Real-World Treatment Patterns and Outcomes With Trifluridine/Tipiracil Monotherapy or in Combination With Bevacizumab in Metastatic Colorectal Cancer

Donald Richards,[1] Ruizhi Zhao,[2] Nadeem Khan,[2] Brenda Chang,[3] Siraje Mahmud,[3] Tehseen Salimi[2]

[1]Texas Oncology, Tyler, TX, USA; [2]Taiho Oncology, Inc., Princeton, NJ, USA; [3]Precision Health Informatics, Dallas, TX, USA

OBJECTIVE

- To describe real-world treatment patterns and outcomes in patients with mCRC receiving FTD/TPI±BEV using data from the largest community oncology practice in the US.

CONCLUSIONS

- In this large, real-world, community practice setting in the US, the majority of mCRC patients who were treated with FTD/TPI received this treatment as a third-line therapy and in combination with BEV.
- A statistically significant and clinically relevant OS benefit (median gain of 5.4 months) was seen with the addition of BEV to FTD/TPI vs monotherapy.
- Patient characteristics were similar in the SUNLIGHT trial, with high rates of previous antiangiogenic use (72% in first and/or second line in SUNLIGHT).
- The study suggests that the finding of clinically relevant OS benefit seen with FTD/TPI+BEV vs FTD/TPI monotherapy in the phase 3 SUNLIGHT trial may extend to the US real-world community oncology setting.
- Additional observational studies, preferably using the target-trial emulation framework and control for confounding, are needed to further elucidate the impact on survival of adding BEV to FTD/TPI in mCRC patients in the real world.

Acknowledgments

Medical writing support was provided by Envision Value & Access, a part of Envision Medical Communications, and funded by Taiho Oncology, Inc.

Author disclosures

This study was funded by Taiho Oncology, Inc.
DR, BC, and SM are employees of Texas Oncology and Precision Health Informatics, paid consultants to Taiho Oncology, Inc. in connection with this study. RZ, NK, and TS are employees of Taiho Oncology, Inc.

This poster was previously presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, IL, May 30–June 3, 2025

Corresponding author

Donald Richards (donald.richards@usoncology.com)

Please scan this Quick Response (QR) code with your smartphone app to view an electronic version of this poster. Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from NCODA® or the authors of this poster.

Poster presented at the Network for Collaborative Oncology Development & Advancement (NCODA) Fall Summit, Orlando, FL, October 15-17, 2025

© Copyright 2025

Background

- Trifluridine/tipiracil (FTD/TPI; Lonsurf®) is an oral antineoplastic agent approved for third-line use in combination with or without bevacizumab (BEV) in metastatic colorectal cancer (mCRC).[1]
- In the Phase 3 SUNLIGHT trial, the addition of BEV to FTD/TPI was associated with a significant improvement in overall survival (OS) compared with FTD/TPI monotherapy.[2]
- In the US, most cancer patients are treated in community-based settings;[3-5] therefore, understanding of patients' treatment patterns and outcomes in community oncology practice can provide crucial insights into real-world delivery, practice decisions, and effectiveness.
- Real-world treatment patterns and outcomes associated with the use of FTD/TPI with or without BEV in the community setting have not been previously studied.

Methods

Study design

- Retrospective observational study using electronic medical records (EMRs) and chart reviews from mCRC patients treated by the Texas Oncology community practice from January 2020 to October 2024.
- Index date was defined as the date of initiation of FTD/TPI therapy.
- A baseline period of 6 months prior to the index date was used to characterize the study population.
- Patients were followed from index date until death, last clinic visit, or end of study period, whichever occurred first.

Data source

- EMRs were obtained from 2 electronic health record systems: (1) iKnowMed Generation 1 and (2) iKnowMed Generation 2 Practice Demographics database.
- All abstracted data were retrospective, and patients were not followed prospectively or contacted to provide additional information.
- Data were extracted from the structured fields of EMRs in participating practices and, when available, via abstraction of patient records.

Patients and cohorts

- For inclusion, patients had to meet the following criteria:
 - Diagnosis of mCRC and receipt of a line of therapy with oxaliplatin and irinotecan from January 2020 to October 2024.
 - Disease progression on a prior line of therapy with oxaliplatin and irinotecan.
 - Age ≥18 years on the index date.
 - Treatment with FTD/TPI as monotherapy or in combination with BEV from January 2020 to October 2024.
- Patients were excluded if they had evidence of clinical trial enrollment.
- Two subcohorts were defined as (1) patients receiving FTD/TPI monotherapy and (2) patients receiving FTD/TPI+BEV, as determined by their first FTD/TPI treatment mode.
 - For combination therapy, BEV use occurred within 5 weeks of FTD/TPI use.

