DAROLUTAMIDE

PROSTATE-SPECIFIC ANTIGEN RESPONSE WITH DAROLUTAMIDE PLUS ANDROGEN-DEPRIVATION THERAPY IN PATIENTS WITH METASTATIC HORMONE-SENSITIVE PROSTATE CANCER IN ARANOTE

Kirollos S. Hanna, PharmD, BCPS, BCOP, FACCC, FAPO[1,2], on behalf of Fred Saad, MD[3], Kunhi Parambath Haresh, MD[4], Neal Shore, MD, FACS[5], Egils Vjaters, MD[6], David Olmos, MD, PhD[7], Natasha Littleton, MSc[8], Isabella Testa, MD[9], Shankar Srinivasan, PhD[10], Frank Verholen, MD[11], Alberto Briganti, MD, PhD[12]

[1]Minnesota Oncology, St. Paul, Minnesota, USA; [2]Mayo Clinic College of Medicine, Rochester, Minnesota, USA; [3]Department of Surgery/Urology, Centre Hospitalier de l'Université de Montréal, University of Montreal, Montreal, Quebec, Canada; [4]All India Institute of Medical Sciences, New Delhi, India; [5]START Carolinas/Carolina Urologic Research Center, Myrtle Beach, South Carolina, USA; [6]P. Stradinš Clinical University Hospital, Riga, Latvia; [7]Department of Medical Oncology, Hospital Universitario 12 de Octubre, Instituto de Investigación Sanitaria Hospital 12 de Octubre (Imas12), Madrid, Spain; [8]Bayer Ltd, Dublin, Ireland; [9]Bayer S.p.A, Milan, Italy; [10]Bayer Healthcare, Whippany, NJ, USA; [11]Bayer Consumer Care AG, Basel, Switzerland; [12]Vita-Salute University San Raffaele, Milan, Italy

