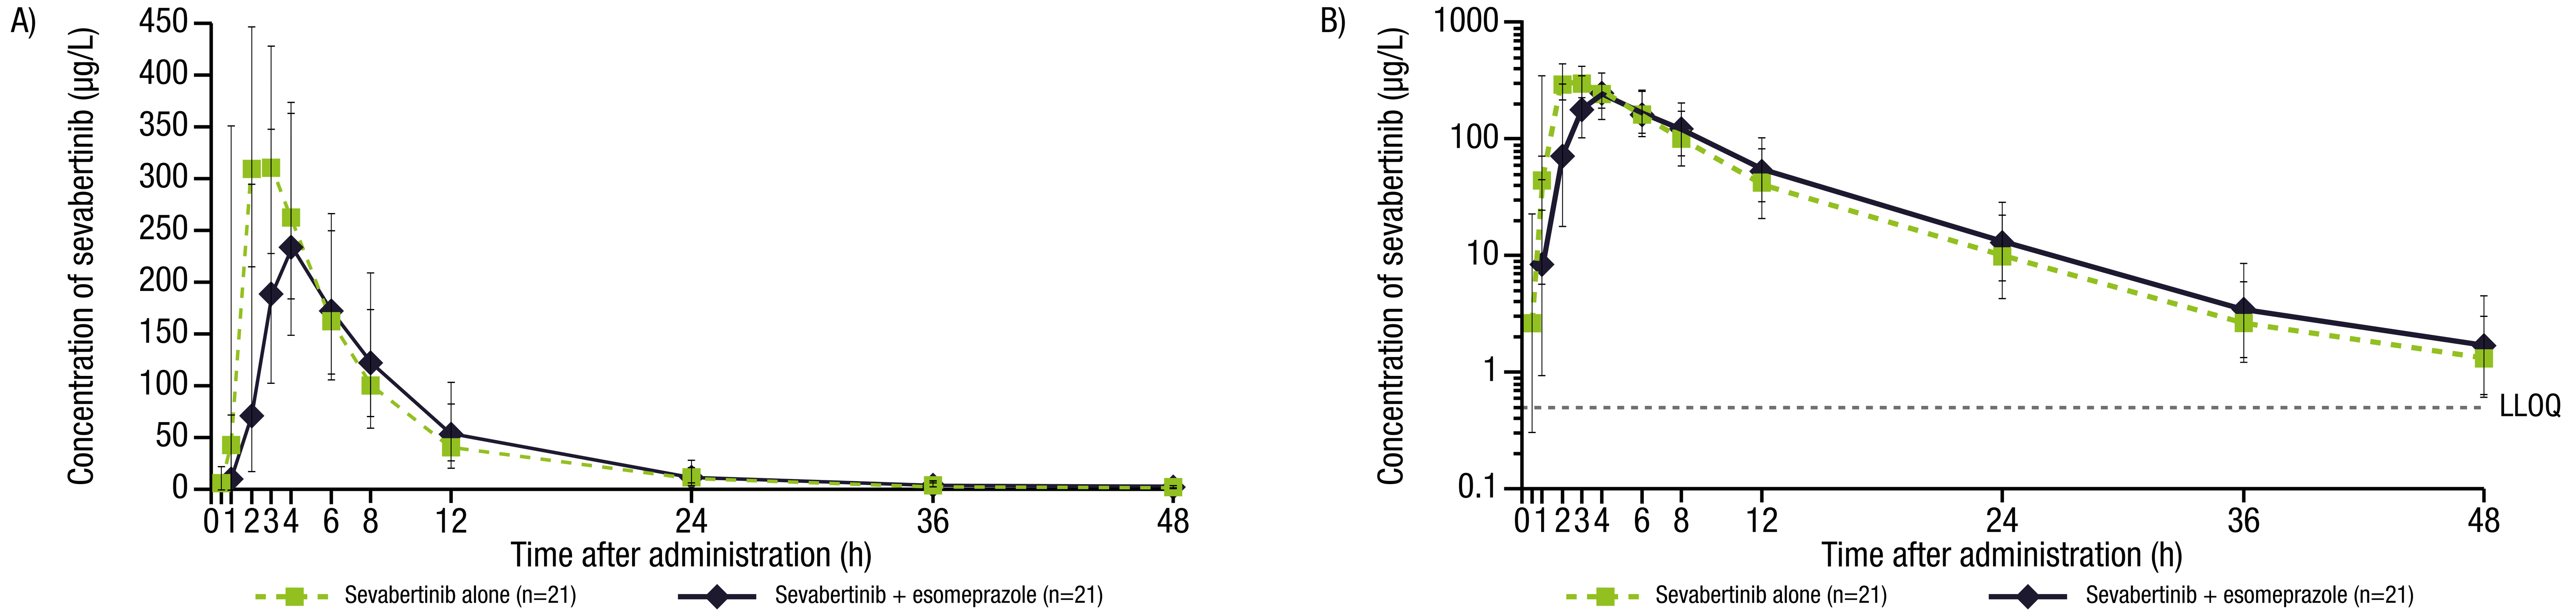
A)
Concentration of sevabertinib (µg/L)
450
400
350
300
250
200
150
100
50
0
0 1 2 3 4 6 8 12 24 36 48
Time after administration (h)
Sevabertinib alone (n=21)
Sevabertinib + esomeprazole (n=21)
B)
Concentration of sevabertinib (µg/L)
1000
100
10
1
0.1
LLOQ
0 1 2 3 4 6 8 12 24 36 48
Time after administration (h)
Sevabertinib alone (n=21)
Sevabertinib + esomeprazole (n=21)

