Overall survival in patients with HR+/HER2- advanced breast cancer treated in a phase 1b trial evaluating gedatolisib in combination with palbociclib and endocrine therapy

Hope S. Rugo[1], Robert Wesolowski[2], Hyo Han[3], Jennifer M. Specht[4], E. Claire Dees[5], Erica M. Stringer-Reasor[6], Peter Kabos[7], Vandana Abramson[8], Amy Weise[9], Anne Schott[10], Mayya Abu-Khalaf[11], Keerthi Gogineni[12], Alexander Spira[13], Charlotte Moser[14], Nadene Zack[14], Samuel Suzuki[14], Sarah Mutka[14], Brian Sullivan[14], Igor Gorbatchevsky[14], Rachel M. Layman[15]

[1]City of Hope Comprehensive Cancer Center, Duarte, CA; [2]The Ohio State University Comprehensive Cancer Center, Columbus, OH; [3]Moffitt Cancer Center, Tampa, FL; [4]Fred Hutchinson Cancer Center, University of Washington, Seattle, WA; [5]University of North Carolina Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [6]O'Neal Comprehensive Cancer Center, University of Alabama at Birmingham, Birmingham, AL; [7]University of Colorado Hospital, Aurora, CO; [8]Vanderbilt University Medical Center, Nashville, TN; [9]Henry Ford Cancer Institute, Detroit, MI; [10]University of Michigan, Ann Arbor, MI; [11]Thomas Jefferson University, Philadelphia, PA; [12]Emory University Hospital, Atlanta, GA; [13]Virginia Cancer Specialists, Fairfax, VA; [14]Celcuity, Minneapolis, MN; [15]The University of Texas MD Anderson Cancer Center, Houston, TX

Presented at
NCODA's 2025 Fall Summit

*Previously presented at the
2024 San Antonio
Breast Cancer Symposium*

BACKGROUND

- The estrogen receptor (ER), CDK4/6, and PI3K/AKT/mTOR (PAM) pathways each promote tumor cell proliferation in HR+, HER2-advanced breast cancer (ABC). [1,2]
- This led to the approval of therapeutic regimens that include various combinations of endocrine therapies (ET), CDK4/6 inhibitors (CDK4/6i) and PAM inhibitors that target a single node of the PI3K/AKT/mTOR pathway for patients with HR+, HER2- ABC. [3-7]
- There is thus a strong scientific rationale to simultaneously blockade the ET, CDK4/6, and PAM pathways in both the first and second line setting to optimize anti-tumor control.
- To evaluate this hypothesis, a Phase Ib study (B2151009) of gedatolisib, a PAM (pan-PI3K and mTORC1/2) inhibitor, palbociclib, a CDK4/6 inhibitor, and ET (letrozole or fulvestrant) in women with HR+/HER2- ABC was conducted. [8]
- In this analysis, we report overall survival data for two patient subgroups evaluated in the B2151009 study:
 - Patients who had prior treatment with a CDK4/6i and fulvestrant and received gedatolisib using the intermittent schedule being studied in an ongoing Phase 3 clinical trial (VIKTORIA-1, NCT05501886).
 - Patients who were treatment naïve in the metastatic setting and were eligible for treatment with a CDK4/6i and letrozole.
- As previously reported, ORR and mPFS for both subgroups of patients compare favorably to published data for current standard of care regimens. Additionally, as previously reported, responses were observed in patients regardless of PIK3CA mutation status and few patients (<10%) discontinued treatment due to an adverse event [8,9]

Figure 1. The ER, CDK4/6, and PAM Pathways Are Interdependent Drivers of HR+/HER2- ABC

Dysregulation of these pathways promotes excessive cell proliferation and resistance to apoptosis

ER and the PAM Pathway

- Activation of the PAM pathway induces estrogen independent ER transcriptional activity by mTOR
- Conversely, ER target gene expression activates upstream effectors of the PI3K/mTOR pathway
- ER also activates the PAM pathway by direct binding to PI3Kα
- **PAM pathway inhibition increases ER activity which increases sensitivity to endocrine therapy**