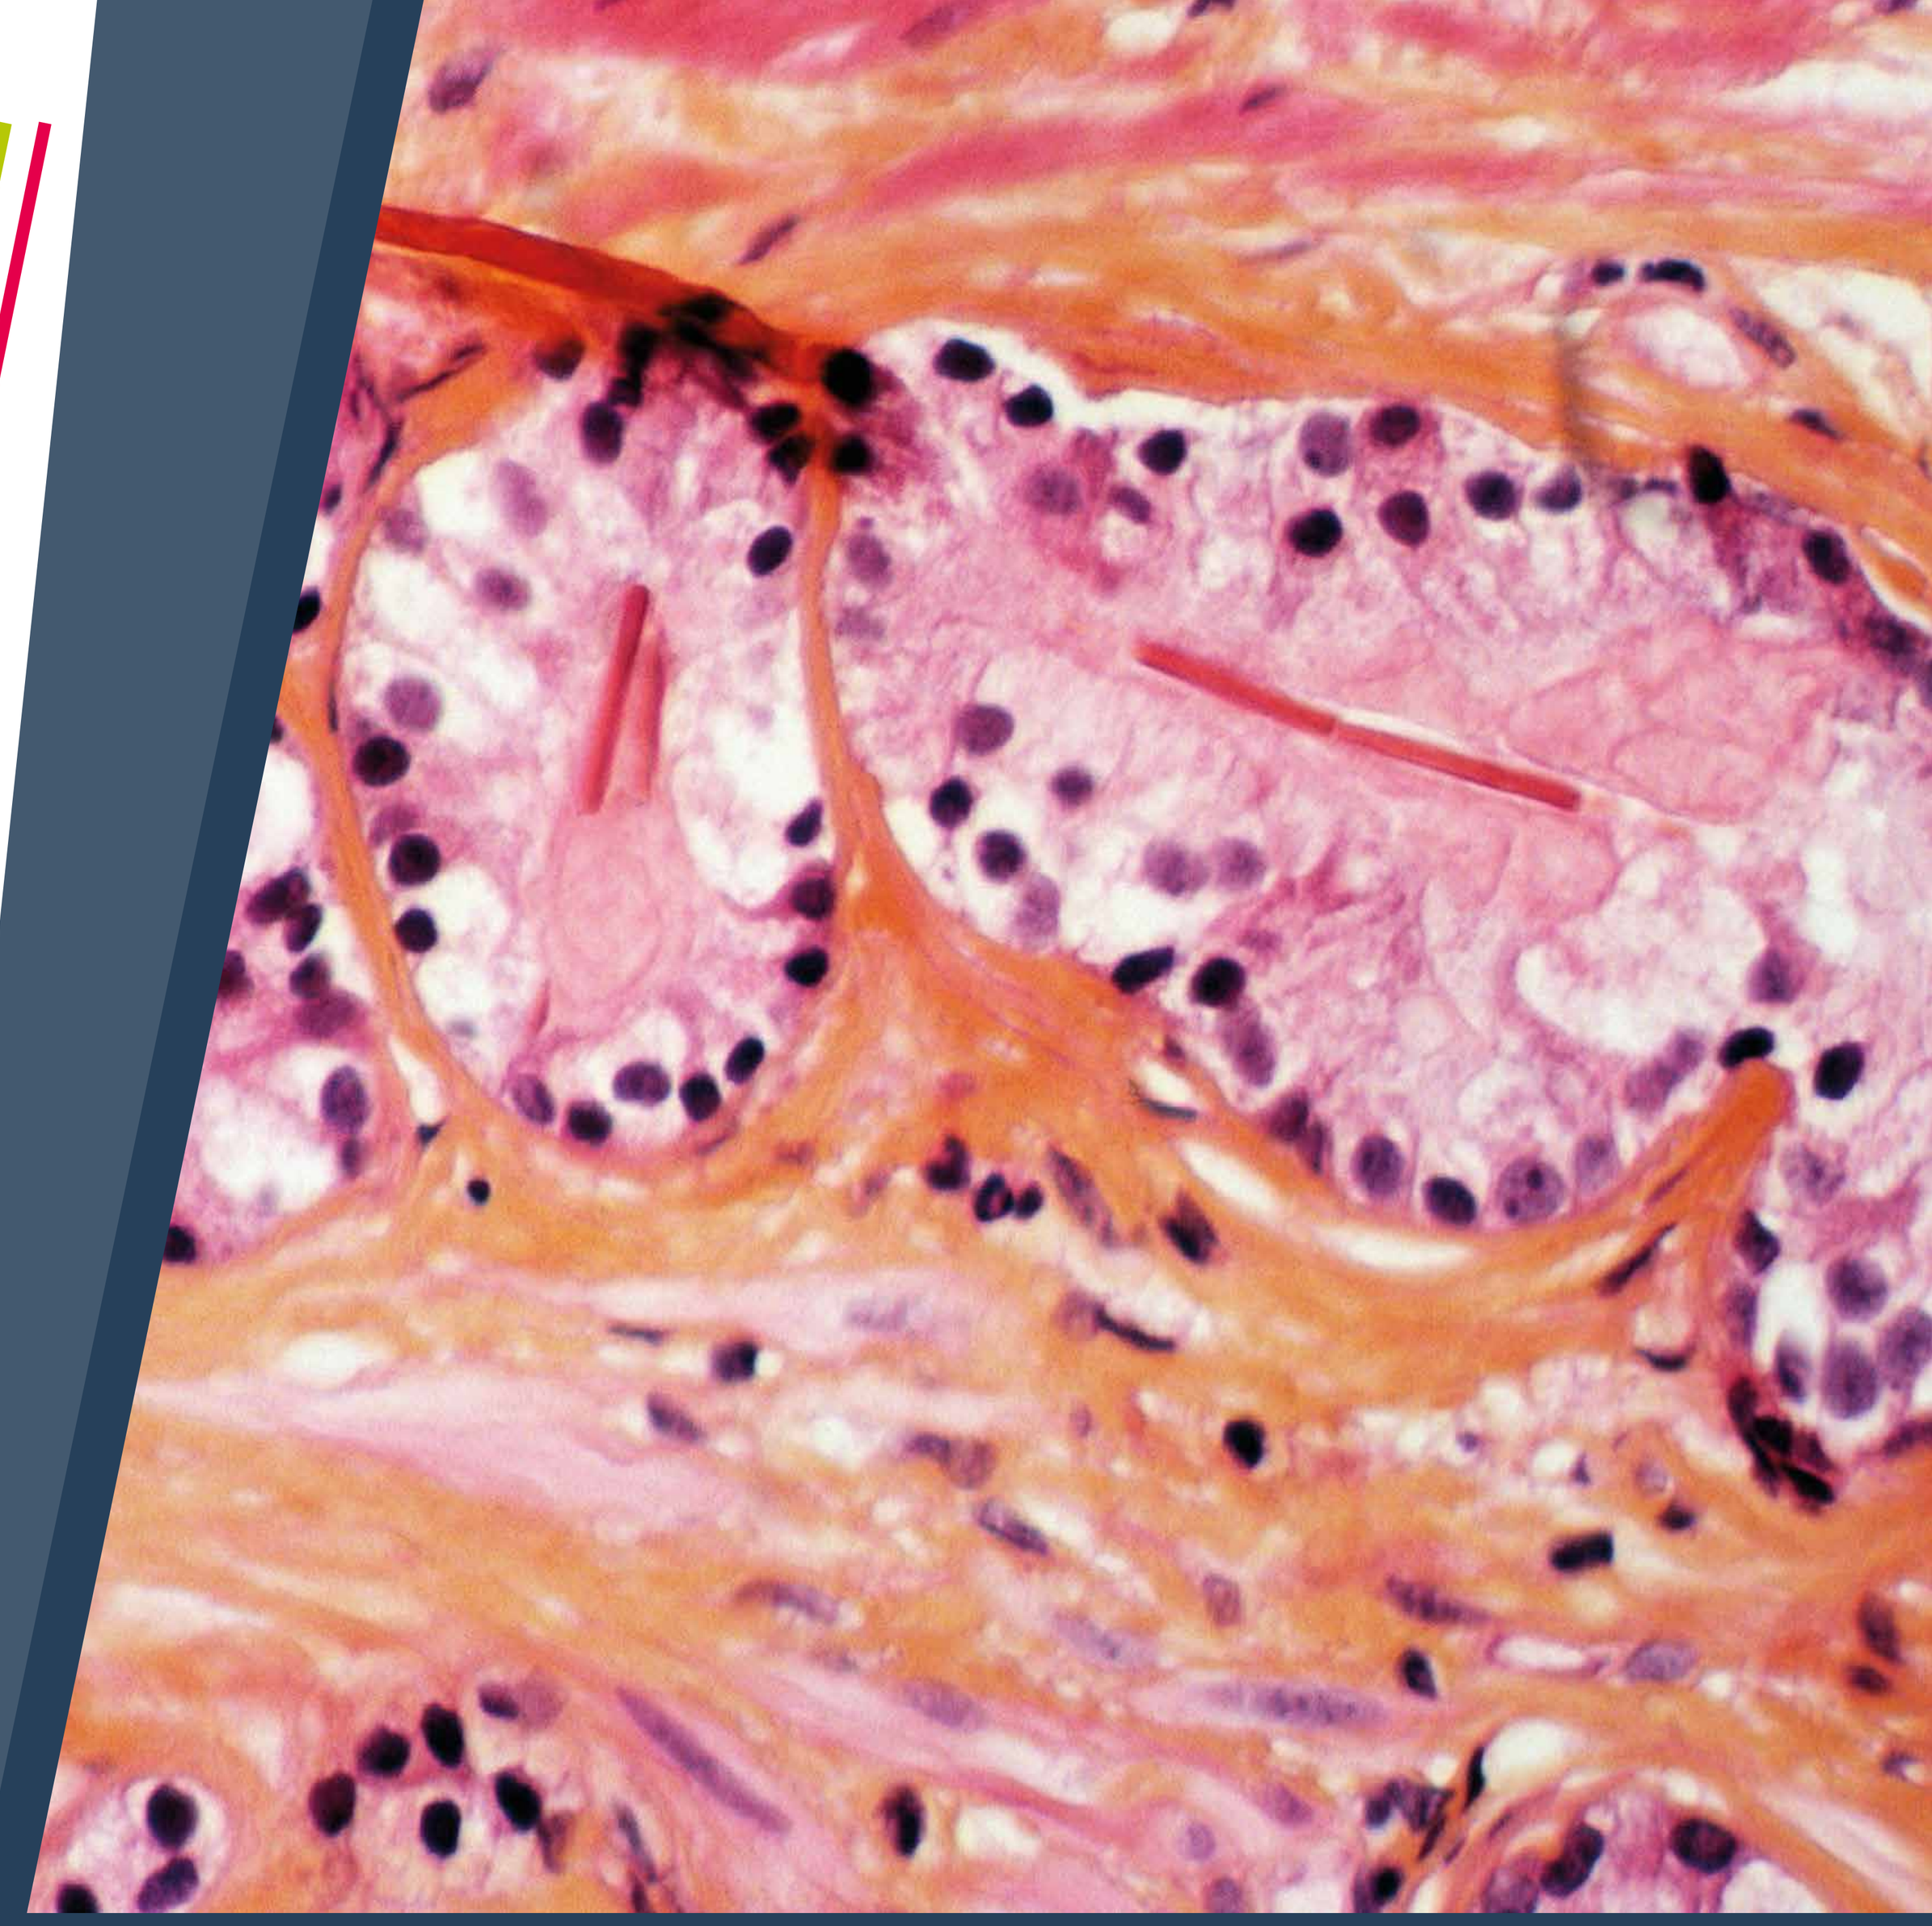

INTRODUCTION

In the phase 3 ARANOTE study (NCT04736199), darolutamide + ADT reduced the risk of radiological progression or death by 46% (HR 0.54; 95% CI 0.41–0.71; *P*<0.0001) versus placebo + ADT in patients with mHSPC[1]

The incidence of treatment-emergent adverse events was low and similar between groups[1]

Patients achieving undetectable PSA (<0.2 ng/mL) at any time had better ECOG performance status, lower Gleason score, and lower baseline PSA values versus patients who did not

METHODS

We report post-hoc analyses of ARANOTE correlating PSA response with outcomes overall and by baseline PSA level

Darolutamide resulted in a deep PSA response

Three times as many patients achieved undetectable PSA (<0.2 ng/mL) at any time vs those on placebo