Outcomes and statistical analyses

- Variables included patients' demographic and clinical characteristics, treatment patterns (including previous lines of therapy), and clinical outcomes, such as OS and time to next treatment or death (TTNTD).
- Time to next treatment was defined as the time interval between index date and initiation of the next line of therapy.
- Symptoms and adverse events reported during follow-up were recorded.
- Continuous variables were described using means, SD, medians, and ranges (min and max), while frequencies and percentages were used for categorical and ordinal variables.
- Time to death (OS) and TTNTD were analyzed using the Kaplan-Meier method with 95% CI.
 - The log-rank test was used to compare groups, along with unadjusted hazard ratios (HR) and associated 95% CIs.
- For OS, patients still alive at the end of follow-up/study end date were censored on the date of last encounter.

Results

Study population and patient characteristics

- A total of 265 patients were included, with the majority (63%) of patients receiving the combination therapy (166 FTD/TPI+BEV, 99 FTD/TPI monotherapy).
- The population was 59% male and 66% white, and mean age was 61 years (35% ≥65), with no notable differences between those receiving FTD/TPI+BEV combination therapy and those on monotherapy (**Table 1**).
- Most patients (89.1%) had Eastern Cooperative Oncology Group (ECOG) performance status scores of 0 or 1.
- Geographical distribution of treatment location around Texas and type of insurance were similar irrespective of BEV use.
- Most patients (87.5%) had Commercial/Medicare Advantage insurance.

Treatment patterns

- The majority of patients received FTD/TPI±BEV as third-line (83%; n=220) or fourth-line (14%; n=38) therapy.
- The most common previous first- and second-line treatment for third-line FTD/TPI patients overall was chemotherapy + an antiangiogenic (first line, 67%; second line, 74%), which was similar regardless of current BEV use.

Table 1. Patient demographics

Demographic	FTD/TPI+BEV (N=166)	FTD/TPI monotherapy (N=99)
Age, mean (SD), years	60.8 (9.8)	61.2 (10.5)
Age ≥65, n (%), years	58 (34.9)	35 (35.4)
Sex, n (%) male	102 (61.4)	55 (55.6)
Race, n (%)		
White	114 (68.7)	62 (62.6)
Black/African American	15 (9.0)	8 (8.1)
Asian	5 (3.0)	0 (0)
Other	30 (18.1)	26 (26.3)
Not reported	2 (1.2)	3 (3.0)
Ethnicity, n (%)		
Hispanic/Latino	46 (27.7)	21 (21.2)
Not Hispanic/Latino	112 (67.5)	71 (71.7)
Not reported	8 (4.8)	7 (7.1)
Location, n (%)		
Dallas–Fort Worth	47 (28.3)	35 (35.4)
Gulf Coast Texas	30 (18.1)	10 (10.1)
Central Texas	26 (15.7)	13 (13.1)
West Texas	24 (14.5)	13 (13.1)
South Texas	22 (13.3)	14 (14.1)
Northeast Texas	17 (10.2)	14 (14.1)

BEV, bevacizumab; FTD/TPI, trifluridine/tipiracil.

Clinical outcomes

- Median OS was 11.6 months with FTD/TPI+BEV and 6.2 months with monotherapy (HR=2.1; 95% CI: 1.5-3.0; *P*<0.001) (**Figure 1**).
- At 6 months, the probability of survival was 0.69 (95% CI: 0.61-0.77) with FTD/TPI+BEV and 0.50 (95% CI: 0.40-0.63) with monotherapy; 12-month survival probability was 0.49 (0.39-0.61) and 0.15 (0.07-0.28), respectively.
- Median TTNTD was 9.4 months for FTD/TPI+BEV and 5.8 months for FTD/TPI alone (HR=1.7; 95% CI, 1.2-2.4; *P*<0.001) (**Figure 2**).
- At 6 months, the probability of remaining free from next treatment or death (event-free probability) was 0.66 (95% CI: 0.58-0.75) with FTD/TPI+BEV and 0.48 (95% CI: 0.38-0.61) with monotherapy; 12-month event-free probability was 0.40 (0.28-0.56) and 0.08 (0.03-0.22), respectively.

Clinical symptoms/adverse events reported during follow-up

- The most commonly observed adverse events were fatigue/asthenia (73%), abdominal discomfort/pain (55%), and nausea (54%) (**Table 2**).
- Some of the notable differences between FTD/TPI+BEV and monotherapy were:
 - neutropenia (37% FTD/TPI+BEV, 27% monotherapy).
 - anemia (34% FTD/TPI+BEV, 26% monotherapy).
 - diarrhea (48% FTD/TPI+BEV, 59% monotherapy).
 - weight loss (45% FTD/TPI+BEV, 24% monotherapy).