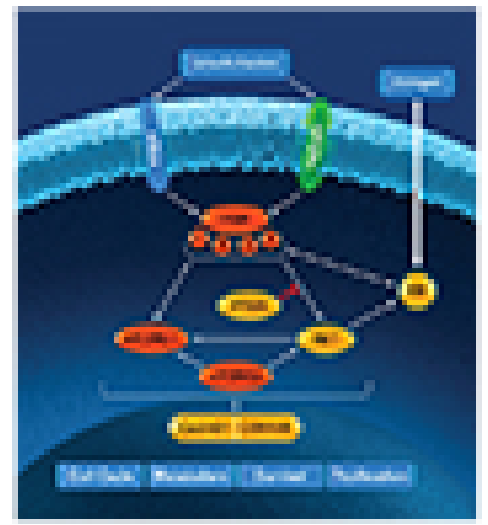

CDK4/6, ER and the PAM Pathway

- Estrogen promotes cyclin D1 transcription and cyclin D1 can cause estrogen independent transcription
- Provides rationale for simultaneously inhibiting ER and CDK4/6
- CDK4/6 inhibition causes incomplete cell cycle arrest – addition of PAM pathway inhibition enables more complete arrest
- **PAM pathway inhibition increases cyclin D1 activity which increases sensitivity to CDK4/6 inhibition**

STUDY DESIGN

1st Line Treatment-Naïve (N=41)	2L/3L CDK4/6i-treated (N=27)
combined subgroup from escalation and expansion arms	
palbociclib + letrozole + gedatolisib (weekly)	palbociclib + fulvestrant + gedatolisib (3 weeks on/1 week off)

- Dosing information: Palbociclib: 125 mg/day in a 3 week on, 1 week off schedule; Letrozole: 2.5 mg/day; Fulvestrant: 500 mg intramuscular injection on cycle 1 days 1 and 15, and every 28 days ± 3 days thereafter; Gedatolisib: 180 mg IV weekly or three weeks on/one week off.
- Tumor assessment performed at baseline and every 8 weeks for at least the first 18 months of treatment until disease progression or the start of a new anti-cancer therapy.
- Endpoints: Primary – objective response per RECIST v1.1 assessed by the investigator; Secondary – safety, duration of response (DOR), and PFS.

RESULTS

Table 1: Baseline Characteristics

Parameters	1L Line Treatment-Naïve (N=41)	2L/3L CDK4/6i-Treated (N=27)
Age, years		
Median (range)	54 (28–73)	59 (34-79)
Prior Therapies – Advanced Breast Cancer, n (%)		
Prior Chemotherapy	NA	5 (19)
Prior SERD or SERM Therapy	NA	10 (37)
Prior Aromatase Inhibitor Therapy	NA	19 (70)
Prior CDK4/6 inhibitor	NA	26 (96)
Number of Prior Therapies – Advanced Breast Cancer, n (%)		
0	41 (100)	0
1	0	18 (67)
2	0	8 (30)
≥3	0	1 (4)
Measurable Baseline Disease, n (%)		
Yes	38 (93)	27 (100)
No	3 (7)	0
Disease Site Involved, n (%)		
Bone Only	1 (2)	0
Bone	26 (63)	18 (67)
Brain	0	0
Liver	15 (37)	17(63)
Lung	7 (17)	6 (22)
Lymph Node	12 (29)	2 (7)
Pleural Effusion	4 (10)	2 (7)
Skin	1 (2)	0
Other	35 (85)	21 (78)
Number of Disease Sites Involved, n (%)		
1-3	35 (85)	24 (89)
≥4	6 (15)	3 (11)
***PIK3CA*, n (%)ᵃ**		
Wild-type	31 (76)	15 (56)
Mutant	9 (22)	11 (41)
Unknown/Missing	1 (2)	1 (4)

ᵃPIK3CA status confirmed by liquid biopsy using a central lab.
NA, not available; SERD, selective estrogen receptor degrader; SERM, selective estrogen receptor modulator.