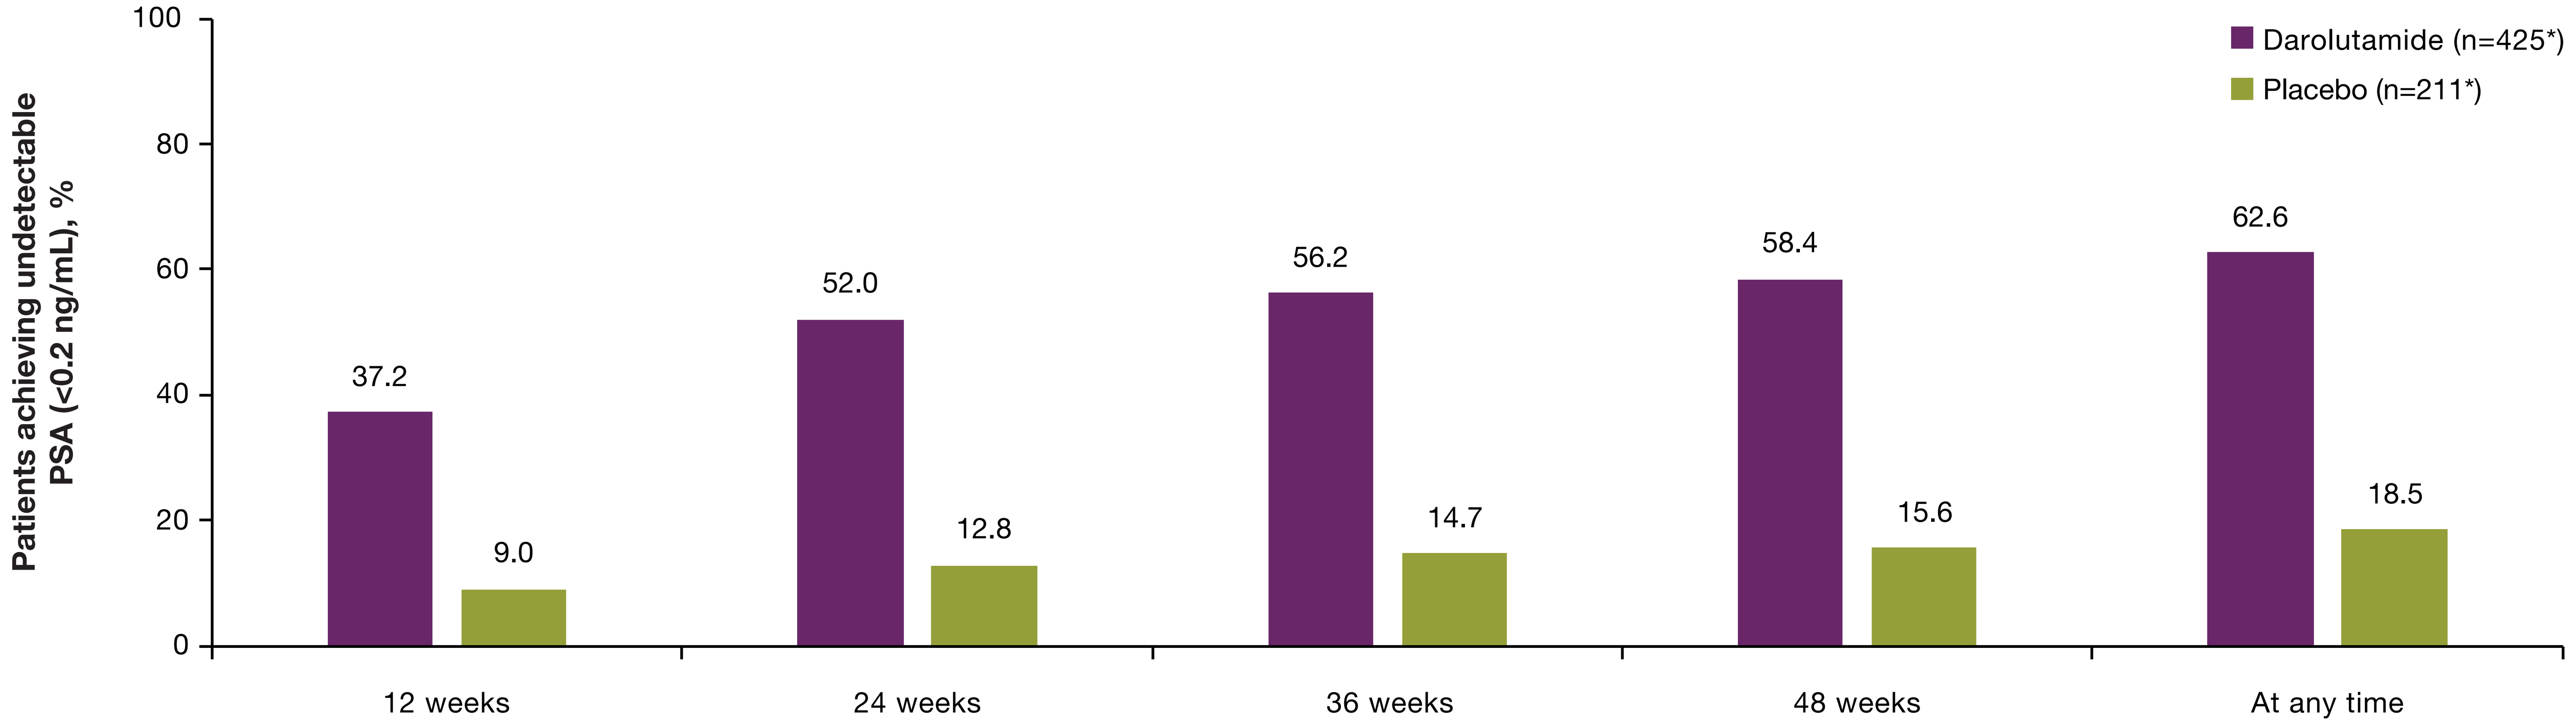
Patients achieving undetectable PSA (<0.2 ng/mL), %
100
80
60
40
20
0
Darolutamide (n=425*)
Placebo (n=211*)
37.2
9.0
52.0
12.8
56.2
14.7
58.4
15.6
62.6
18.5
12 weeks
24 weeks
36 weeks
48 weeks
At any time

*Excludes patients with undetectable PSA (<0.2 ng/mL) at baseline (darolutamide n=11; placebo n=8). PSA, prostate-specific antigen.