Table 2. Clinical symptoms/adverse events reported during follow-up

Parameter	FTD/TPI+BEV (N=166)	FTD/TPI monotherapy (N=99)
Symptom/adverse event, n (%)		
Fatigue/asthenia	124 (74.7)	70 (70.7)
Abdominal discomfort/pain	94 (56.6)	51 (51.5)
Nausea	88 (53.0)	56 (56.6)
Diarrhea	80 (48.2)	58 (58.6)
Constipation/intestinal obstruction	76 (45.8)	50 (50.5)
Weight loss	75 (45.2)	24 (24.2)
Loss of appetite/decreased appetite	59 (35.5)	28 (28.3)
Back pain	37 (22.3)	31 (31.3)
Clinical abnormalities (top 3), n (%)		
Neutropenia	62 (37.3)	27 (27.3)
Anemia	43 (25.9)	34 (34.3)
Infection	3 (1.8)	3 (3.0)

Figure 1. Overall survival with FTD/TPI+BEV vs FTD/TPI monotherapy

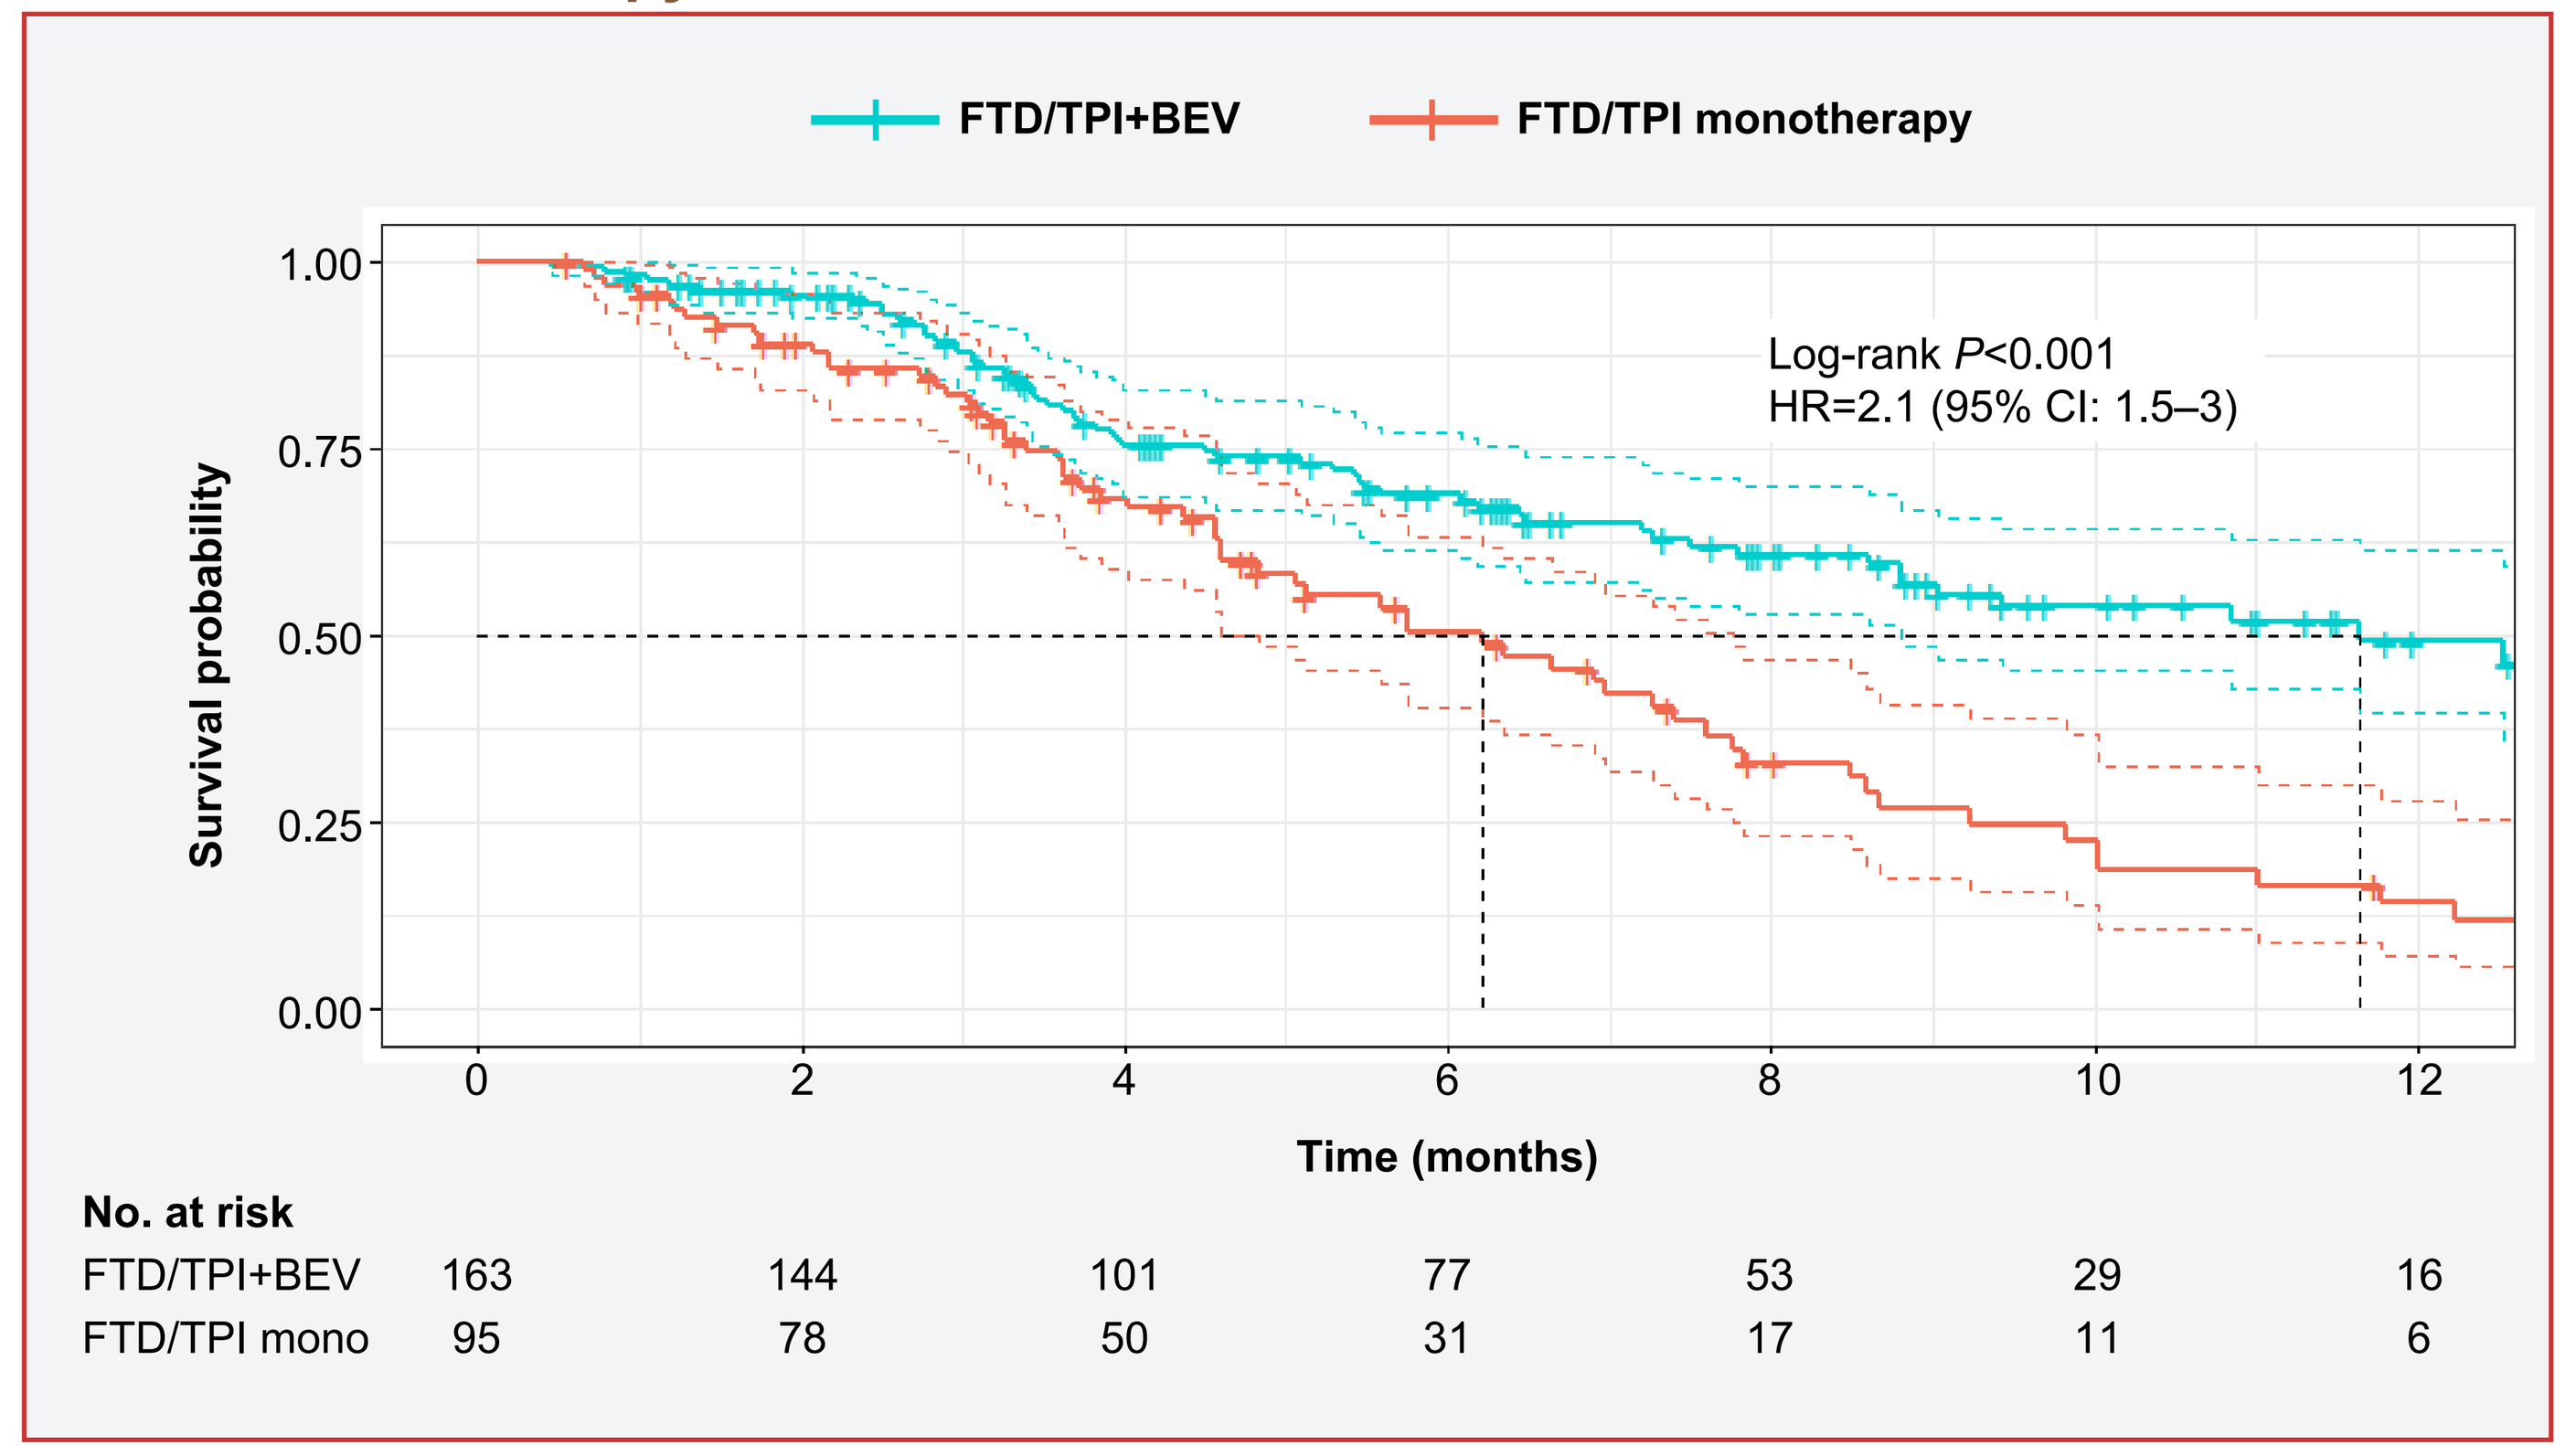
FTD/TPI+BEV
FTD/TPI monotherapy
Log-rank P<0.001
HR=2.1 (95% CI: 1.5–3)
Survival probability
1.00
0.75
0.50
0.25
0.00
0
2
4
6
8
10
12
Time (months)
No. at risk
FTD/TPI+BEV 163 144 101 77 53 29 16
FTD/TPI mono 95 78 50 31 17 11 6