Figure 2. Overall Survival 1L Treatment-Naive, N=41

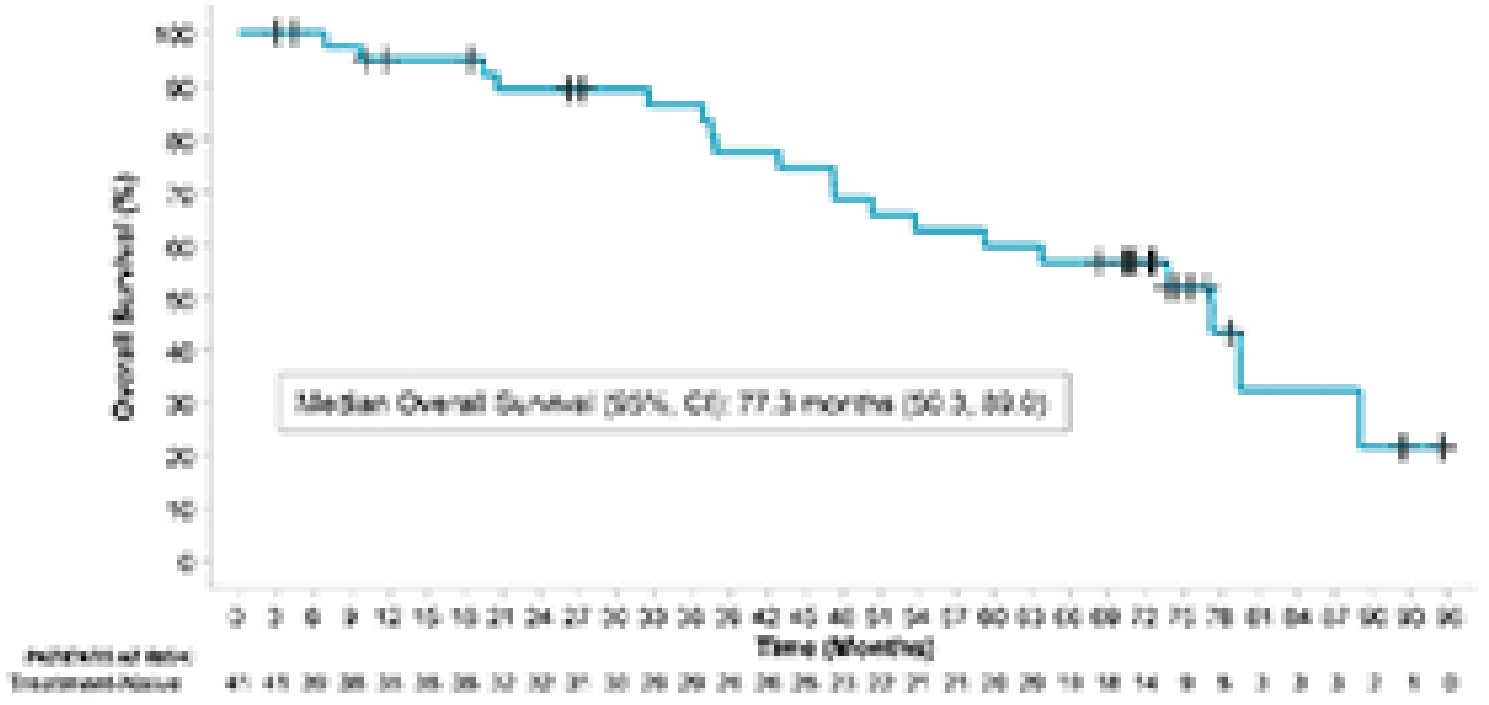
Overall Survival (%)
Median Overall Survival (95%, CI): 77.3 months (50.3, 89.0)
Time (Months)
Patients at Risk:
Treatment-Naive

Figure 3. Overall Survival 2L/3L CDK4/6I-Treated (N=27)