Patients on darolutamide who achieved undetectable PSA (<0.2 ng/mL) had a lower risk of radiological progression or death

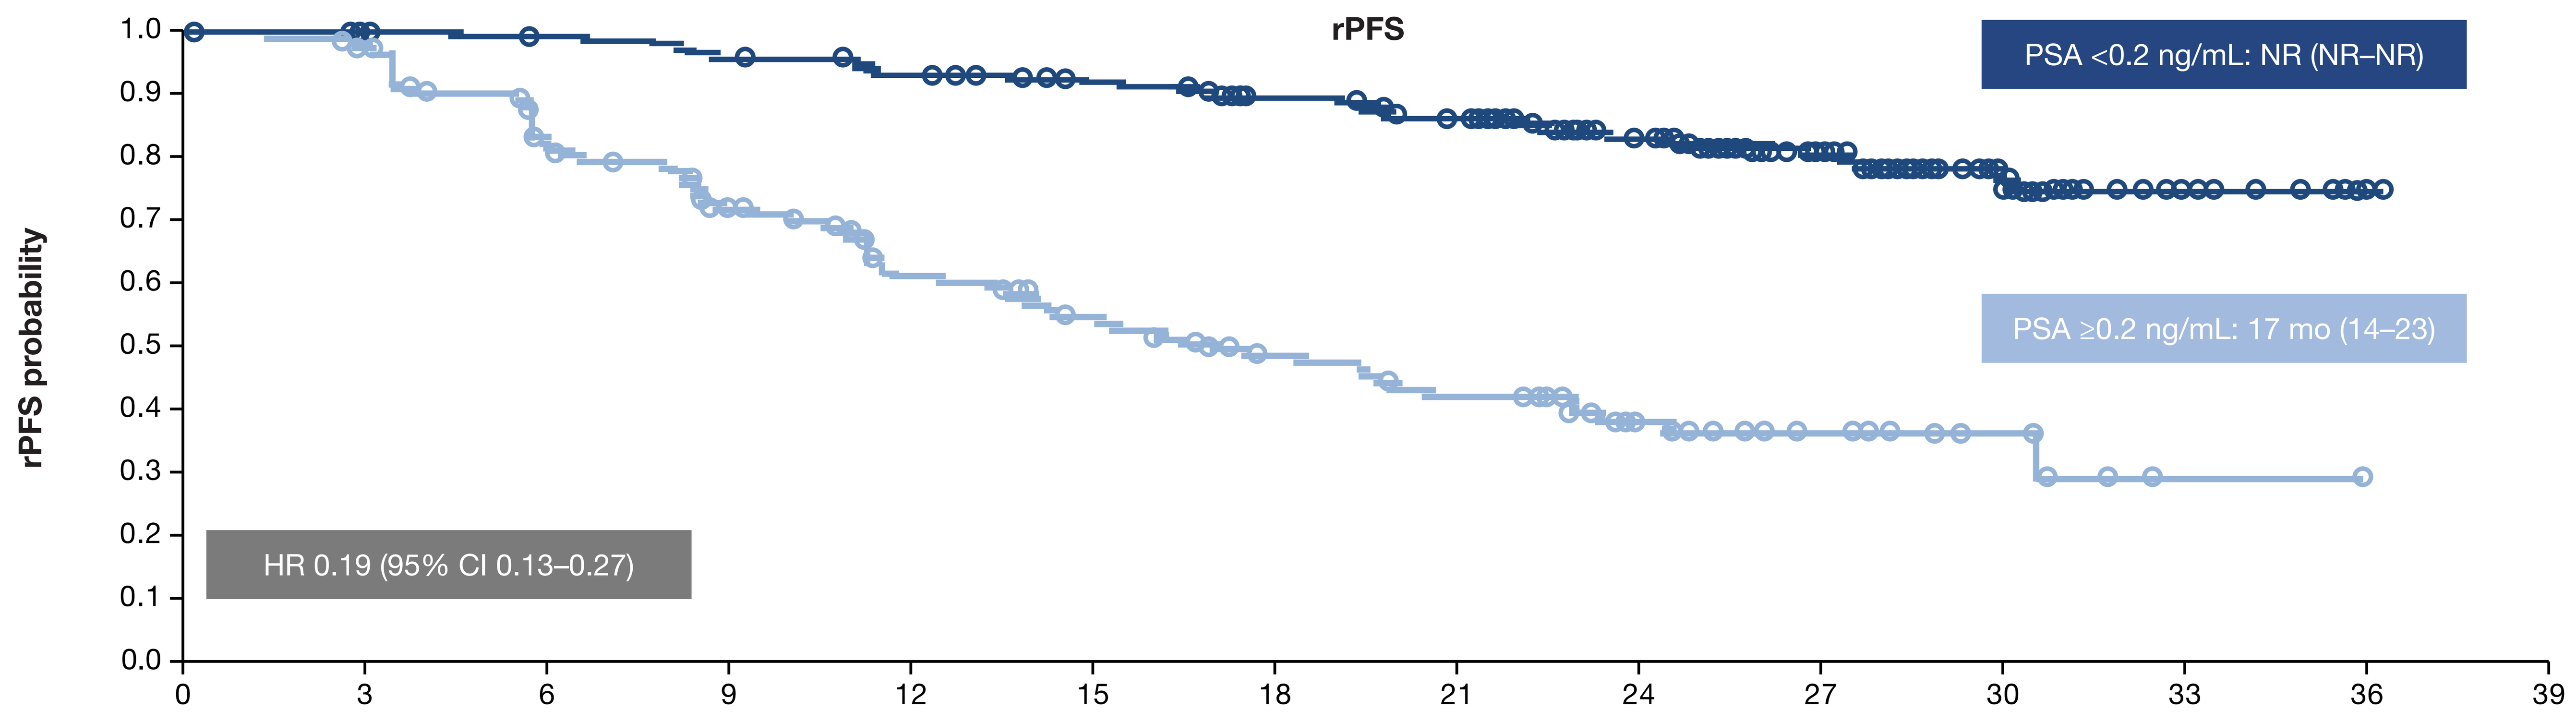
rPFS
rPFS probability
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
PSA <0.2 ng/mL: NR (NR–NR)
PSA ≥0.2 ng/mL: 17 mo (14–23)
HR 0.19 (95% CI 0.13–0.27)
0
3
6
9
12
15
18
21
24
27
30
33
36
39
Time, months