Patients who died within 2 weeks from the start of FTD/TPI were excluded.
BEV, bevacizumab; FTD/TPI, trifluridine and tipiracil.

Figure 2. Time to next treatment or death with FTD/TPI+BEV vs FTD/TPI monotherapy

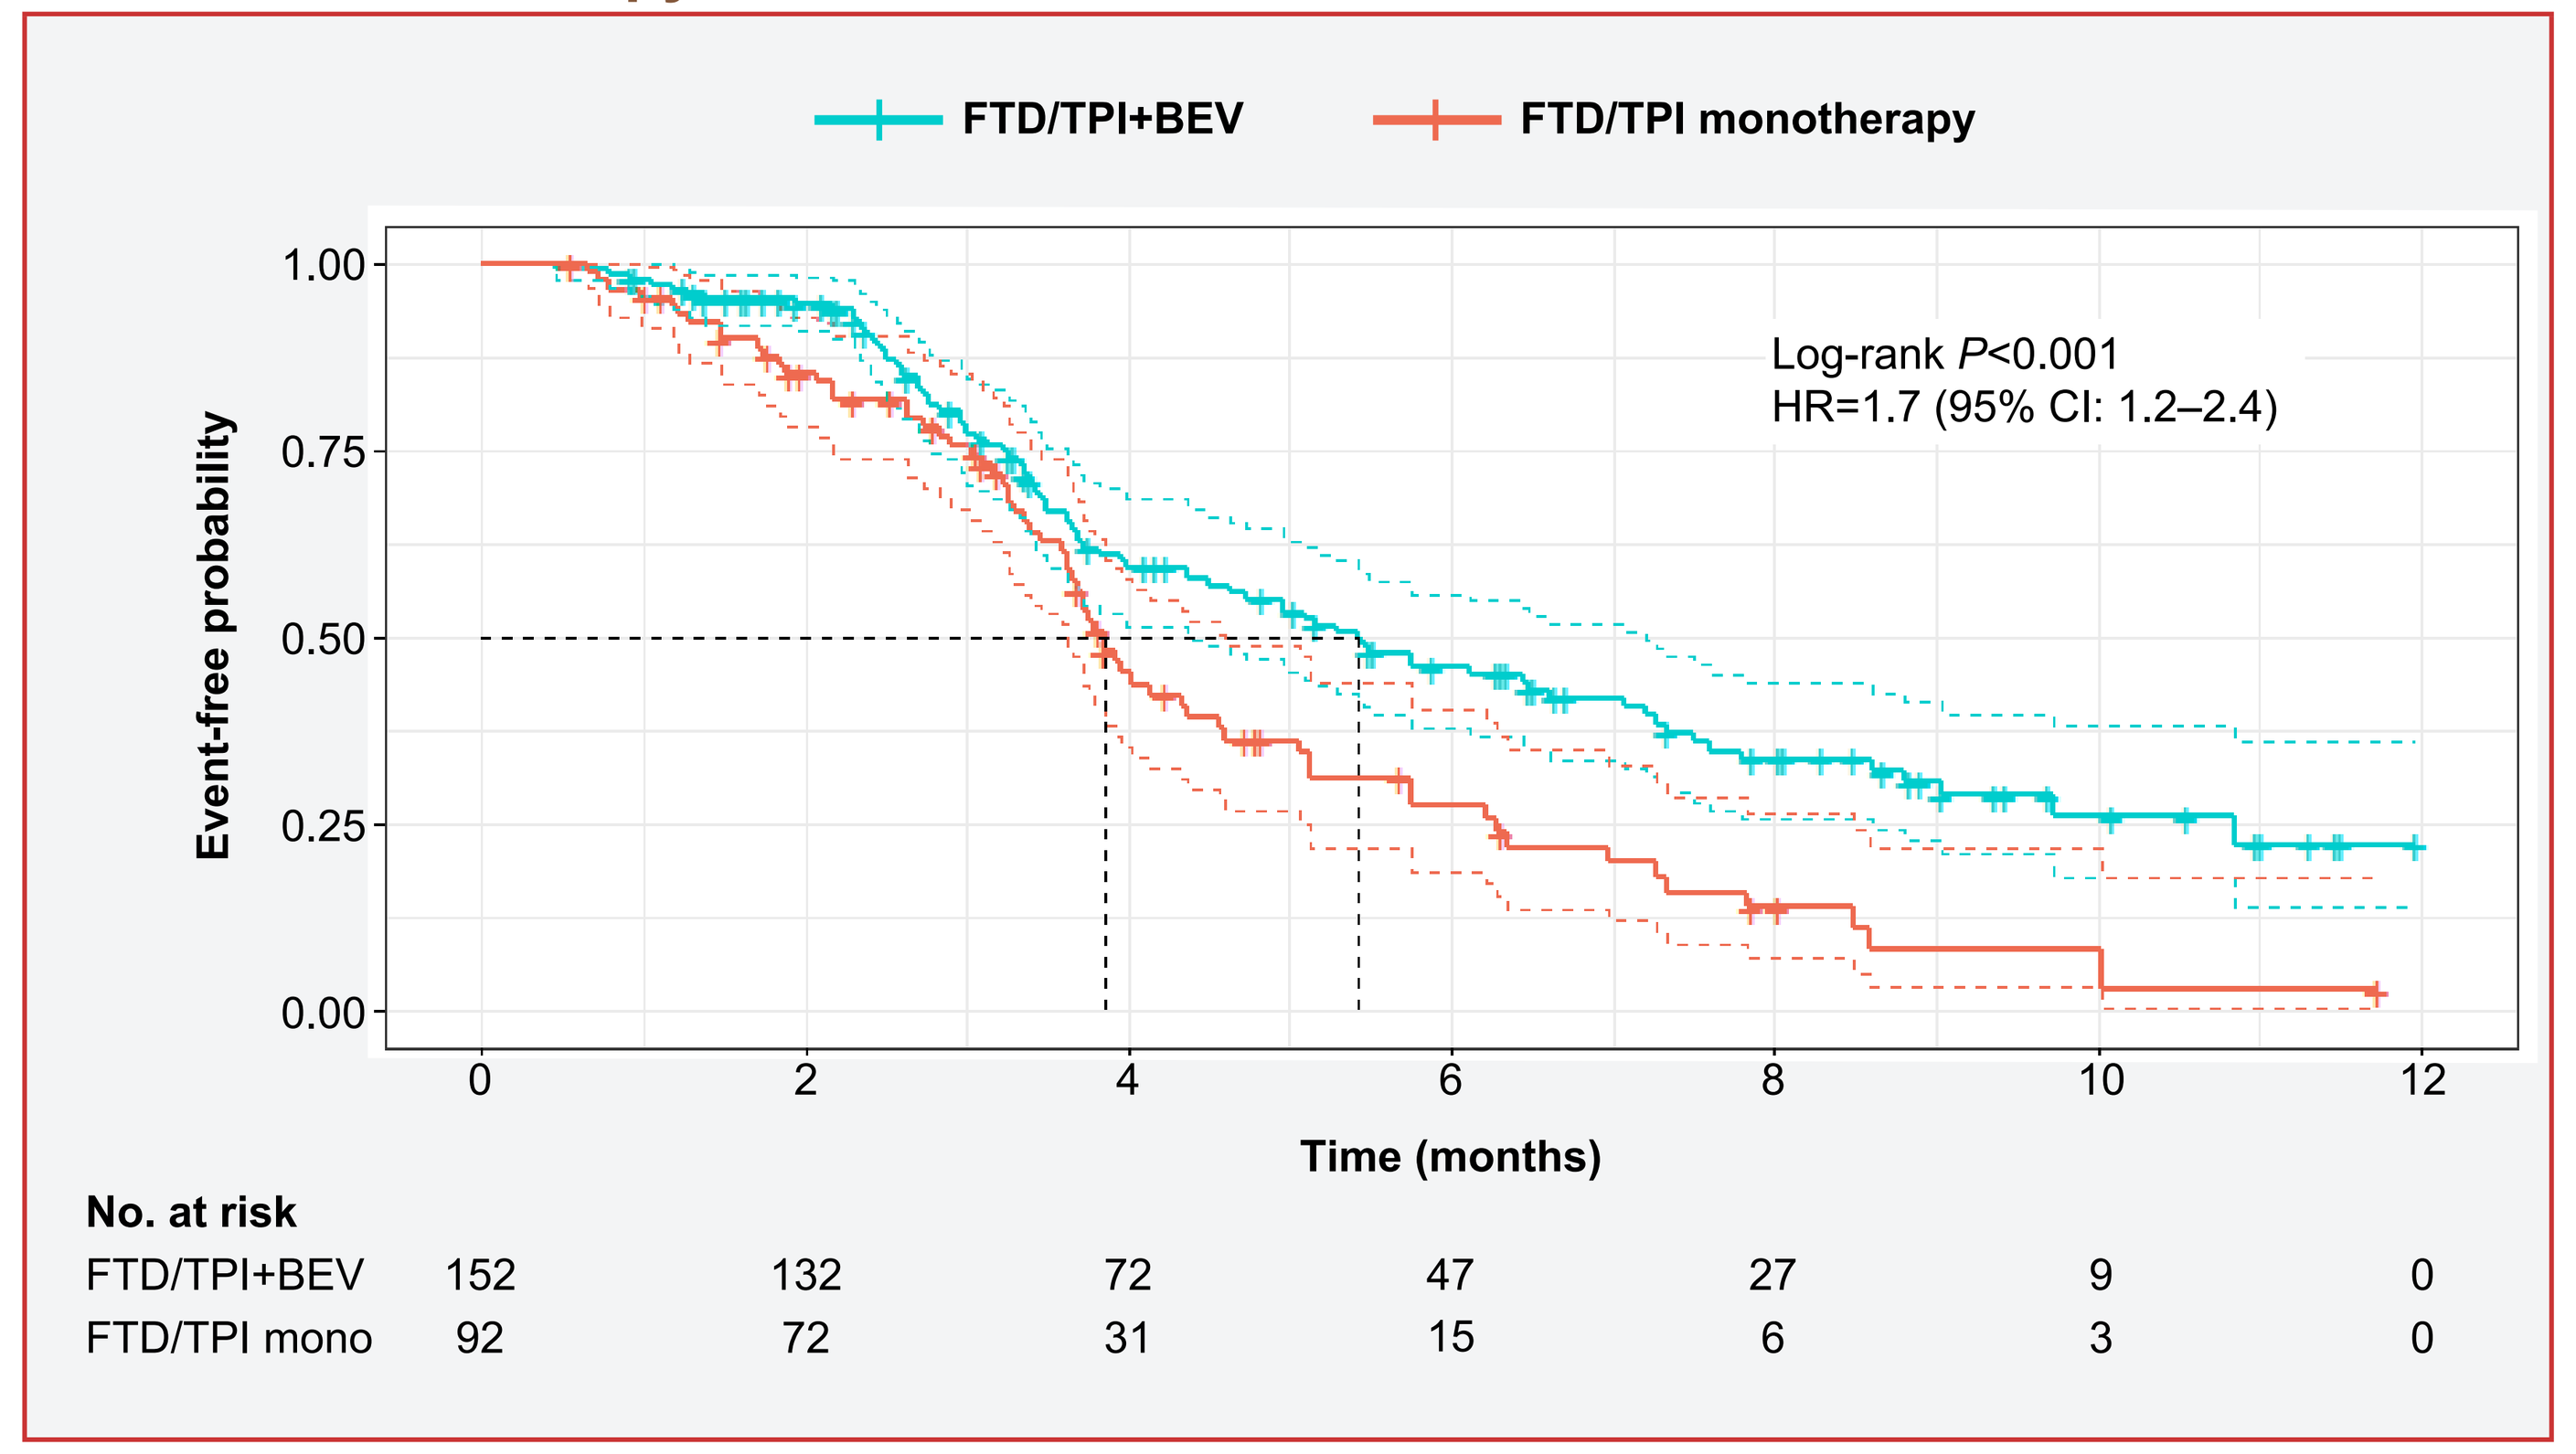
FTD/TPI+BEV
FTD/TPI monotherapy
Log-rank P<0.001
HR=1.7 (95% CI: 1.2–2.4)
Event-free probability
1.00
0.75
0.50
0.25
0.00
0
2
4
6
8
10
12
Time (months)
No. at risk
FTD/TPI+BEV 152 132 72 47 27 9 0
FTD/TPI mono 92 72 31 15 6 3 0

Patients who died within 2 weeks from the start of FTD/TPI were excluded.
BEV, bevacizumab; FTD/TPI, trifluridine and tipiracil.

Limitations

- This study was observational and descriptive in nature and is subject to inherent limitations; hence, the results should be interpreted with caution.
- Detailed clinical characteristics of patients could not be fully captured if access to chart data was not available.
- Grade of adverse events/clinical abnormalities were not captured in this database.
- Duration of treatment may have been incomplete for patients who were lost to follow-up if they continued treatment outside of Texas Oncology.

References

1. LONSURF (trifluridine and tipiracil). Prescribing Information. Taiho Oncology, Inc. 2023. Accessed August 2023.
2. Prager GW, et al. *N Engl J Med*. 2023;388(18):1657-1667.
3. Garg AK. *Am J Clin Oncol*. 2020;43(8):537-538.
4. Community Oncology Alliance FACT Sheet. 2017. Available at: https://communityoncology.org/wp-content/uploads/2017/08/What-is-Comm-Onc.pdf
5. Unger JM, et al. *J Natl Cancer Inst*. 2019;111(3):245-255.