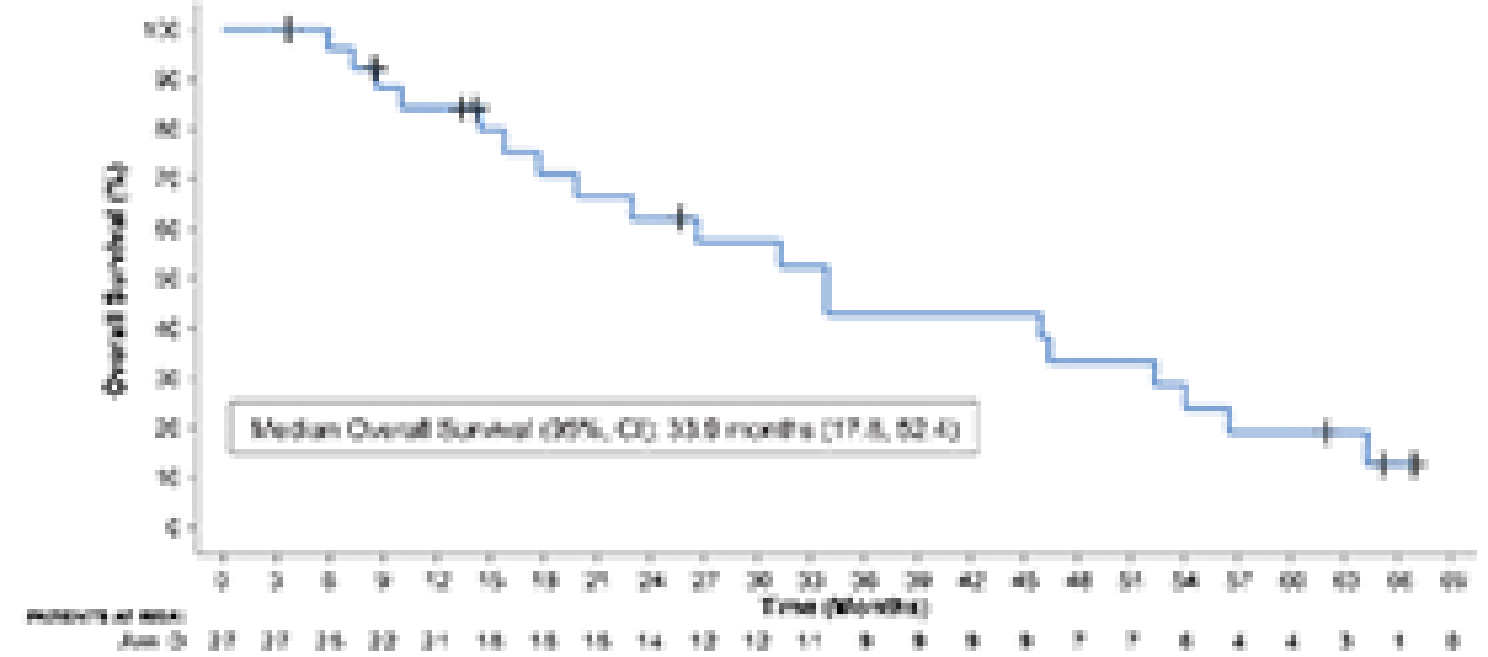
Overall Survival (%)
Median Overall Survival (95%, CI): 33.9 months (17.8, 52.4)
Time (Months)
Patients at Risk:

CONCLUSIONS

- Gedatolisib in combination with palbociclib and ET demonstrated encouraging preliminary efficacy in patients with HR+/HER2- ABC. mPFS was 48.4 months in the treatment-naïve subgroup and 12.9 months in the subgroup previously treated with a CDK4/6i who received gedatolisib on an intermittent schedule. [8,9]
- A subsequent analysis of these patients after additional follow-up showed encouraging OS results that compare favorably to data published for currently available standard-of-care regimens.
 - Median OS for the treatment-naïve subgroup was 77.3 months.
 - Median OS for the 2/3L CDK4/6i-treated patients who received gedatolisib on an intermittent schedule was 33.9 months.
- Combination therapy was well tolerated with few patients discontinuing treatment due to an adverse event. [6,7]
- These encouraging results support further evaluation of this combination therapy in patients with HR+/HER2- ABC.
 - VIKTORIA-1 (NCT05501886), an ongoing randomized Phase 3 study, is evaluating patients who previously received a CDK4/6i.
 - A randomized Phase 3 study evaluating treatment naïve patients (VIKTORIA-2) is currently enrolling.

REFERENCES:
[1] Alves R, et al. Int J Mol Sci. 2023;24(5):4522.
[2] Dinegar A, et al. Int J Mol Sci. 2023;25(12):5844.
[3] Turner NC, et al. N Engl J Med. 2015;373(3):209-219.
[4] André F, et al. N Engl J Med. 2019;380(20):1929-1940.
[5] Rugo HS, et al. J Clin Oncol. 2020;38(suppl 15): abstr 1006.
[6] Baselga J, et al. N Engl J Med. 2012;366(6):520-529.
[7] Dhakal A, et al. Breast Cancer (Auckl). 2020;14:1178223420944864.
[8] Layman RM, et al. Lancet Oncol. 2024;25(4):474-487.
[9] Rugo HS, et al. ESMO Open. 2023;8(1): [illegible]

ACKNOWLEDGEMENTS:
- Patients and their families/caregivers for participating in this trial
- Investigators and their support staff who participated in this work
- Celcuity Inc. for sponsoring the study

Poster PDF:
Scan the QR code below to download a PDF of this poster.

RELATED PUBLICATIONS:
Scan the QR codes below to access related publications