Number of patients at risk

	0	3	6	9	12	15	18	21	24	27	30	33	36	39
PSA <0.2 ng/mL	266	263	260	252	243	234	222	208	147	90	43	8	1	0
PSA ≥0.2 ng/mL	159	138	107	85	67	55	43	36	24	13	6	1	0	0

CI, confidence interval; HR, hazard ratio; mo, months; NR, not reached; PSA, prostate-specific antigen; rPFS, radiological progression-free survival.
Reprinted from Saad F, et al. Eur Urol Oncol. 2025. https://doi.org/10.1016/j.euo.2025.06.010, with permission from Elsevier.

CONCLUSIONS

- Darolutamide provided deep and durable PSA responses in the overall population and across baseline PSA subgroups
- Undetectable PSA with darolutamide correlated with clinical benefit in terms of radiological progression or death and longer times to mCRPC and PSA progression
- The safety profile of darolutamide was consistent across subgroups, regardless of PSA response and baseline PSA

SCAN ME FOR MORE INFO

Reference: 1. Saad F, et al. J Clin Oncol 2024;42(36):4271–81.

ADT, androgen-deprivation therapy; CI, confidence interval; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HR, hazard ratio; mHSPC, metastatic hormone-sensitive prostate cancer; PSA, prostate-specific antigen.

Presented at the National Community Oncology Dispensing Association (NCODA) International Fall Summit 2025, October 16–17, Orlando, FL, USA

Originally presented at the Annual European Association of Urology (EAU) Congress 2025, March 21–24, Madrid, Spain

This study was supported by Bayer AG. Medical writing services were provided by Rachael Powis, PhD, of Adelphi Communications Ltd (Bollington, UK), and Daniel Booth, PhD, on behalf of Adelphi Communications Ltd (Bollington, UK), funded by Bayer, in accordance with Good Publication Practice 2022 guidelines