Data shown as geometric mean ± standard deviation. Sevabertinib alone was administered under intervention B (low fat).
AUC, area under the curve; C_{max}, maximum observed drug concentration; LLOQ, lower limit of quantification; h, hours.

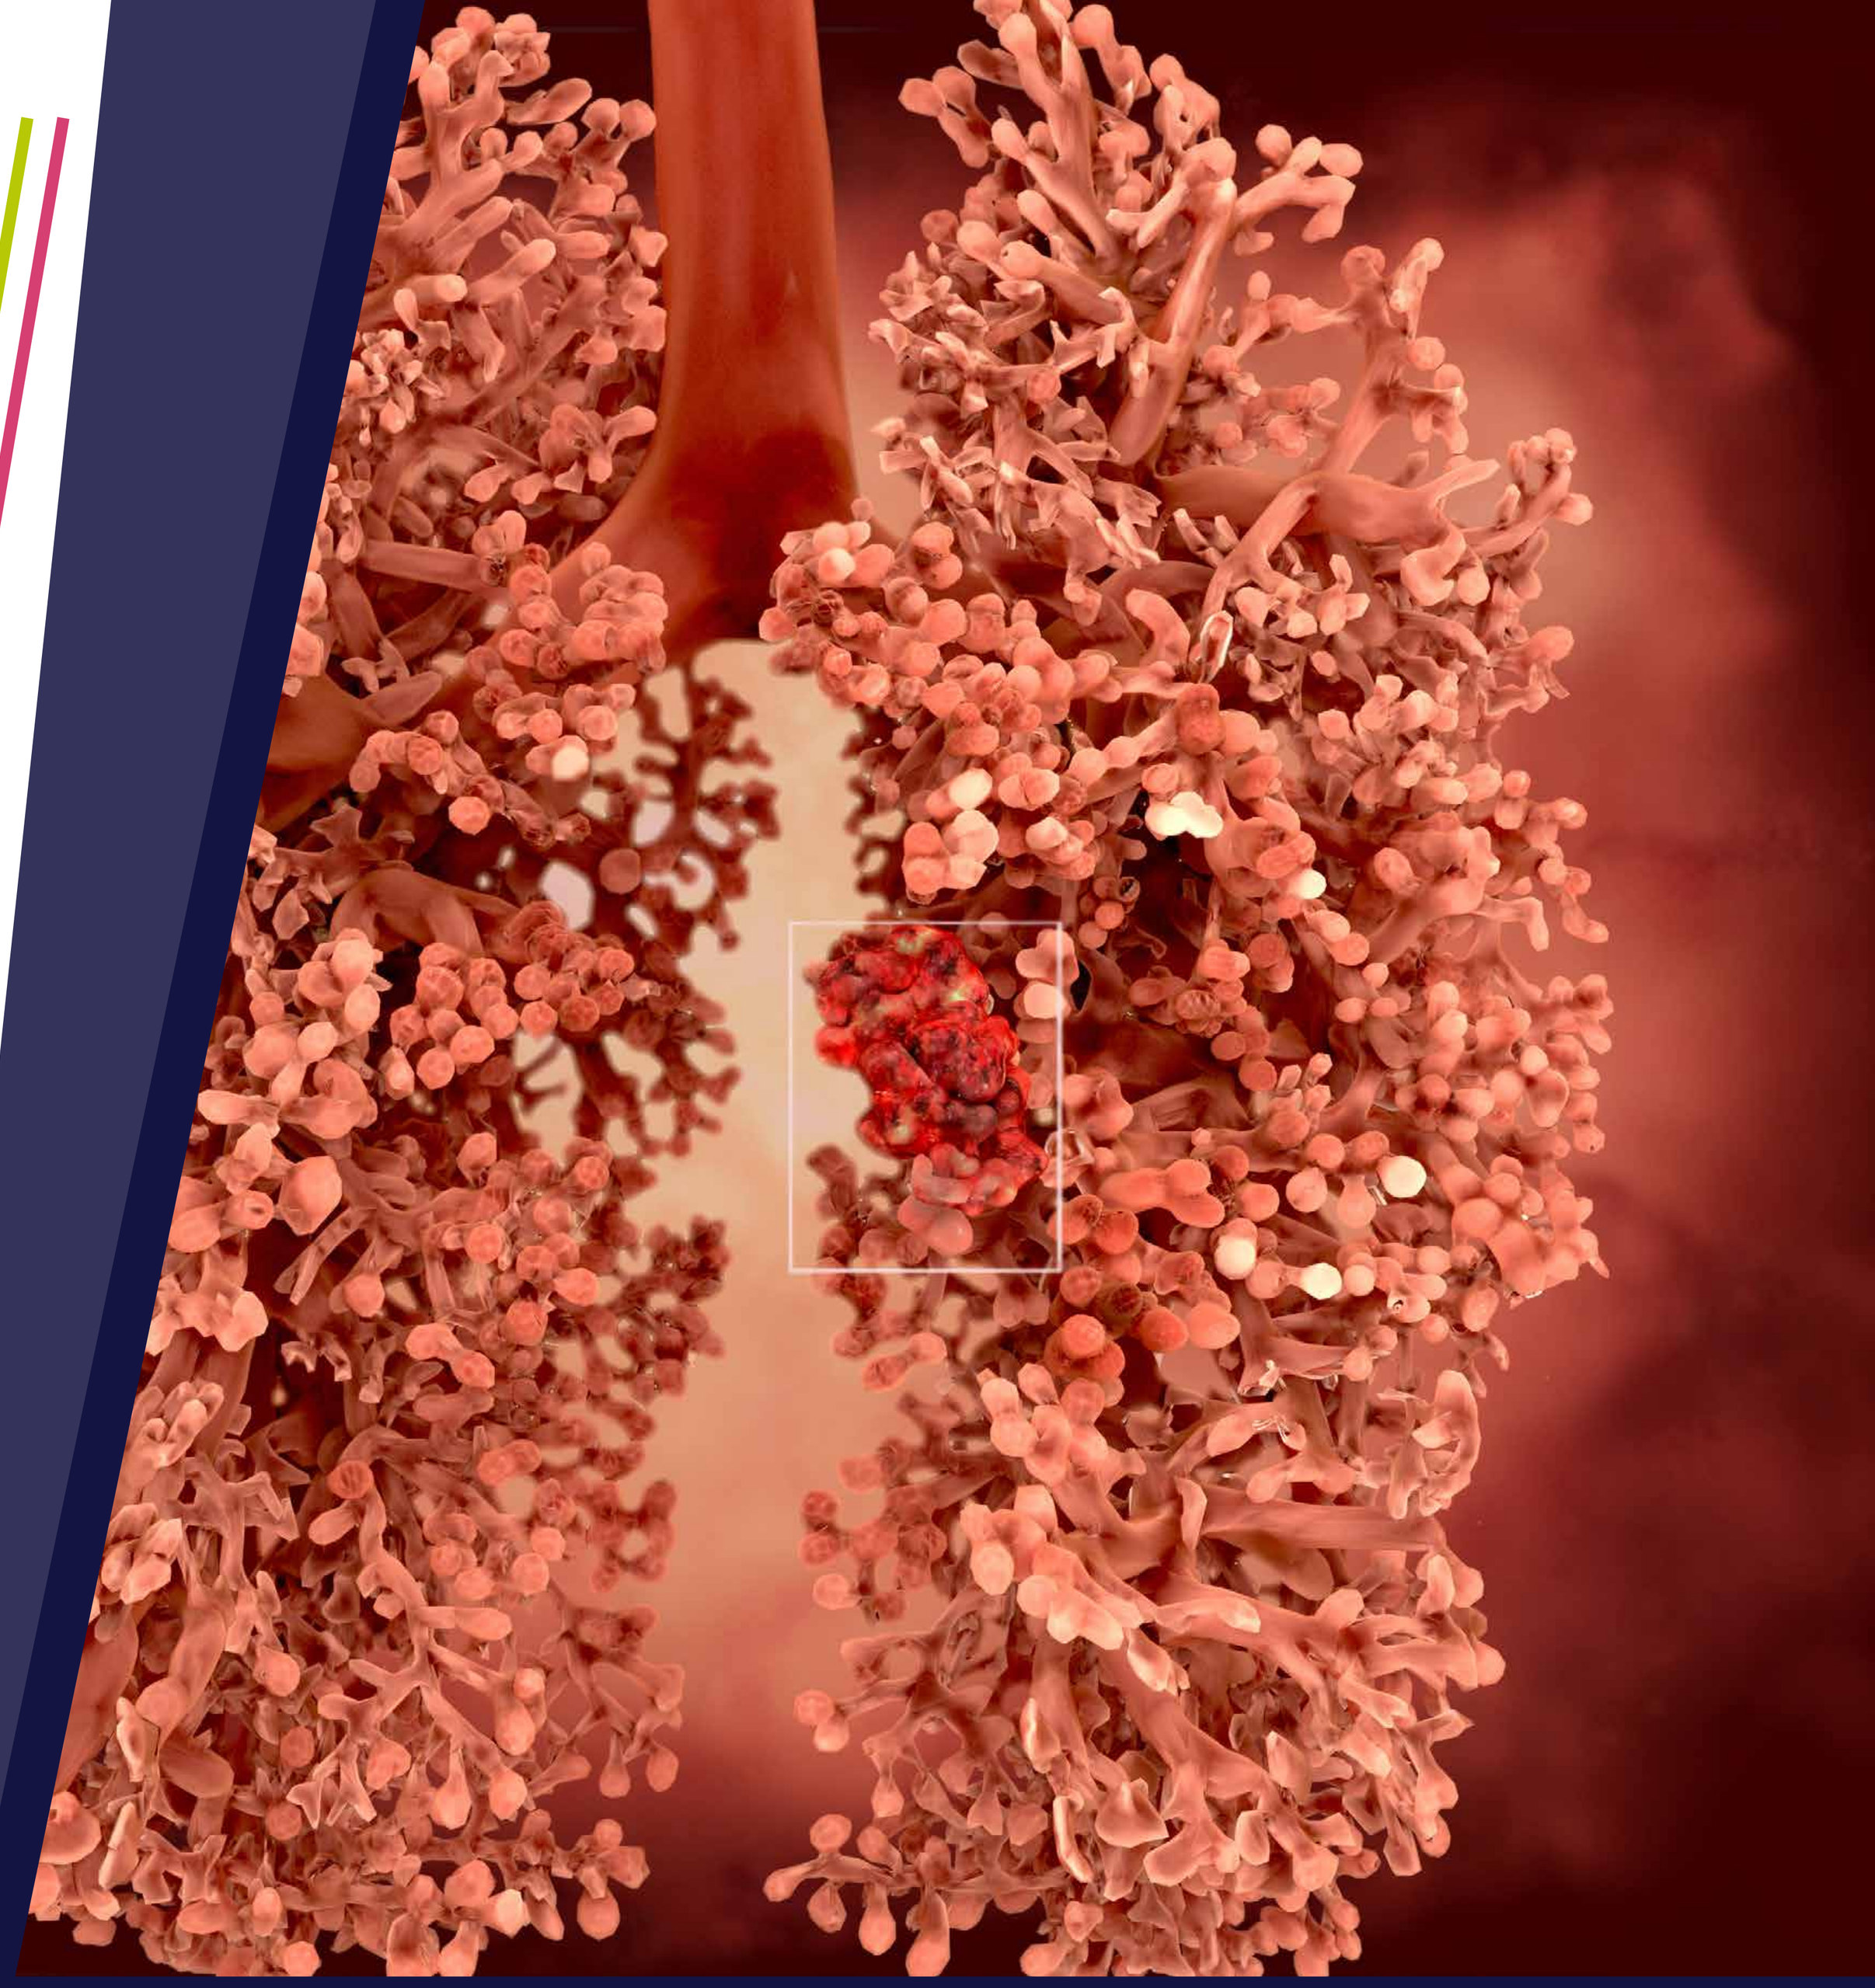

CONCLUSIONS

- Sevabertinib exposure was decreased slightly when administered in a fed state, compared with a fasted state, with no clinically relevant difference between meal types
- Sevabertinib exposure was not affected by esomeprazole co-administration, although a slight decrease in peak sevabertinib concentration was observed
- Sevabertinib was safe and well tolerated in healthy participants

SCAN ME FOR MORE INFO

References: 1. Siegel F et al. Cancer Res 2023;83(Suppl 7):1098-1099;
2. Siegel F et al. EJC Supplements 2022;174(Suppl 1):S9-S10.

Presented at the National Community Oncology Dispensing Association (NCODA) International Fall Summit 2025, October 16–17, Orlando, FL, USA

Originally presented at the American Society for Clinical Pharmacology and Therapeutics (ASCPT) Annual Meeting 2025, May 28–31, Washington, D.C, USA

This study was supported by Bayer AG. Medical writing services were provided by Rachael Powis, PhD, of Adelphi Communications Ltd (Bollington, UK), funded by Bayer, in accordance with Good Publication Practice 2022 guidelines.